TPS6587: A Phase 1 Single-Arm, Open-Label Study of Emavusertib (CA-4948) in Combination with Azacitidine and Venetoclax in Acute Myeloid Leukemia Patients in Complete Response with Measurable Residual Disease

Adolfo de la Fuente, MD[1], Claudio Cerchione, MD, PhD[2], Sebastian Scholl, MD[3], Jan Moritz Middeke, MD[4], Nitika, PhD[5], Maureen E Lane, PhD[5], Wanying Zhao, PhD[5], Steven Angelides, MS[5], Reinhard von Roemeling, MD[5], Uwe Platzbecker, MD[6]

[1]Department of Hematology, MD Anderson Cancer Center Madrid, Madrid, Spain; [2]Istituto Romagnolo per lo Studio dei Tumori, Meldola, Italy; [3]Universitätsklinikum Jena, Jena, Germany; [4]Technische Universität Dresden, Dresden, Germany; [5]Curis Inc., Lexington, MA; [6]Department of Hematology, University Hospital Leipzig, Leipzig, Germany.

INTRODUCTION

Acute myeloid leukemia (AML) is a heterogenous disease and exhibits a dynamic mutational landscape as the disease progresses.[1] In the VIALE-A study, composite complete response (CRc), which includes complete response (CR) or CR with incomplete hematological recovery (CRi) in association with measurable residual disease (MRD) of <1 residual blast/1000 leukocytes or MRD negative (MRD−) resulted in longer duration of response, event-free survival, and OS than patients who achieved CRc but were MRD positive (MRD+).[2] Single cell sequencing performed in older AML patients showed that primary and adaptive resistance in venetoclax-based combinations was commonly characterized by acquisition or enrichment of clones with FMS-like tyrosine kinase 3 (FLT3)-internal tandem duplication (ITD) mutations.[3] There is no established role for a FLT3 inhibitor in front-line setting for patients who are unsuitable for intensive chemotherapy. The current standard of care is the combination of azacitidine plus venetoclax; however, FLT3-driven relapses are common.[4,5]

Emavusertib is a novel potent oral inhibitor of interleukin-1 receptor-associated kinase 4 (IRAK4) with additional inhibitory activity against FLT3 and CDC-like kinases (CLK1/2/4). Clinical studies with emavusertib monotherapy have demonstrated a significant reduction in blasts, along with CR, in patients who received prior therapy with an hypomethylating agent (HMA) and FLT3 inhibitor. Additionally, emavusertib in combination with azacitidine and venetoclax demonstrated significant anti-leukemic effects in all AML cell lines, including azacitidine- or venetoclax-resistant cell lines.[6] MCL-1 is a prime driver of resistance to venetoclax and targeting IRAK4 has been shown to affect transcription and post translational regulation of MCL-1.[7,8] Given the significant role of FLT3 mutations and MCL-1 in conferring resistance to the combination therapy of azacitidine and venetoclax, we designed this phase 1b study to assess the efficacy of emavusertib in combination with venetoclax and azacitidine in AML patients in CR with MRD. (EUCTR#2023-505828-58).

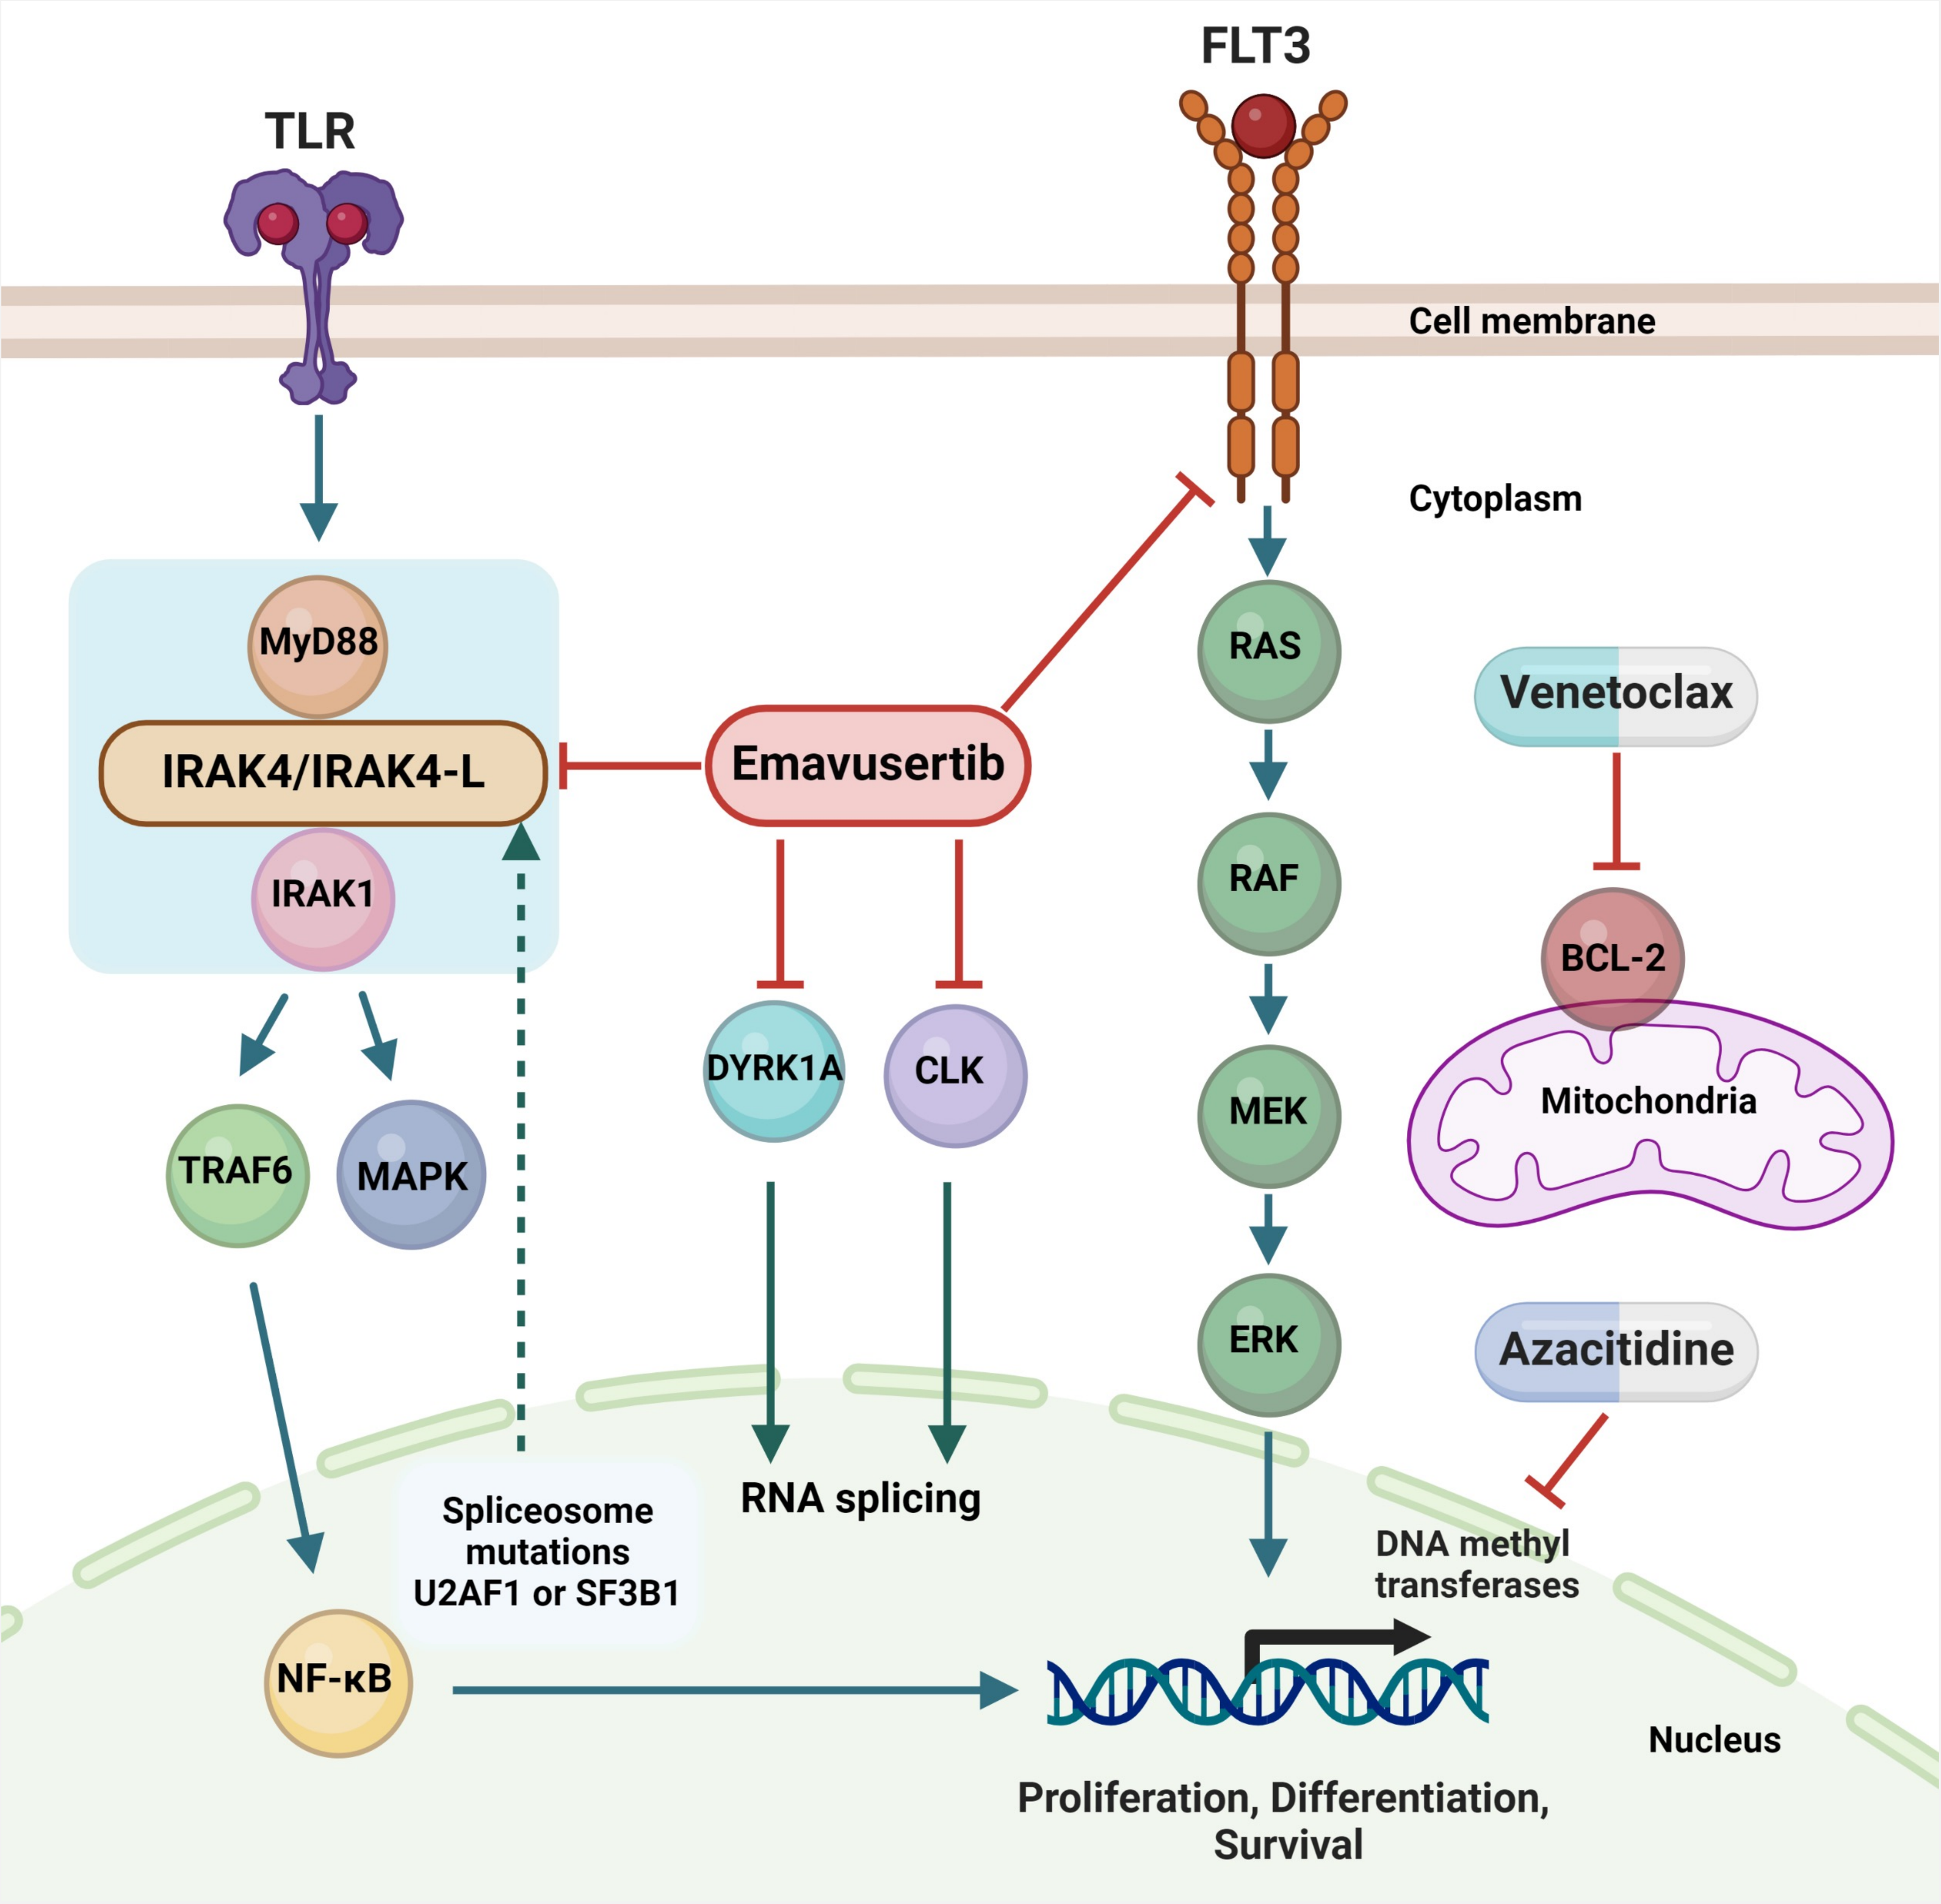
TLR
FLT3
Cell membrane
Cytoplasm
MyD88
IRAK4/IRAK4-L
IRAK1
Emavusertib
RAS
Venetoclax
RAF
BCL-2
Mitochondria
DYRK1A
CLK
MEK
TRAF6
MAPK
ERK
Azacitidine
Spliceosome mutations U2AF1 or SF3B1
RNA splicing
DNA methyl transferases
NF-κB
Nucleus
Proliferation, Differentiation, Survival

The mechanism of action of emavusertib in combination with azacitidine and venetoclax

STUDY OBJECTIVES

Primary	• Evaluate the safety and tolerability of different dosing schedules of emavusertib as an add-on agent to the combination of azacitidine and venetoclax
Secondary	• Conversion of MRD+ to MRD- status • Characterize the pharmacokinetic profiles of emavusertib, azacitidine, and venetoclax • Assess the effects of the triplet regimen (emavusertib + azacitidine + venetoclax) on dynamics of MRD status and the relationship to outcomes • Evaluate anti-cancer activity of the triplet regimen
Exploratory	• Evaluate the molecular profile of peripheral blood at baseline and following treatment with the triplet regimen

STUDY DESIGN

This is a Phase 1, single-arm, open-label study evaluating safety and tolerability, PK, and conversion of MRD status with emavusertib as an add-on agent in AML patients who have received azacitidine + venetoclax as first-line therapy after no more than 6 cycles and have achieved CR or CRh with MRD+ status.

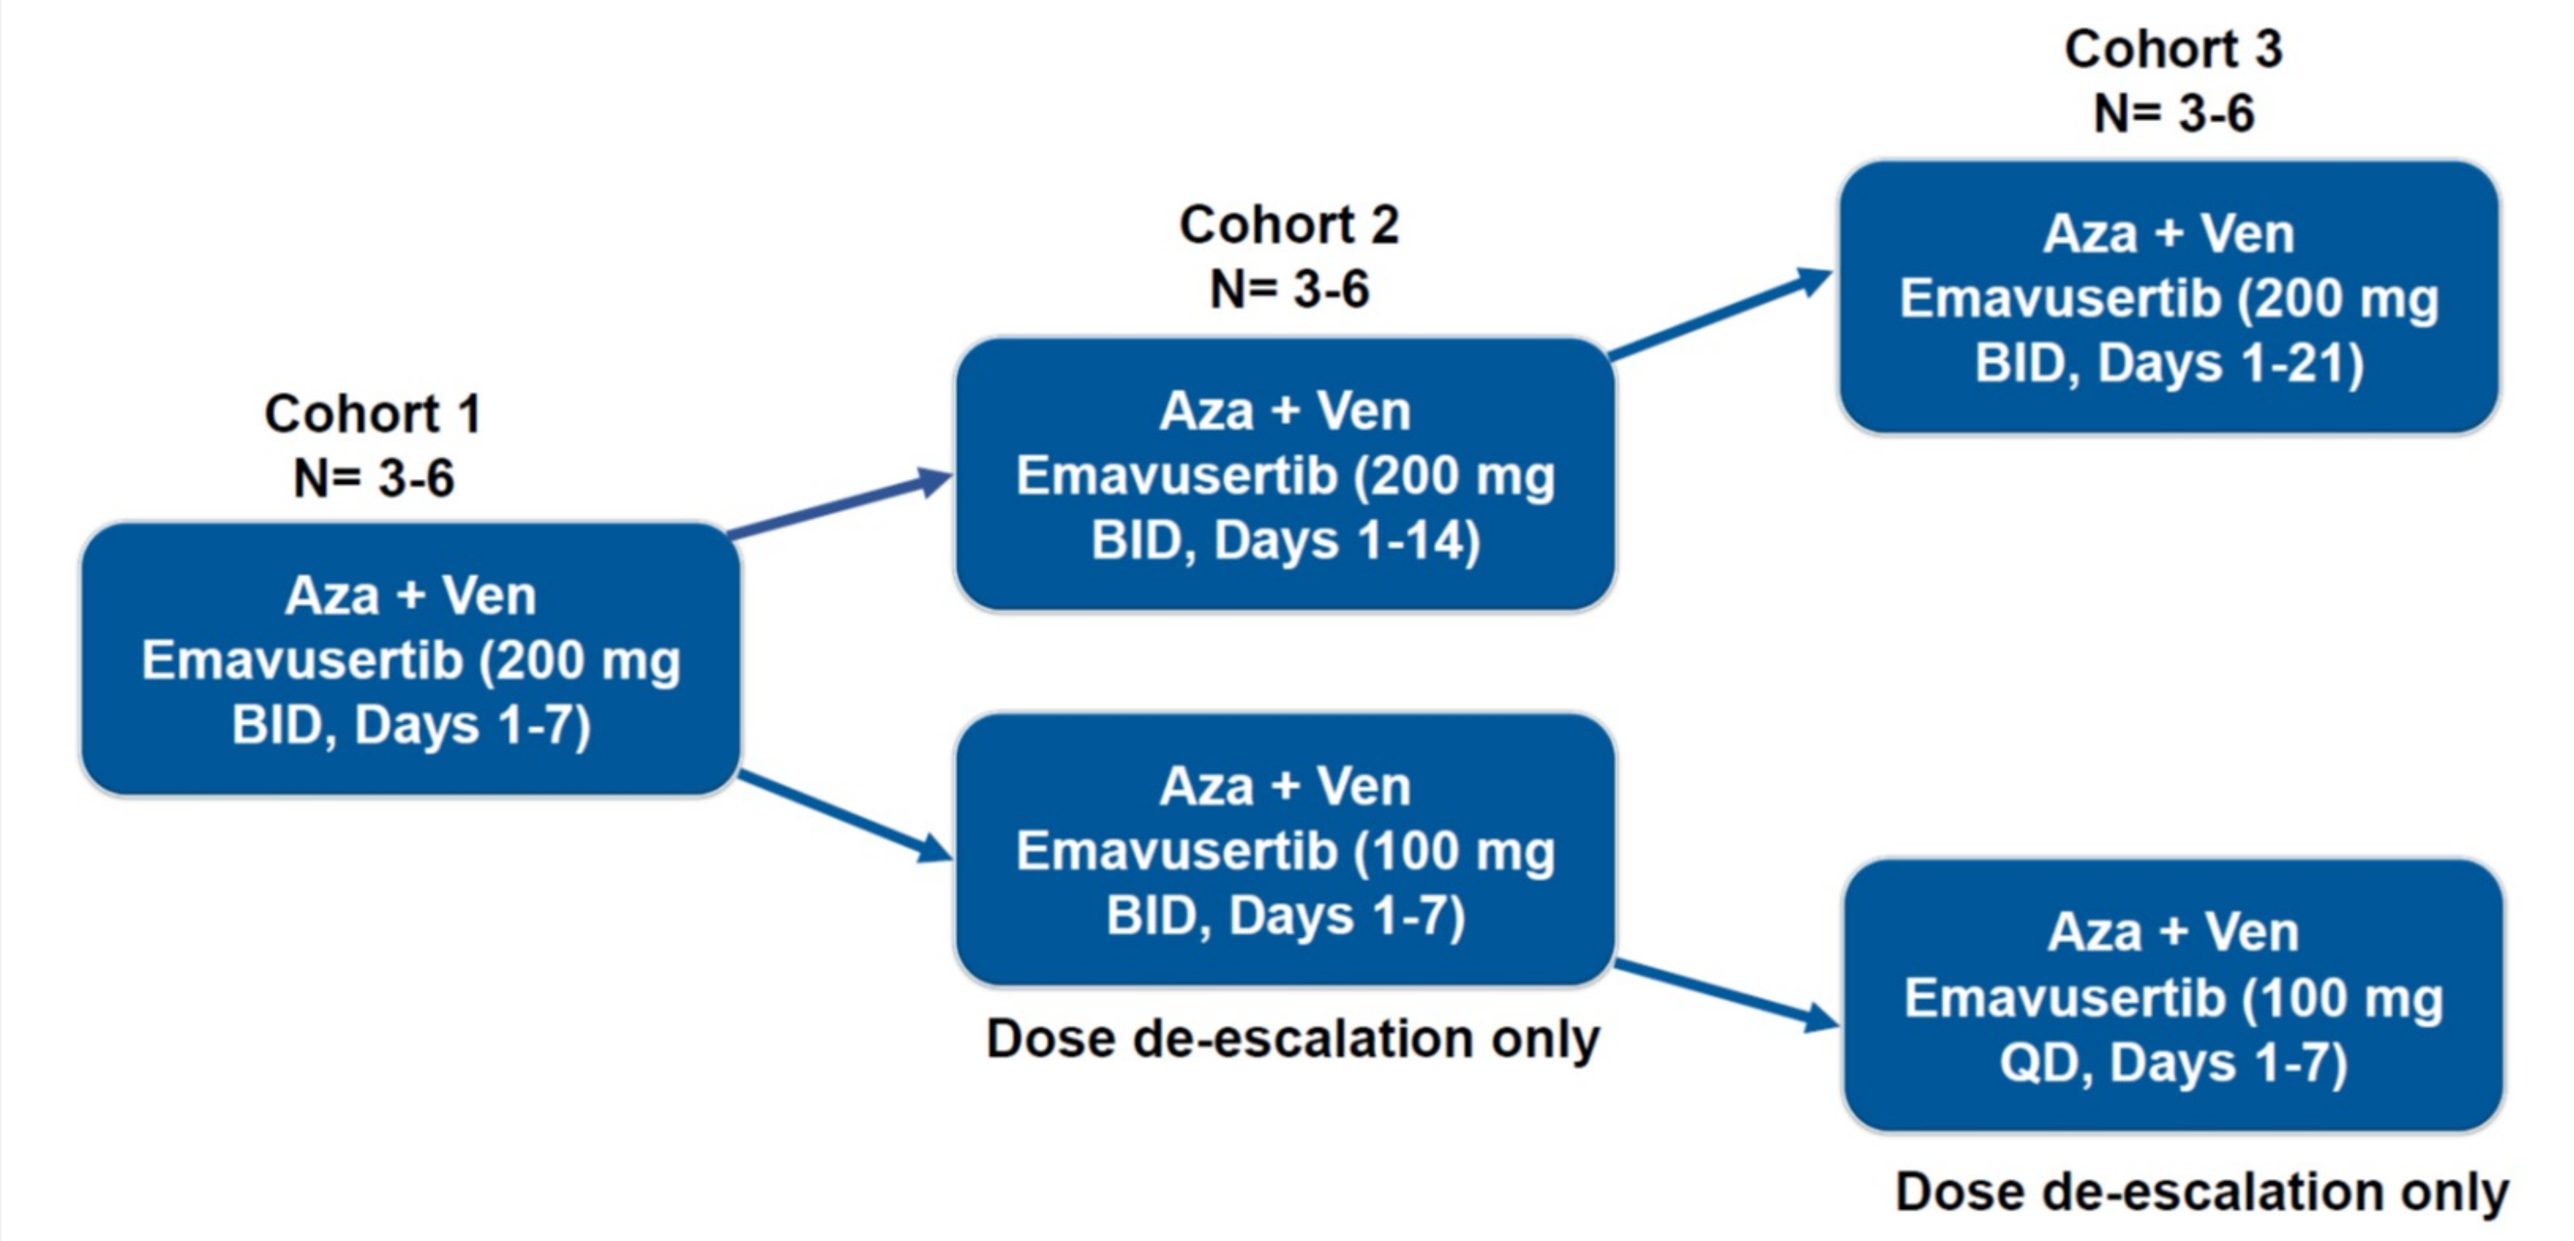
Cohort 1
N= 3-6
Aza + Ven
Emavusertib (200 mg
BID, Days 1-7)
Cohort 2
N= 3-6
Aza + Ven
Emavusertib (200 mg
BID, Days 1-14)
Cohort 3
N= 3-6
Aza + Ven
Emavusertib (200 mg
BID, Days 1-21)
Aza + Ven
Emavusertib (100 mg
BID, Days 1-7)
Dose de-escalation only
Aza + Ven
Emavusertib (100 mg
QD, Days 1-7)
Dose de-escalation only

STUDY SITES

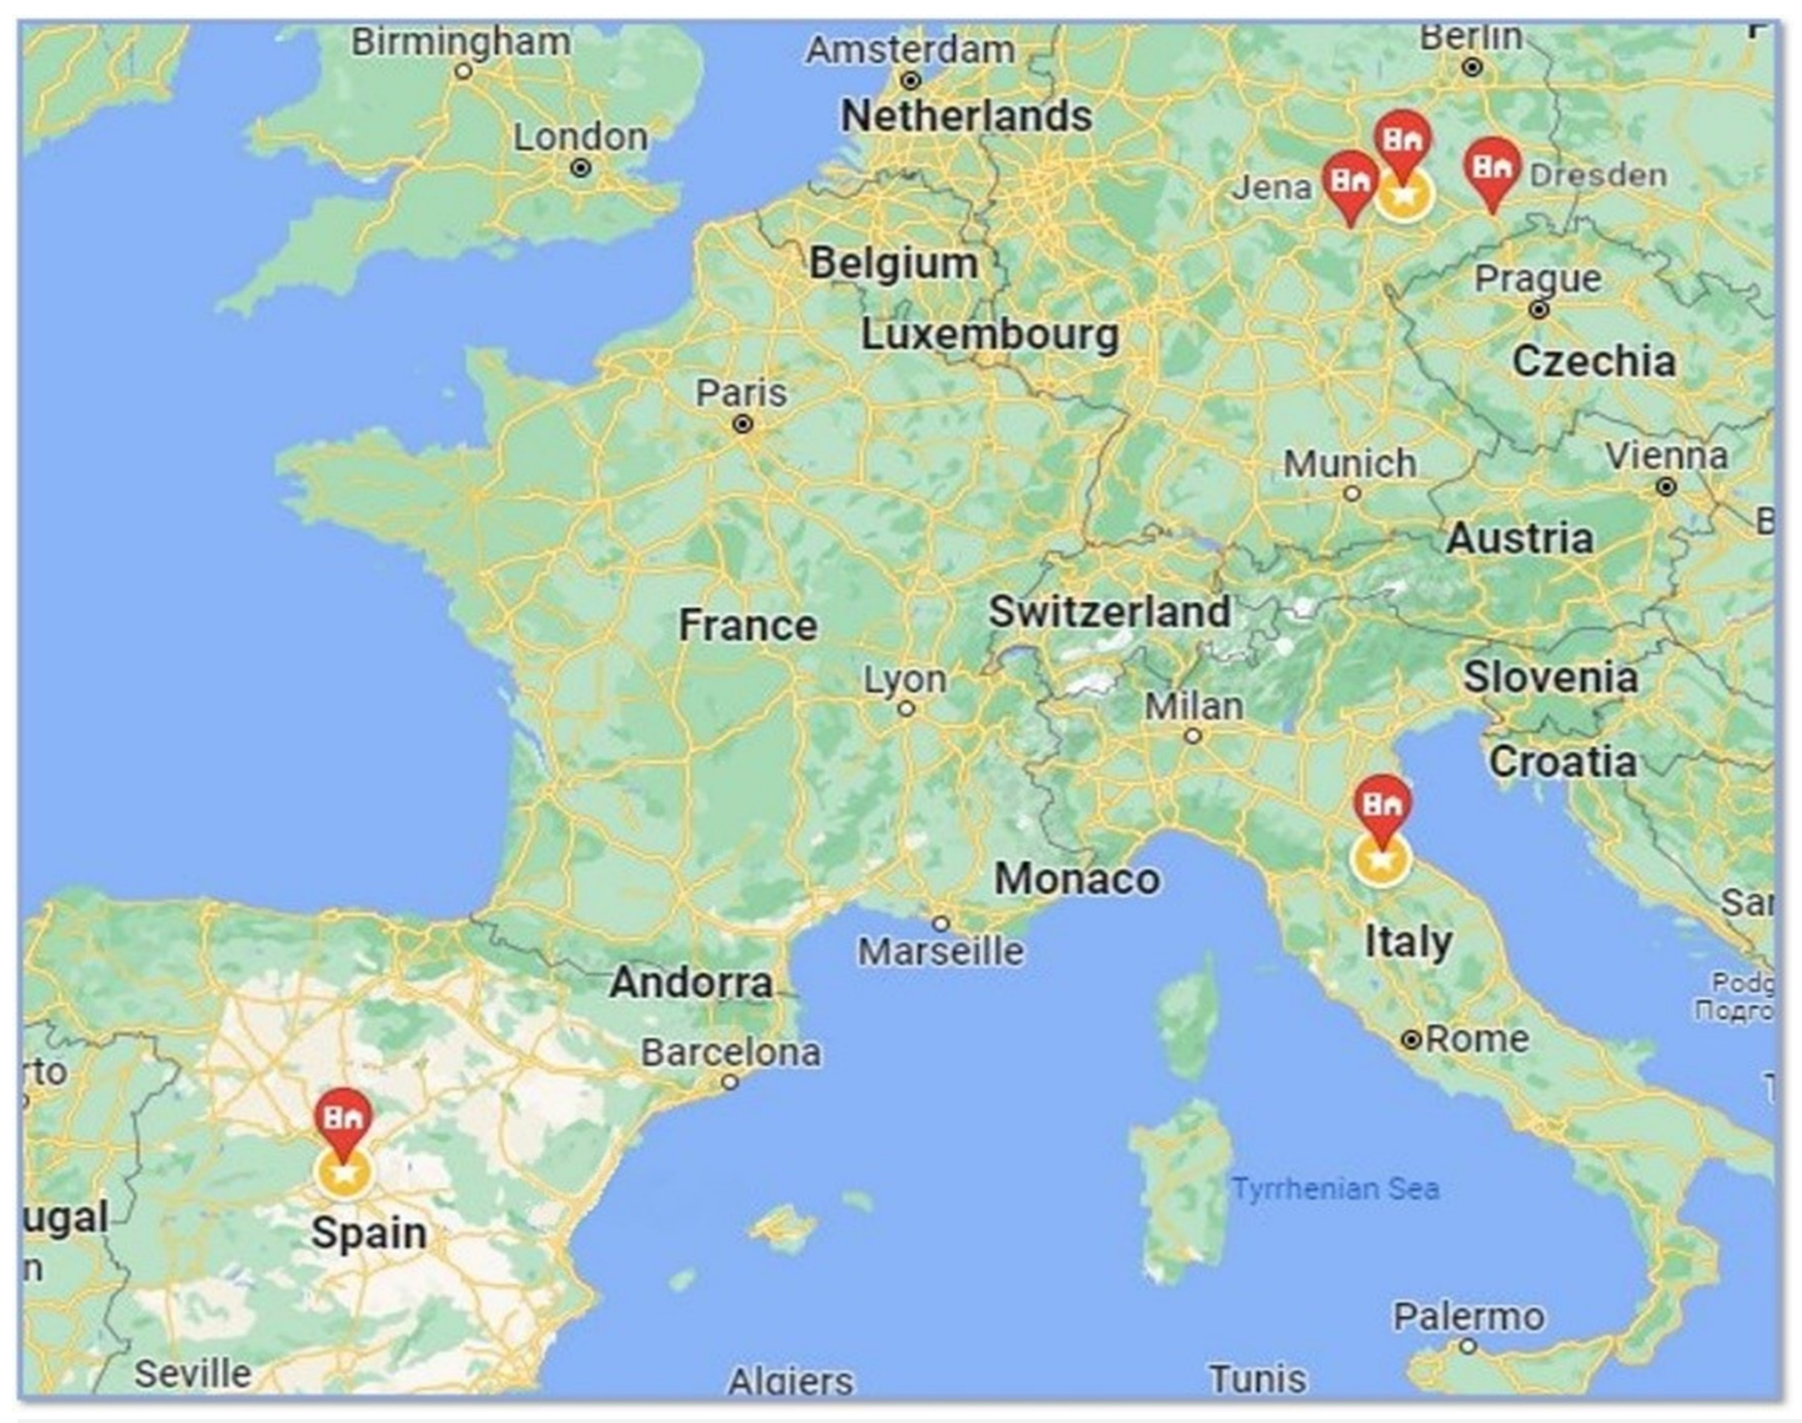
Birmingham
London
Amsterdam
Netherlands
Berlin
Jena
Dresden
Belgium
Luxembourg
Prague
Czechia
Paris
Munich
Vienna
Austria
France
Switzerland
Lyon
Milan
Slovenia
Croatia
Monaco
Marseille
Italy
Andorra
Barcelona
Rome
Spain
Tyrrhenian Sea
Palermo
Seville
Algiers
Tunis

- MD Anderson, Madrid, Spain
- Istituto Romagnolo per lo Studio dei Tumori, Meldola, Italy
- University of Jena, Jena, Germany
- University of Dresden, Dresden, Germany
- University Hospital Leipzig, Leipzig, Germany

STUDY ELIGIBILITY

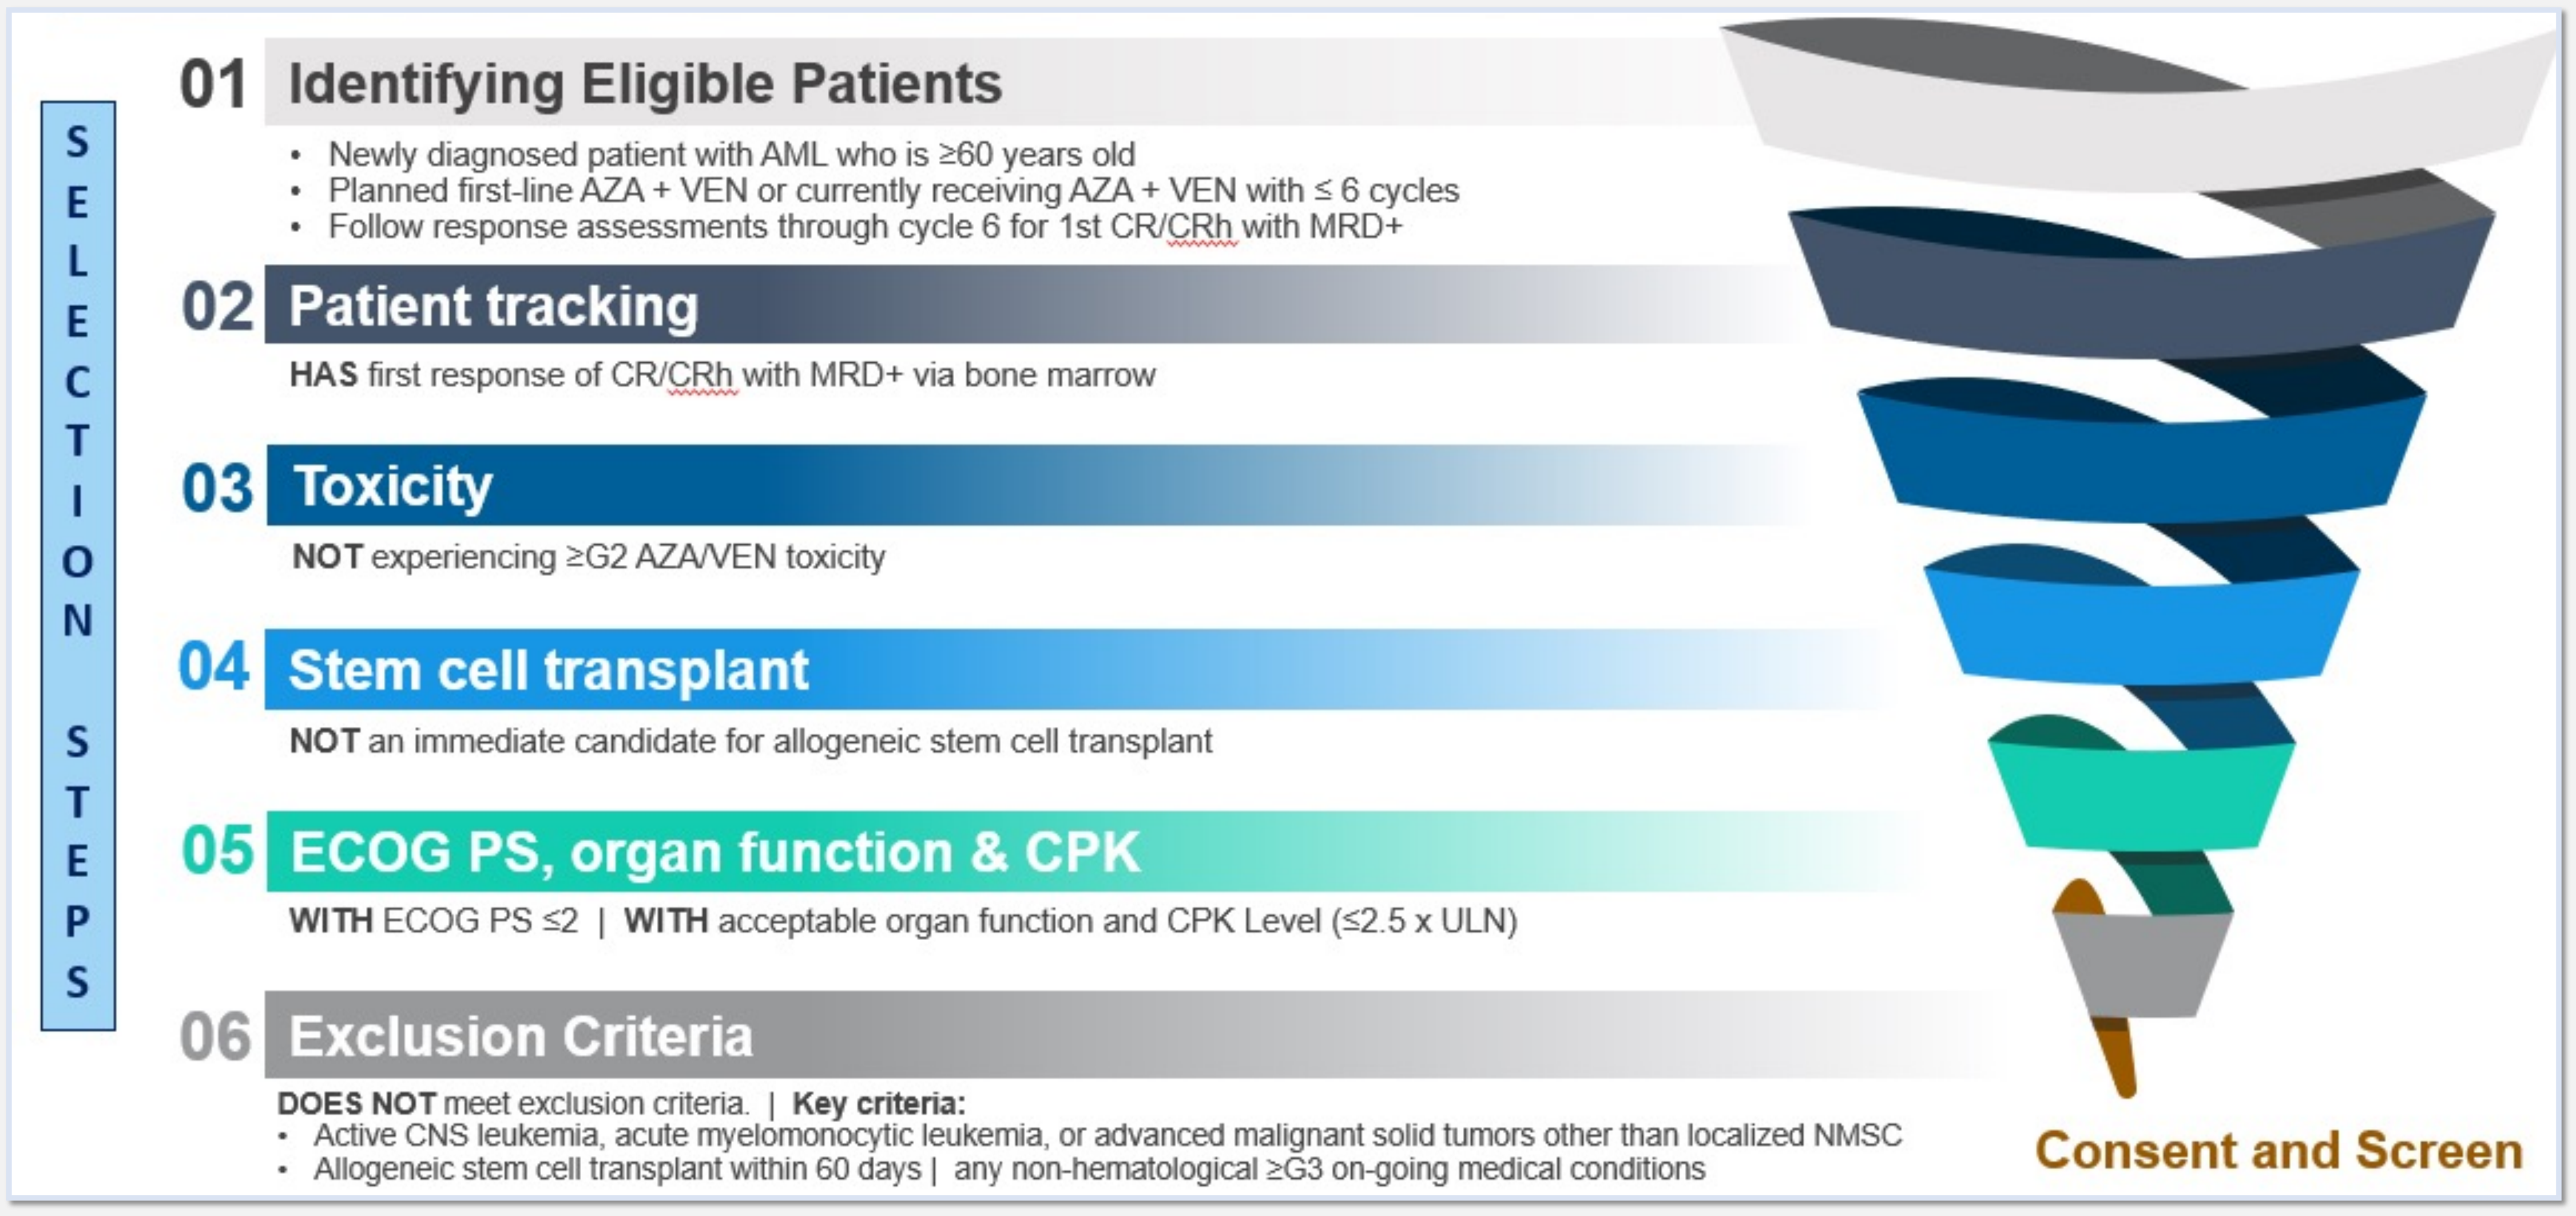
SELECTION STEPS
01 Identifying Eligible Patients
• Newly diagnosed patient with AML who is ≥60 years old
• Planned first-line AZA + VEN or currently receiving AZA + VEN with ≤ 6 cycles
• Follow response assessments through cycle 6 for 1st CR/CRh with MRD+
02 Patient tracking
HAS first response of CR/CRh with MRD+ via bone marrow
03 Toxicity
NOT experiencing ≥G2 AZA/VEN toxicity
04 Stem cell transplant
NOT an immediate candidate for allogeneic stem cell transplant
05 ECOG PS, organ function & CPK
WITH ECOG PS ≤2 | WITH acceptable organ function and CPK Level (≤2.5 x ULN)
06 Exclusion Criteria
DOES NOT meet exclusion criteria. | Key criteria:
• Active CNS leukemia, acute myelomonocytic leukemia, or advanced malignant solid tumors other than localized NMSC
• Allogeneic stem cell transplant within 60 days | any non-hematological ≥G3 on-going medical conditions
Consent and Screen

BIOANALYTICAL AND BIOMARKER PLAN

Pharmacokinetics	• Patient safety and drug clearance
Genomic analysis	• Genetic, epigenetic and mutational analysis of bone marrow and peripheral blood • MRD analysis by next generation sequencing (exploratory objective)
Transcriptomic analysis	• RNA-sequencing analysis of bone marrow and peripheral blood
Proteomic analysis	• MRD analysis by flow cytometry (secondary objective) • Cytokine/chemokine/growth factor quantification
Machine learning analysis	• Correlational analysis of biomarker and clinical data to determine predictive biomarkers of response.

SUMMARY

- Emavusertib monotherapy appears to be well tolerated in patients with relapsed or refractory AML, regardless of mutation status, previously treated with HMA, venetoclax, and FLT3 inhibitors.
- Emavusertib in combination with azacitidine and venetoclax demonstrated synergistic anti-leukemic effects in AML cell lines.
- Adding emavusertib to the azacitidine/venetoclax doublet in MRD+ patients at the time of CR may enable patients to achieve MRD negativity without significant toxicity.
- This triplet combination has a potential to become new regimen in front-line therapy for AML patients, regardless of mutation status.
- Exploratory biomarkers will be analyzed to determine predictive biomarkers of response.
- The study is currently enrolling patients.

REFERENCES

1. Metzeler K.H. et al., Spectrum and prognostic relevance of driver gene mutations in acute myeloid leukemia. Blood 2016; 128 (5): 686–698.
2. Pratz K.W. et al., Measurable Residual Disease Response and Prognosis in Treatment-Naïve Acute Myeloid Leukemia With Venetoclax and Azacitidine. JCO 40, 855-865(2022).
3. DiNardo C.D. et al., Molecular patterns of response and treatment failure after frontline venetoclax combinations in older patients with AML. Blood. 2020 Mar 12;135(11):791-803.
4. DiNardo C.D. et al., Azacitidine and Venetoclax in Previously Untreated Acute Myeloid Leukemia. N Engl J Med. 2020 Aug 13;383(7):617-629.
5. Short N.J. et al., Azacitidine, Venetoclax, and Gilteritinib in Newly Diagnosed and Relapsed or Refractory FLT3-Mutated AML. JCO 0, JCO.23.01911
6. Ugolkov et al., 2021. IRAK4 inhibitor CA-4948 potentiates antitumor effects of azacitidine and Venetoclax in human acute myeloid leukemia. EHA 325144; EP390.
7. Li Z. et al., Inhibition of IRAK1/4 sensitizes T cell acute lymphoblastic leukemia to chemotherapies. J Clin Invest. 2015 Mar 2;125(3):1081-97.
8. Hertlein E.K. et al. NF-κB Is Active in Mcl-1 Promoter Regulation in Human CLL.. Blood 2008; 112 (11): 2265.

CONTACT INFORMATION

Reinhard von Roemeling, MD
Senior VP, Clinical Development
rvonroemeling@curis.com
Curis Inc.
128 Spring Street
Lexington
MA 02421
www.curis.com