Preliminary Safety, Efficacy and Molecular Characterization in Higher-Risk Myelodysplastic Syndrome Patients Treated with Single Agent Emavusertib (CA-4948)

G. Garcia-Manero[1], D. Sallman[2], S. Tarantolo[3], A. Verma[4], S. Groepper[5], A. Mims[6], Y. Abaza[7], J. Dugan[8], V. Garcia de Soria[9], G. Choudhary[10], M. Lane[10], W. Zhao[10], A. Jain[10], C Gallagher[10], U. Platzbecker[11]

1. University of Texas, MD Anderson Cancer Center, Houston, TX; 2. H. Lee Moffitt Cancer Center and Research Institute, Tampa, FL; 3. Nebraska Cancer Specialists, Omaha, NE; 4. Montefiore Medical Center, Bronx, NY; 5. Univ. of Düsseldorf, Düsseldorf, Germany; 6. The Ohio State University, Columbus, OH; 7. Northwestern Medicine, Chicago, IL; 8. Novant Health Cancer Institute, Winston-Salem, NC; 9. Hospital Universitario de La Princesa, Madrid, Spain; 10. Curis Inc., Lexington, MA; 11. University Hospital Leipzig, Leipzig, Germany

INTRODUCTION

- Myelodysplastic syndromes (MDS) are a heterogeneous group of hematologic malignancies in older adults, marked by dysplastic hematopoiesis, cytopenias, and exhibit a dynamic mutational landscape as the disease progresses.[1]
- In higher- risk MDS (hrMDS), the primary treatment goal is to alter the natural progression of the disease, delaying the onset of acute myeloid leukemia and enhancing overall survival. However, current treatments are typically not curative, and many patients face relapse or resistance to first-line therapies.[2]
- Splicing factor mutations (SFm) are frequently reported in AML and MDS and have been associated with disease progression and poor prognosis in relapsed/refractory (R/R) diseases.[1,2]
- Emavusertib is a potent oral inhibitor of IRAK4, FLT3 (ITD and TKD), and CLK (1, 2, and 4), conferring preclinical efficacy advantages when compared with other IRAK4 or FLT3 inhibitors. Treatment with emavusertib inhibits the NF-kB and MAPK pathways, thus offering a potential mechanism to address known pathways of resistance to BCL2 and FLT3 inhibitors.[2,3,4,5]
- As of October 12, 2024, the ongoing TakeAim Leukemia trial (NCT04278768) has treated 49 hrMDS patients in monotherapy. Among patients with < 3 prior lines of therapy who received 300 mg BID, 7 patients were treated for either 7 or 14 days in a 28-day cycle and 14 patients with SFm received continuous dosing.

METHOD

- The safety, clinical activity, and potential biomarkers of emavusertib in relapsed/refractory (R/R) AML and hrMDS (IPSS-R score> 3.5) are being investigated.
- We present preliminary efficacy of emavusertib in hrMDS patients with SFm (U2AF1 and SF3B1) at continuous dosing and those at intermittent dosing, including molecular disease characterization.
- Mutational profiles of patients were documented based on local testing results. Bone marrow and peripheral blood of enrolled patients were collected at the baseline and on treatment.

STUDY POPULATION

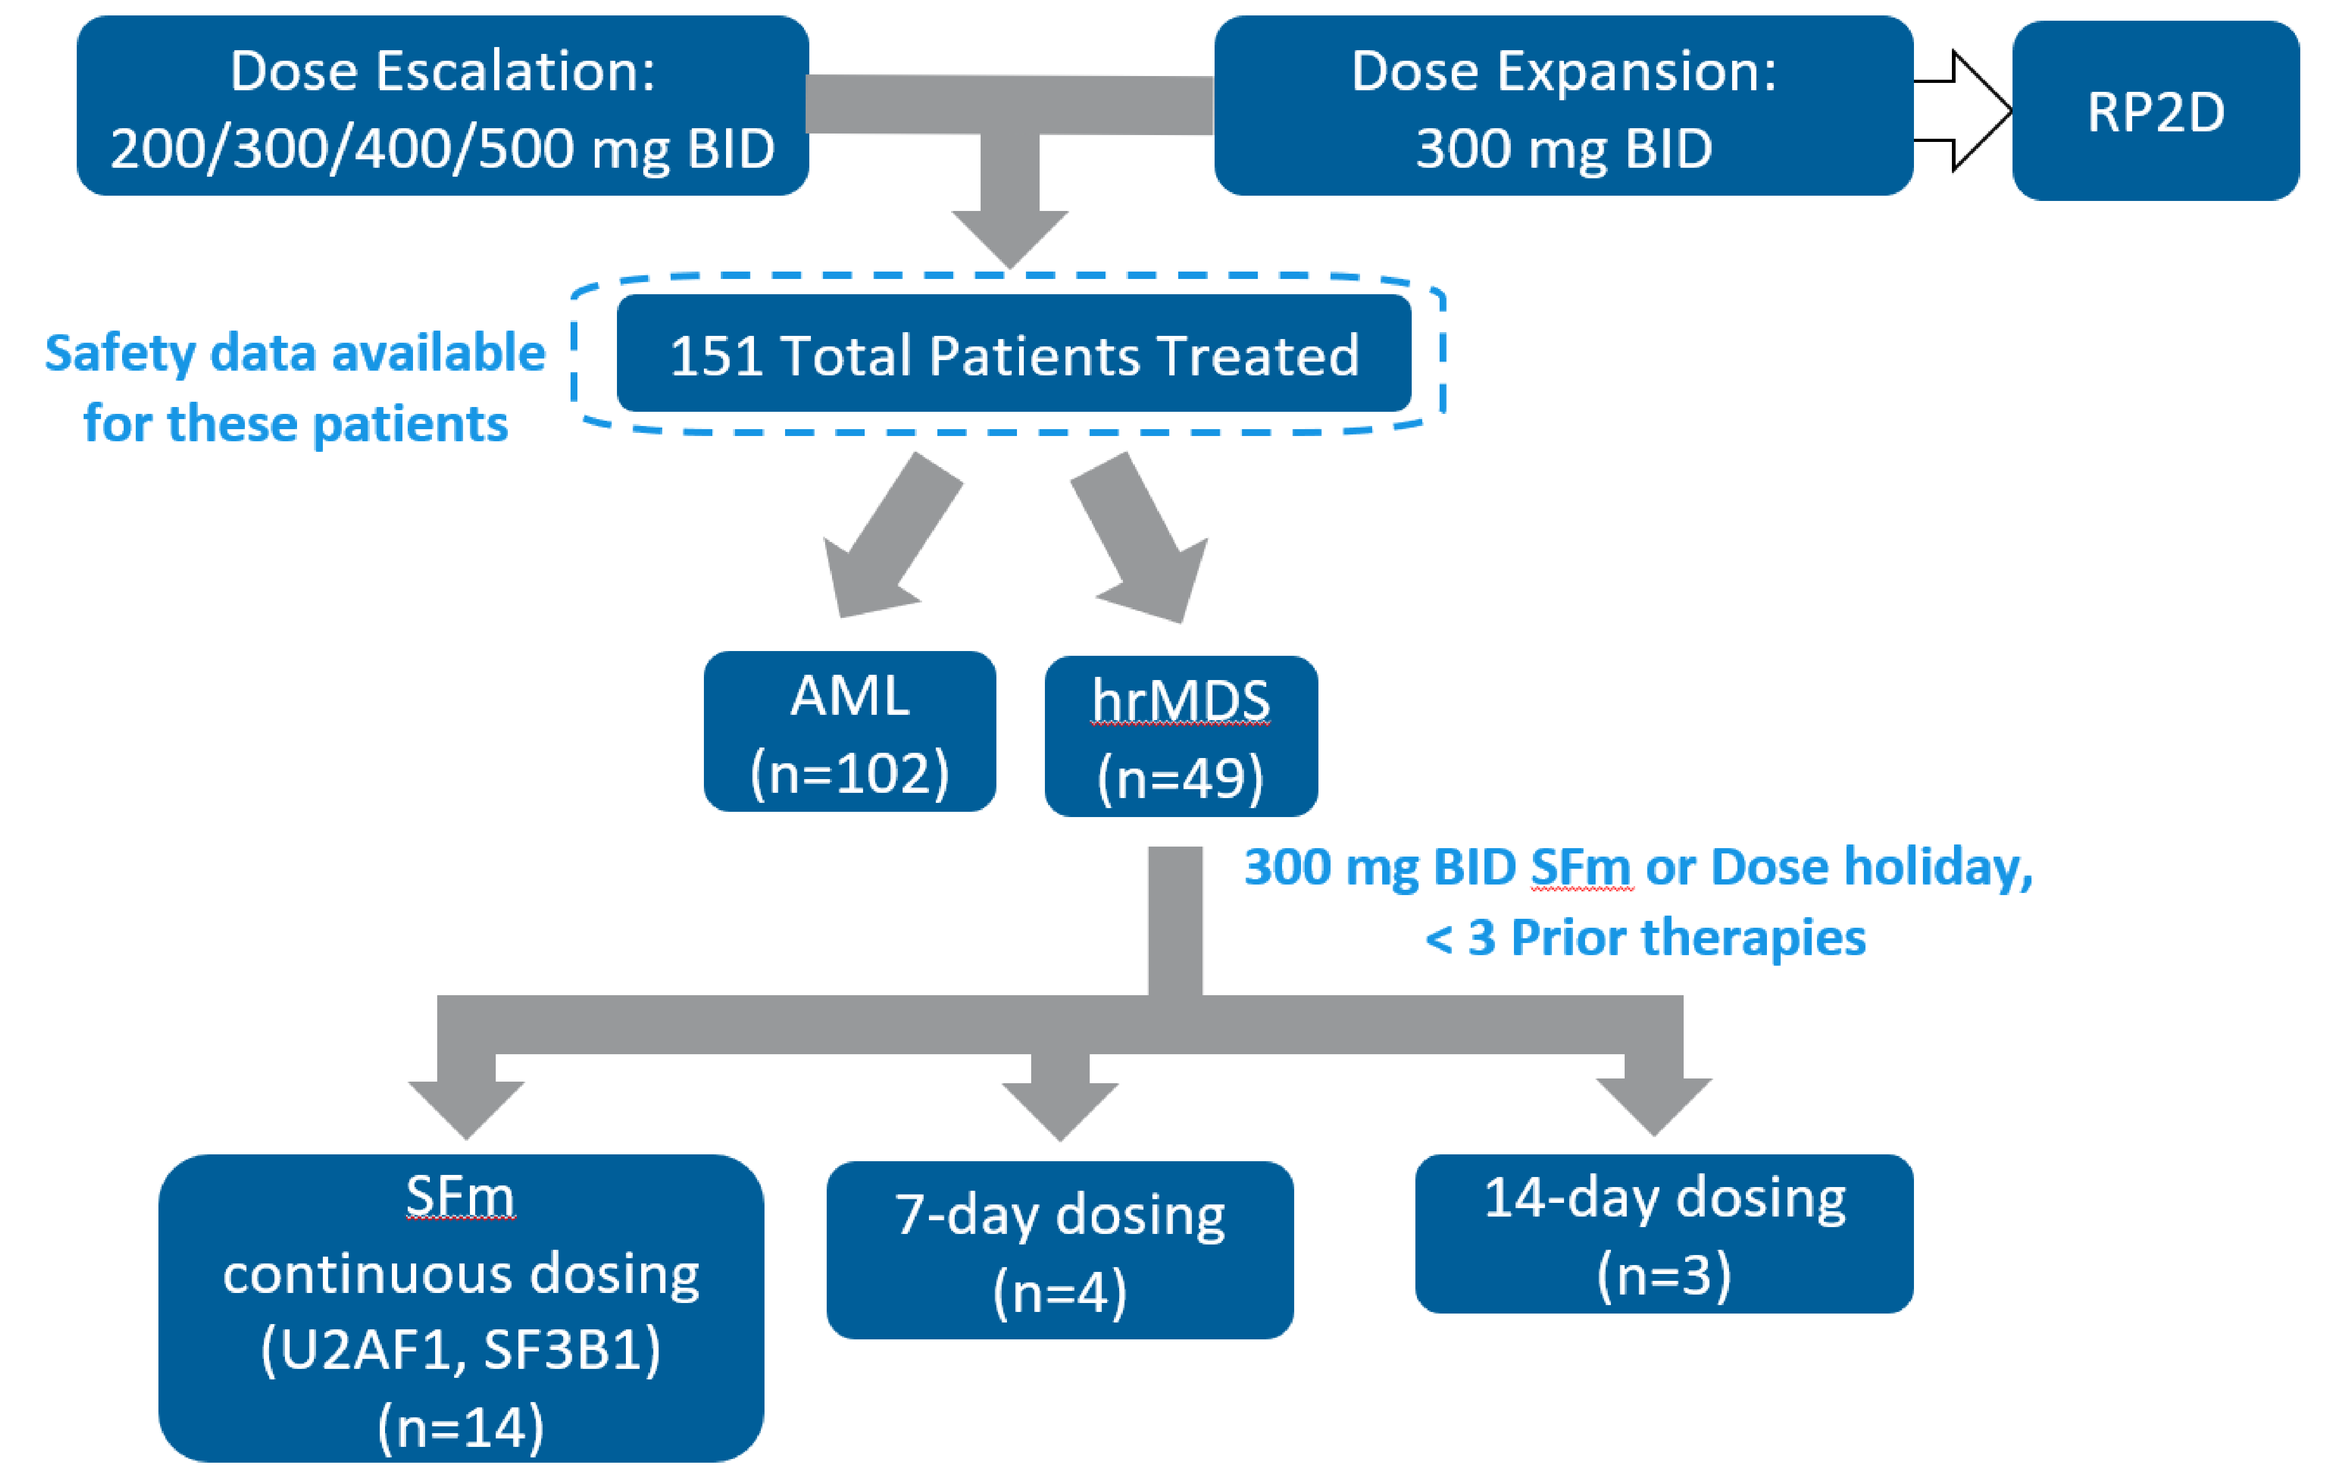
Dose Escalation: 200/300/400/500 mg BID
Dose Expansion: 300 mg BID
RP2D
Safety data available for these patients
151 Total Patients Treated
AML (n=102)
hrMDS (n=49)
300 mg BID SFm or Dose holiday, < 3 Prior therapies
SFm continuous dosing (U2AF1, SF3B1) (n=14)
7-day dosing (n=4)
14-day dosing (n=3)

RESULTS

Baseline Characteristics

		Continuous dosing	Dose holiday	
		hr-MDS SFm (N=14)	hr-MDS - 7 Days (N=4)	hr-MDS - 14 Days (N=3)
Male n (%) : Female n (%)		12 (85.7) : 2 (14.3)	0 : 4 (100)	1 (33.3) : 2 (66.7)
Age (yrs): median (range)		74.5 (69, 79)	77.5 (75, 85)	74 (57, 81)
Race n (%)	Asian	1 (7.1)	0	0
	Black or African American	0	0	0
	White	11 (78.6)	4 (100)	3 (100)
	Others	2 (14.3)	0	0
Median bone marrow blast (%) (range)		8.5 (3, 16)	3.5 (2, 6)	7 (7, 8)
1 prior line n (%): 2 prior lines n (%)		8 (57.1): 6 (42.9)	3 (75): 1 (25)	3 (100): 0
Neutrophils (G/L): median (range)		0.75 (0.09, 3.54)	0.57 (0.19, 0.79)	0.44 (0.23, 3.03)
Platelets (G/L): median (range)		39.5 (3, 243)	24 (17, 71)	31 (31, 32)

Single-agent activity in hrMDS

A hrMDS patients with *SFm continuous dosing*

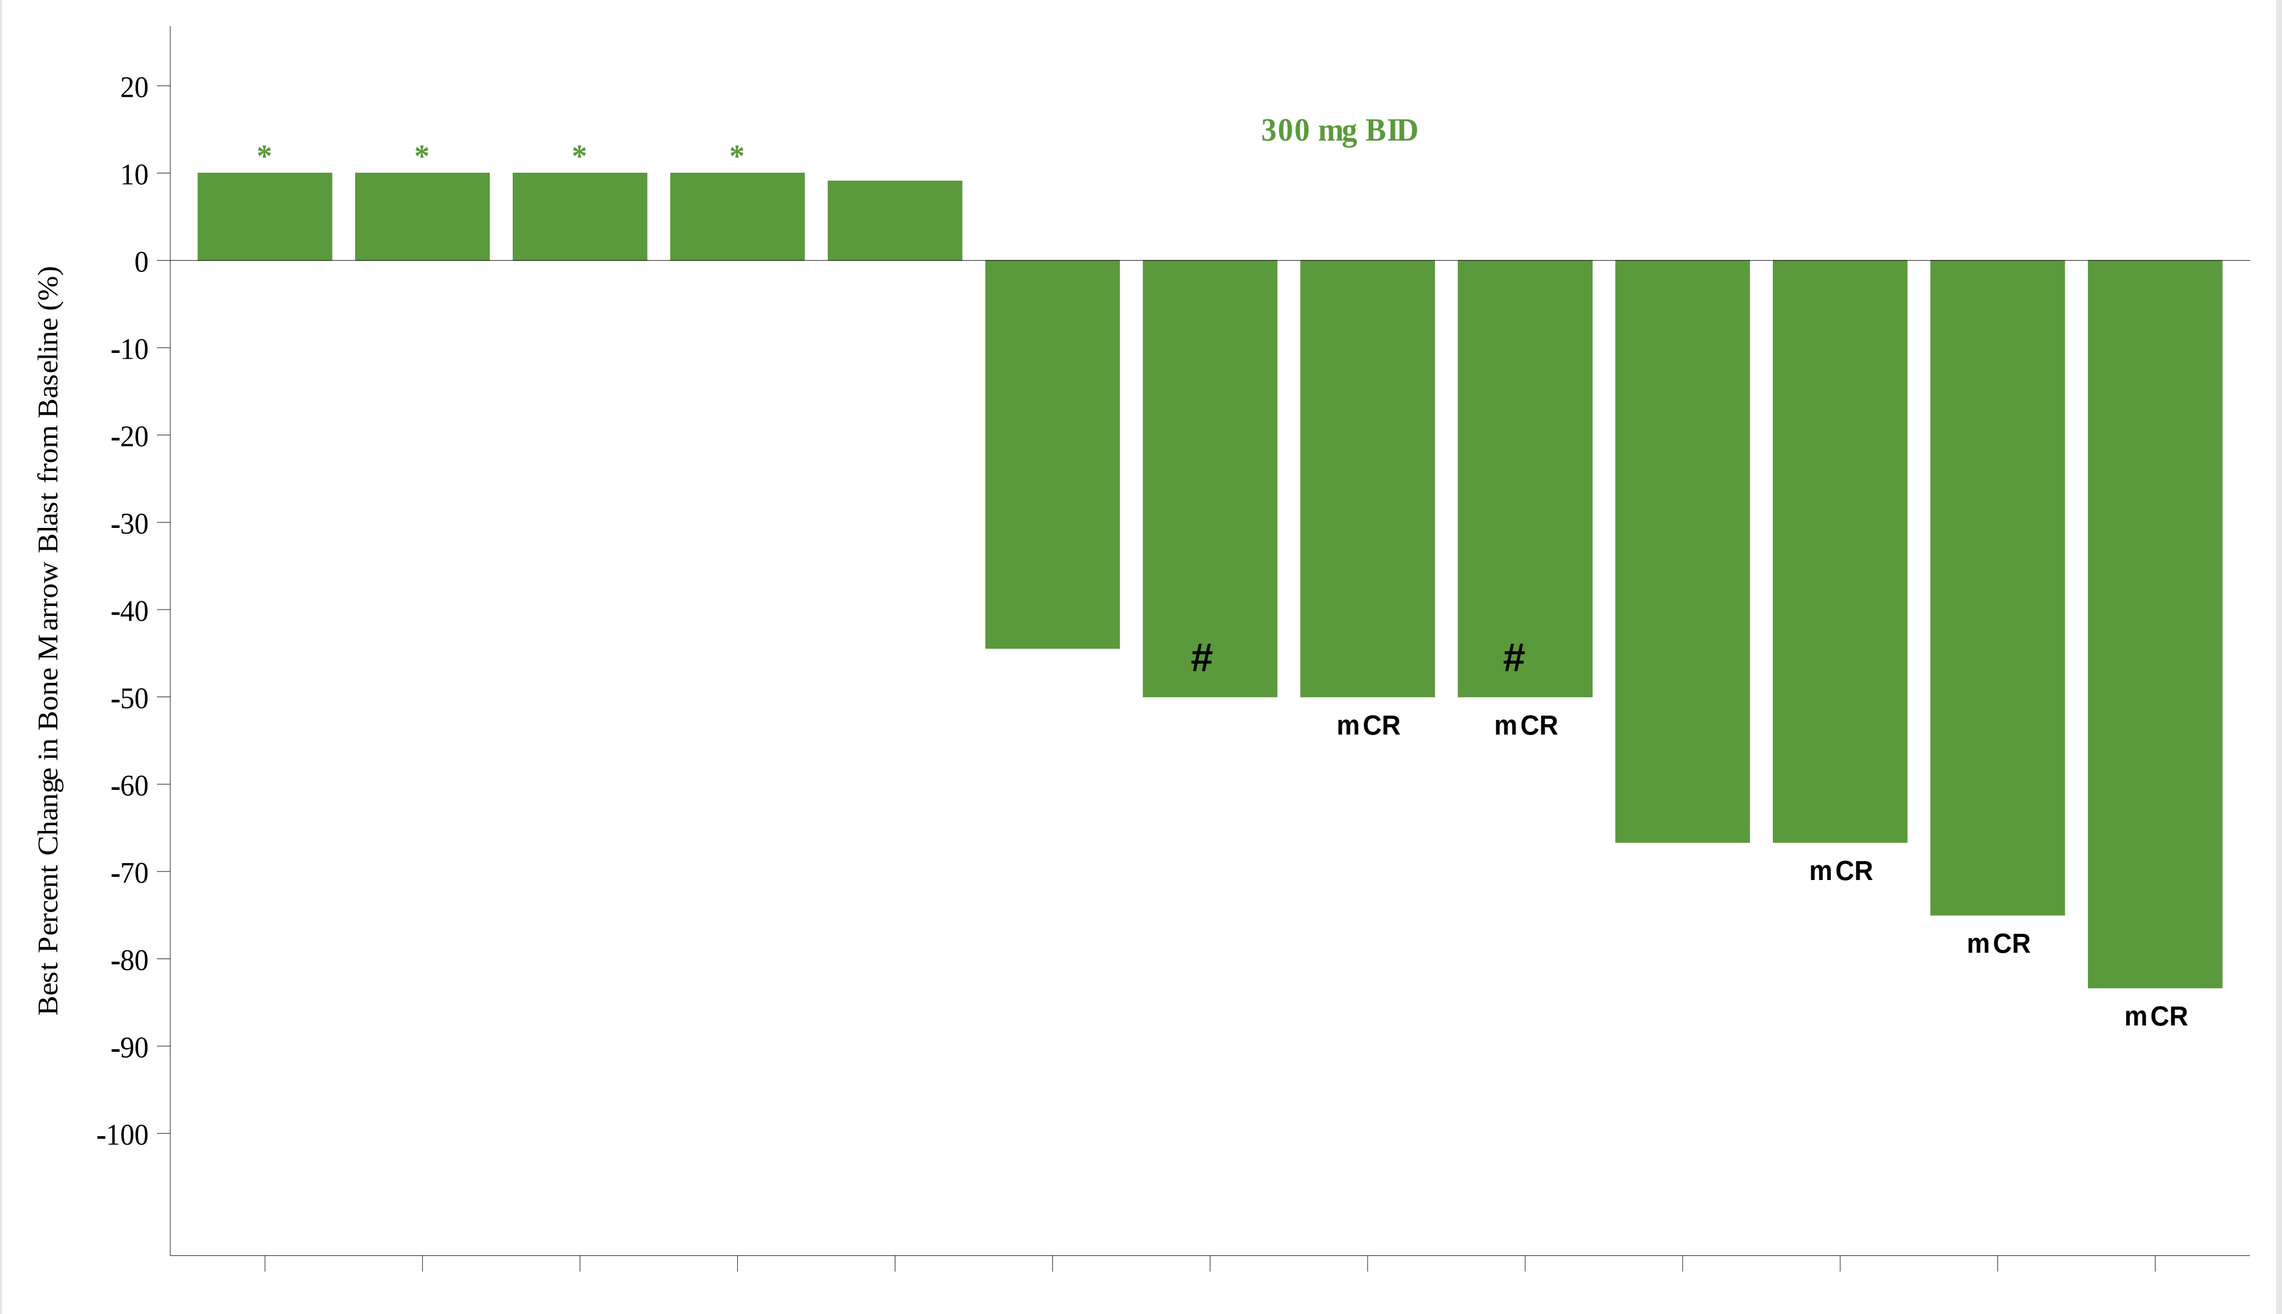
300 mg BID
Best Percent Change in Bone Marrow Blast from Baseline (%)
mCR
#

B hrMDS patients dosed for either *7- or 14-day in 28-day cycle*

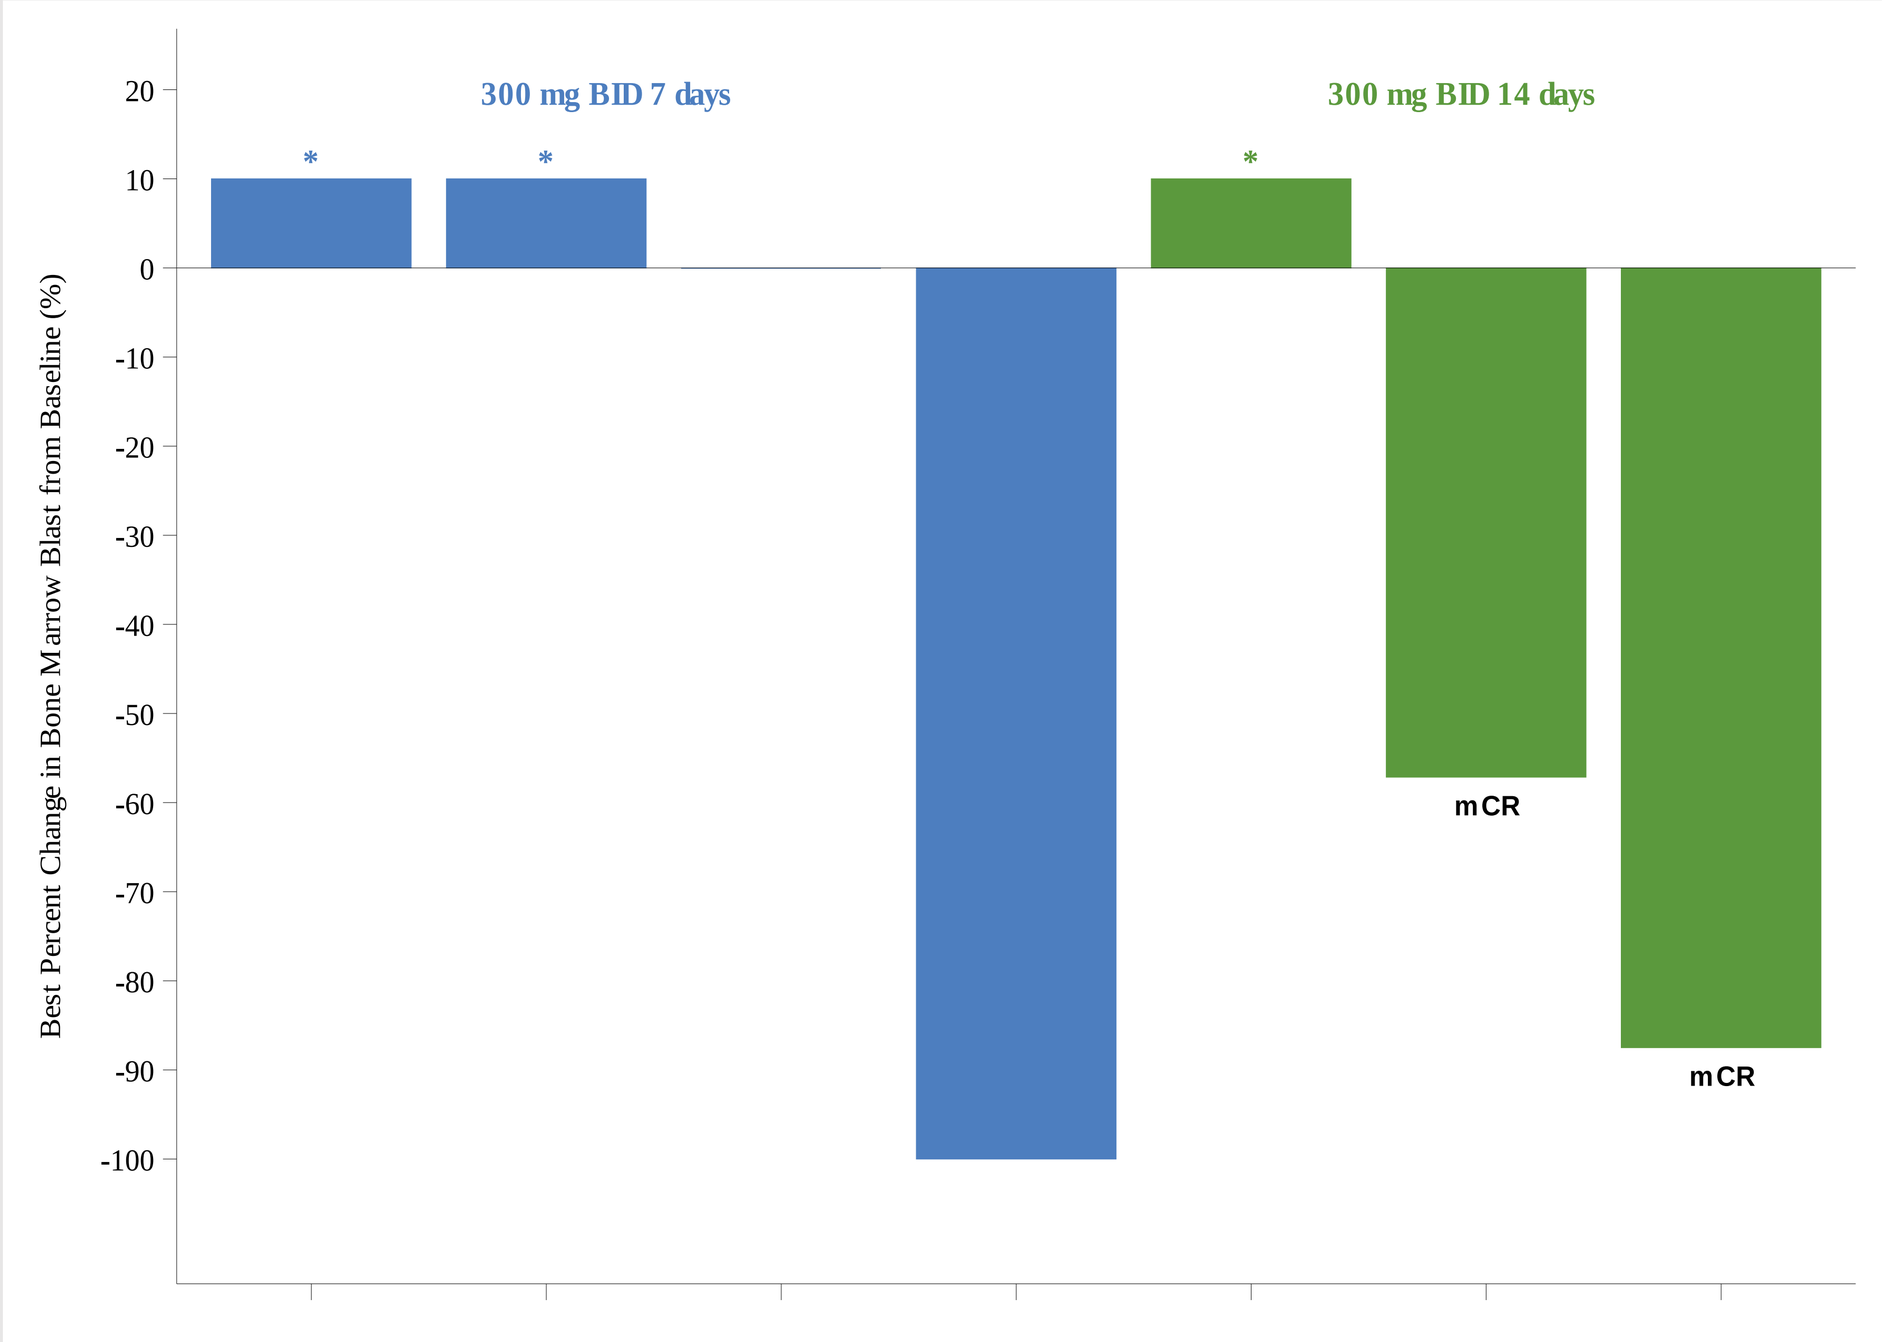
300 mg BID 7 days
300 mg BID 14 days
Best Percent Change in Bone Marrow Blast from Baseline (%)
mCR

Includes all patients that had baseline and post-treatment bone marrow blast assessments.
* Indicates best percentage change from baseline >10%.
Patient went to stem cell or bone marrow transplantation.
- One patient was not included EOT because of AE before first assessment.

* indicates the best percentage change from baseline >10%

Treatment-related adverse events (TRAEs) Grade ≥ 3 in all patients

Grade 3+ Treatment-Related Adverse Event reported in > 1 patients, n (%)	200 mg BID (N = 27)	300 mg BID (N = 99)	300 mg BID 7 days (N = 4)	300 mg BID 14 days (N = 3)	400 mg BID (N = 15)	500 mg BID (N = 3)	Total (N=151)
# of patients having grade 3+ TRAEs	4 (14.8)	36 (36.4)	0	1 (33.3)	7 (46.7)	2 (66.7)	50 (33.1)
Platelet count decreased	1 (3.7)	5 (5.1)	0	0	2 (13.3)	0	8 (5.3)
Blood creatine phosphokinase increased	0	7 (7.1)	0	0	0	0	7 (4.6)
Anaemia	0	6 (6.1)	0	0	0	0	6 (4.0)
Neutropenia	0	5 (5.1)	0	0	1 (6.7)	0	6 (4.0)
Rhabdomyolysis *	0	2 (2.0)	0	0	1 (6.7)	1 (33.3)	4 (2.6)
Aspartate aminotransferase increased	1 (3.7)	2 (2.0)	0	0	0	0	3 (2.0)
Lipase increased	0	2 (2.0)	0	1 (33.3)	0	0	3 (2.0)
Neutrophil count decreased	0	2 (2.0)	0	0	1 (6.7)	0	3 (2.0)
Alanine aminotransferase increased	2 (7.4)	0	0	0	0	0	2 (1.3)
Dizziness	1 (3.7)	1 (1.0)	0	0	0	0	2 (1.3)
Fatigue	0	1 (1.0)	0	0	1 (6.7)	0	2 (1.3)
Leukopenia	0	2 (2.0)	0	0	0	0	2 (1.3)
Orthostatic hypotension	0	2 (2.0)	0	0	0	0	2 (1.3)
Syncope	0	1 (1.0)	0	0	0	1 (33.3)	2 (1.3)
Thrombocytopenia	0	2 (2.0)	0	0	0	0	2 (1.3)

Note: After discussion with regulatory authorities of investigator-reported AEs, objective laboratory criteria for the determination of rhabdomyolysis were adopted from existing approved drug labels (CPK >10 x ULN and SCr ≥ 1.5 x ULN). Previously, reported events of rhabdomyolysis were determined by subjective criteria. Using the objective laboratory criteria, rhabdomyolysis was reported in 1/151 patients.

Duration of treatment in hrMDS patients

A hrMDS patients with *SFm continuous dosing*

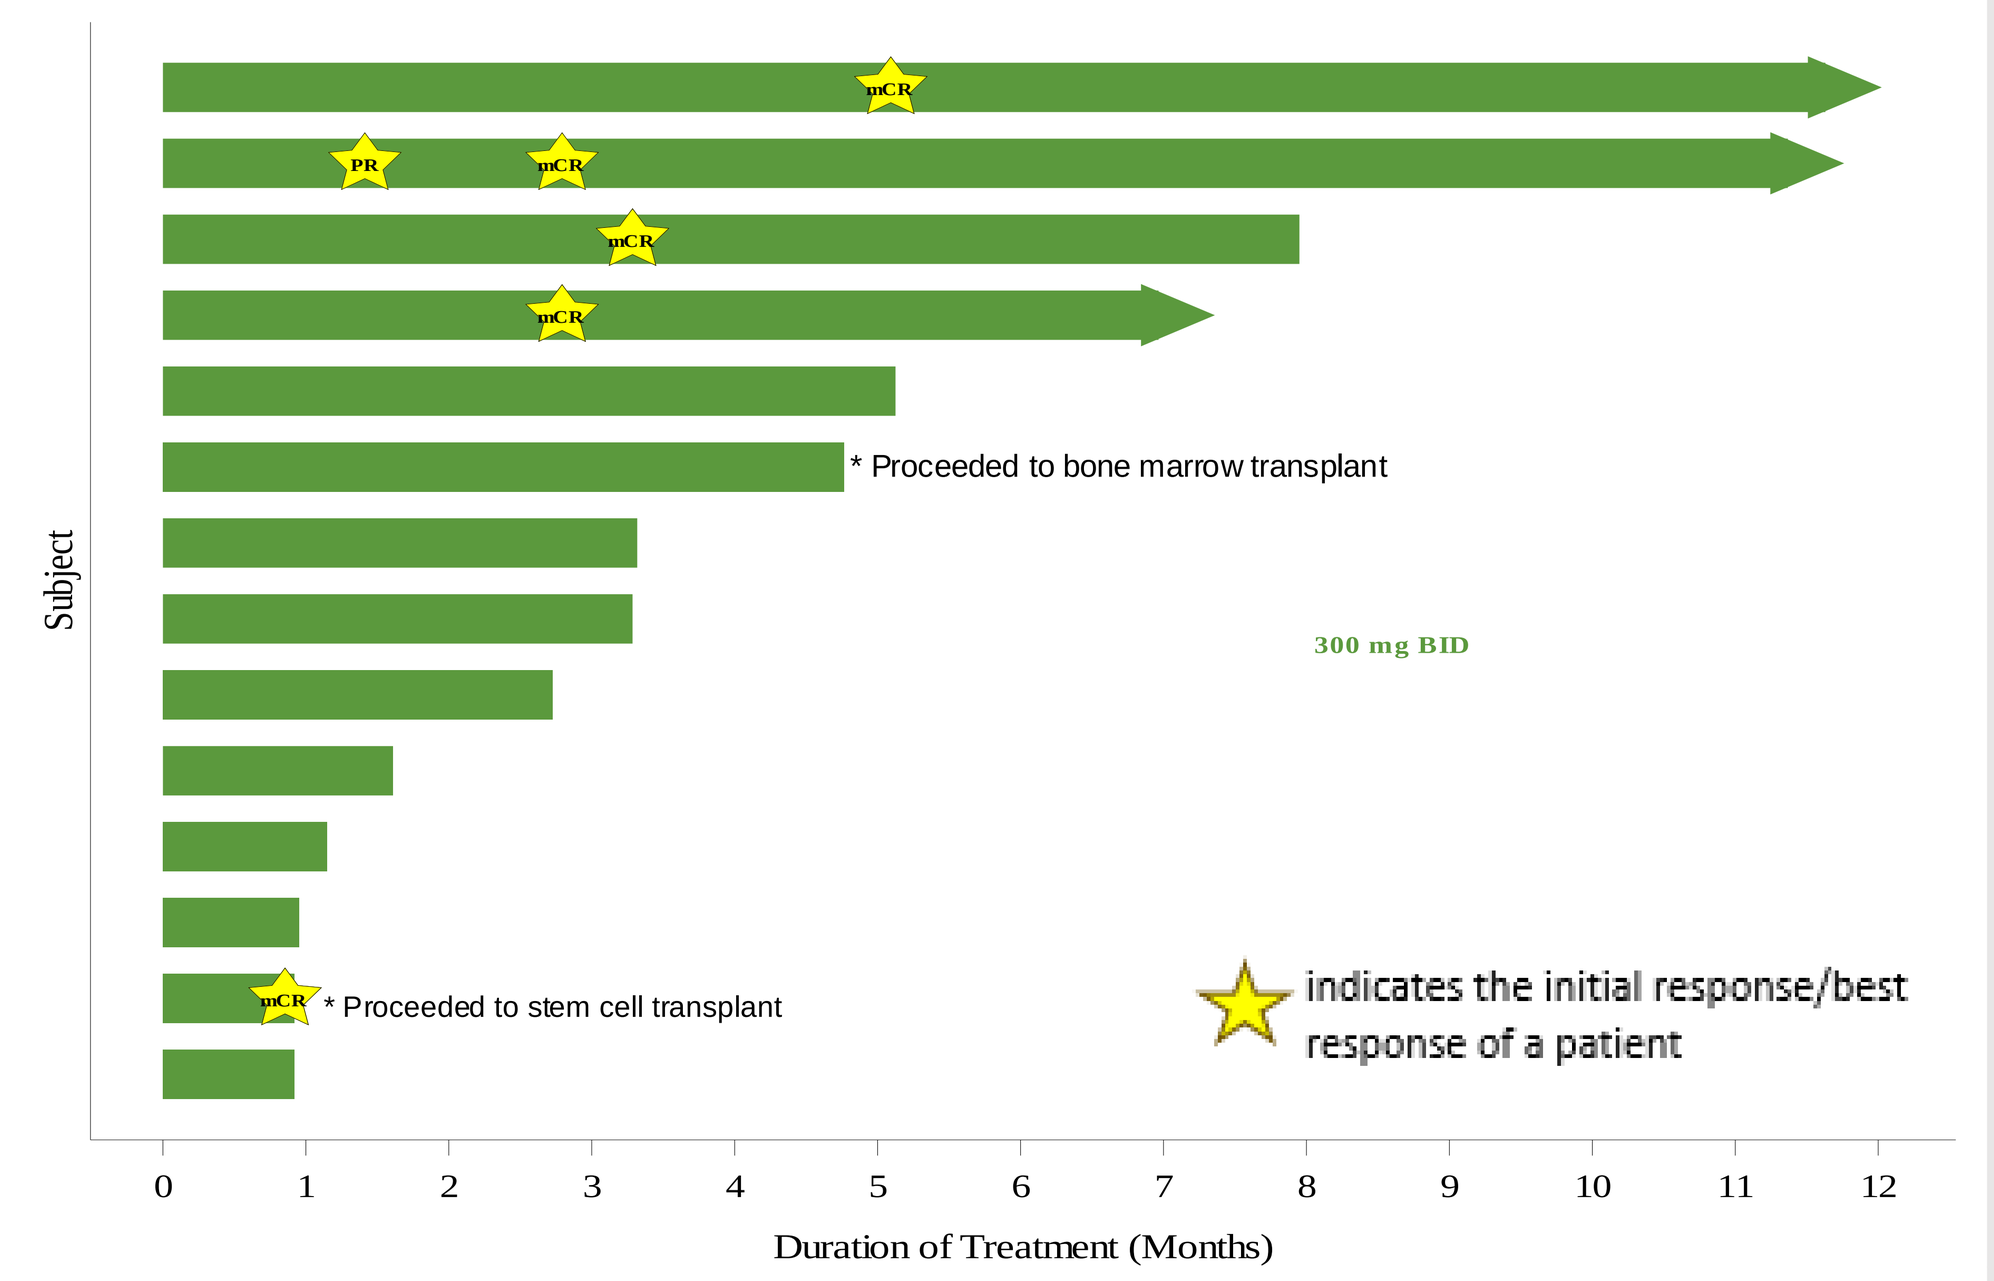
mCR
PR
* Proceeded to bone marrow transplant
300 mg BID
* Proceeded to stem cell transplant
indicates the initial response/best response of a patient
Subject
Duration of Treatment (Months)

Median time to first response was 85 days

B hrMDS patients dosed for either *7- or 14-day in 28-day cycle*

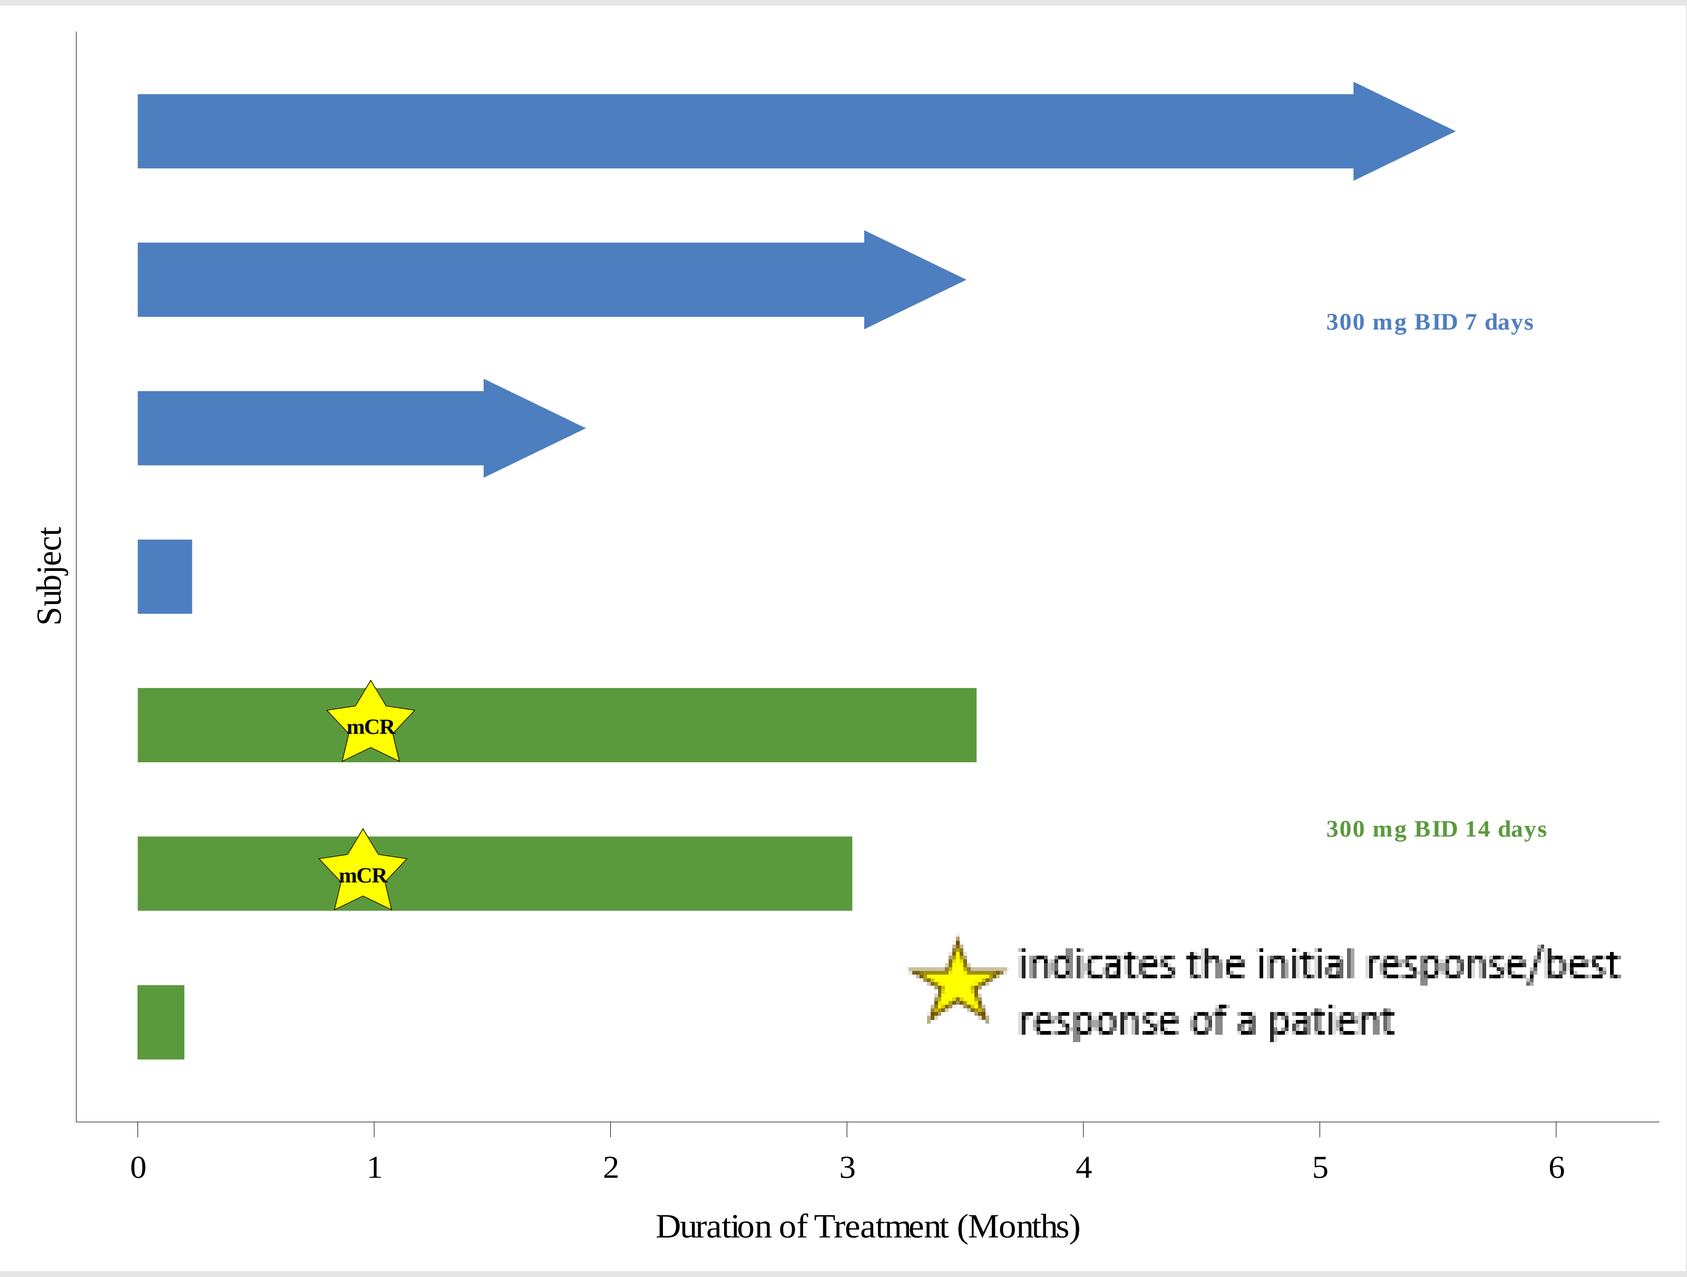
300 mg BID 7 days
300 mg BID 14 days
mCR
indicates the initial response/best response of a patient
Subject
Duration of Treatment (Months)

Clinical activity in responders (mCR) in hrMDS

Patient #	Age	Sex	IPSS-R SCORE	# prior therapy	Prior BCL2i	Prior HMA	Dosing schedule	Co-mutations At Baseline
1	77	M	5.5	1	N	Y	Continuous	DNMT3A, U2AF1, BCOR, STAG2, BCORL1, ETV6, SET3BP1
2	77	M	4.5	1	N	Y	Continuous	SF3B1, TET2, SMC1A, CHEK2
3	74	F	5.5	2	N	Y	Continuous	ASXL1, DNMT3A, SF3B1, TET2, EZH2
4	74	F	6	1	N	Y	Continuous	SF3B1, IDH1
5	69	M	6	2	N	Y	Continuous	BCOR, DNMT3A, STAG2, TET2, U2AF1, RUNX1
6	57	F	6	1	Y	Y	Dose holiday – 14 day	WT1, GATA2
7	81	F	7	1	N	Y	Dose holiday – 14 day	ASXL1, ETNK1, PHF6, RUNX1, SRSF2, TET2, U2AF1, BCOR

CONCLUSIONS

- Emavusertib has an acceptable and manageable safety profile in R/R AML and hrMDS patients.
- The mutation profiles of responders indicate that emavusertib may be able to target diverse underlying genetic mechanisms of resistance to venetoclax and hypomethylating regimens. This is suggestive of the disease-modifying activity of emavusertib.
- Emavusertib has demonstrated anti-leukemic activity in patients with *SFm* and in the 14-day dosing cohort.
- Enrollment in this trial is continuing at the RP2D dose of 300 mg BID in hrMDS patients with < 3 prior lines of therapy in the 7- and 14-day dosing cohort.

REFERENCES

1. Garcia-Manero G. Am J Hematol. 2023; 98(8), 1307-1325
2. Platzbecker, U *et al.* Leukemia. 2021;35(8):2182-2198
3. Grob T *et al* J Clin Oncol 2023; 41(4) 756-765
4. Smith M *et al.* Nat Cell Biol. 2019 May;21(5):640-650
5. Gummadi VR et al. ACS Med Chem Lett 2020; 11(12):2374-2381

ACKNOWLEDGEMENTS

We would like to thank the patients, their families and caregivers for their invaluable contribution and participation in this study.

CONTACT INFORMATION

Maureen E. Lane, Ph.D.
VP, Clinical and Translational Science, CURIS
mlane@curis.com