DEVELOPMENT OF POTENTIAL BIOMARKERS FOR IRAK4 INHIBITOR EMAVUSERTIB IN HUMAN ACUTE MYELOID LEUKEMIA

A. UGOLKOV[1], M. PILICHOWSKA[2], C. LI[1], R. HOK[1], M. SAMSON[1], M. LANE[1], R. VON ROEMELING[1], R. MARTELL[1] / [1]Curis, Inc., Lexington, MA, USA | [2]Tufts Medical Center, Boston, MA, USA

INTRODUCTION

Acute myeloid leukemia (AML), the second most common form of leukemia in adults, remains a highly fatal disease. Interleukin-1 receptor-associated kinase 4 (IRAK4) has been demonstrated as a potential therapeutic target in human AML. IRAK4-mediated activation of NF-κB signaling pathway could play a critical role in NF-κB-regulated survival and chemoresistance of cancer cells (Fig. 1). The results of ongoing Phase 1 study demonstrated clinical activity of IRAK4 inhibitor emavusertib (CA-4948) in patients with relapsed/refractory AML and high-risk MDS (hrMDS) [1]. Our preliminary results demonstrated that mutations of splicing factor genes U2AF1 and SF3B1 are associated with a clinical response to emavusertib therapy in hrMDS/AML patients [1]. Because mutations of splicing factor genes are rarely detected in de novo AML cases [2], we developed an immunohistochemical (IHC) assay and explored expression of potential biomarkers in bone marrow (BM) samples with the goal to support the development of a companion diagnostic for emavusertib.

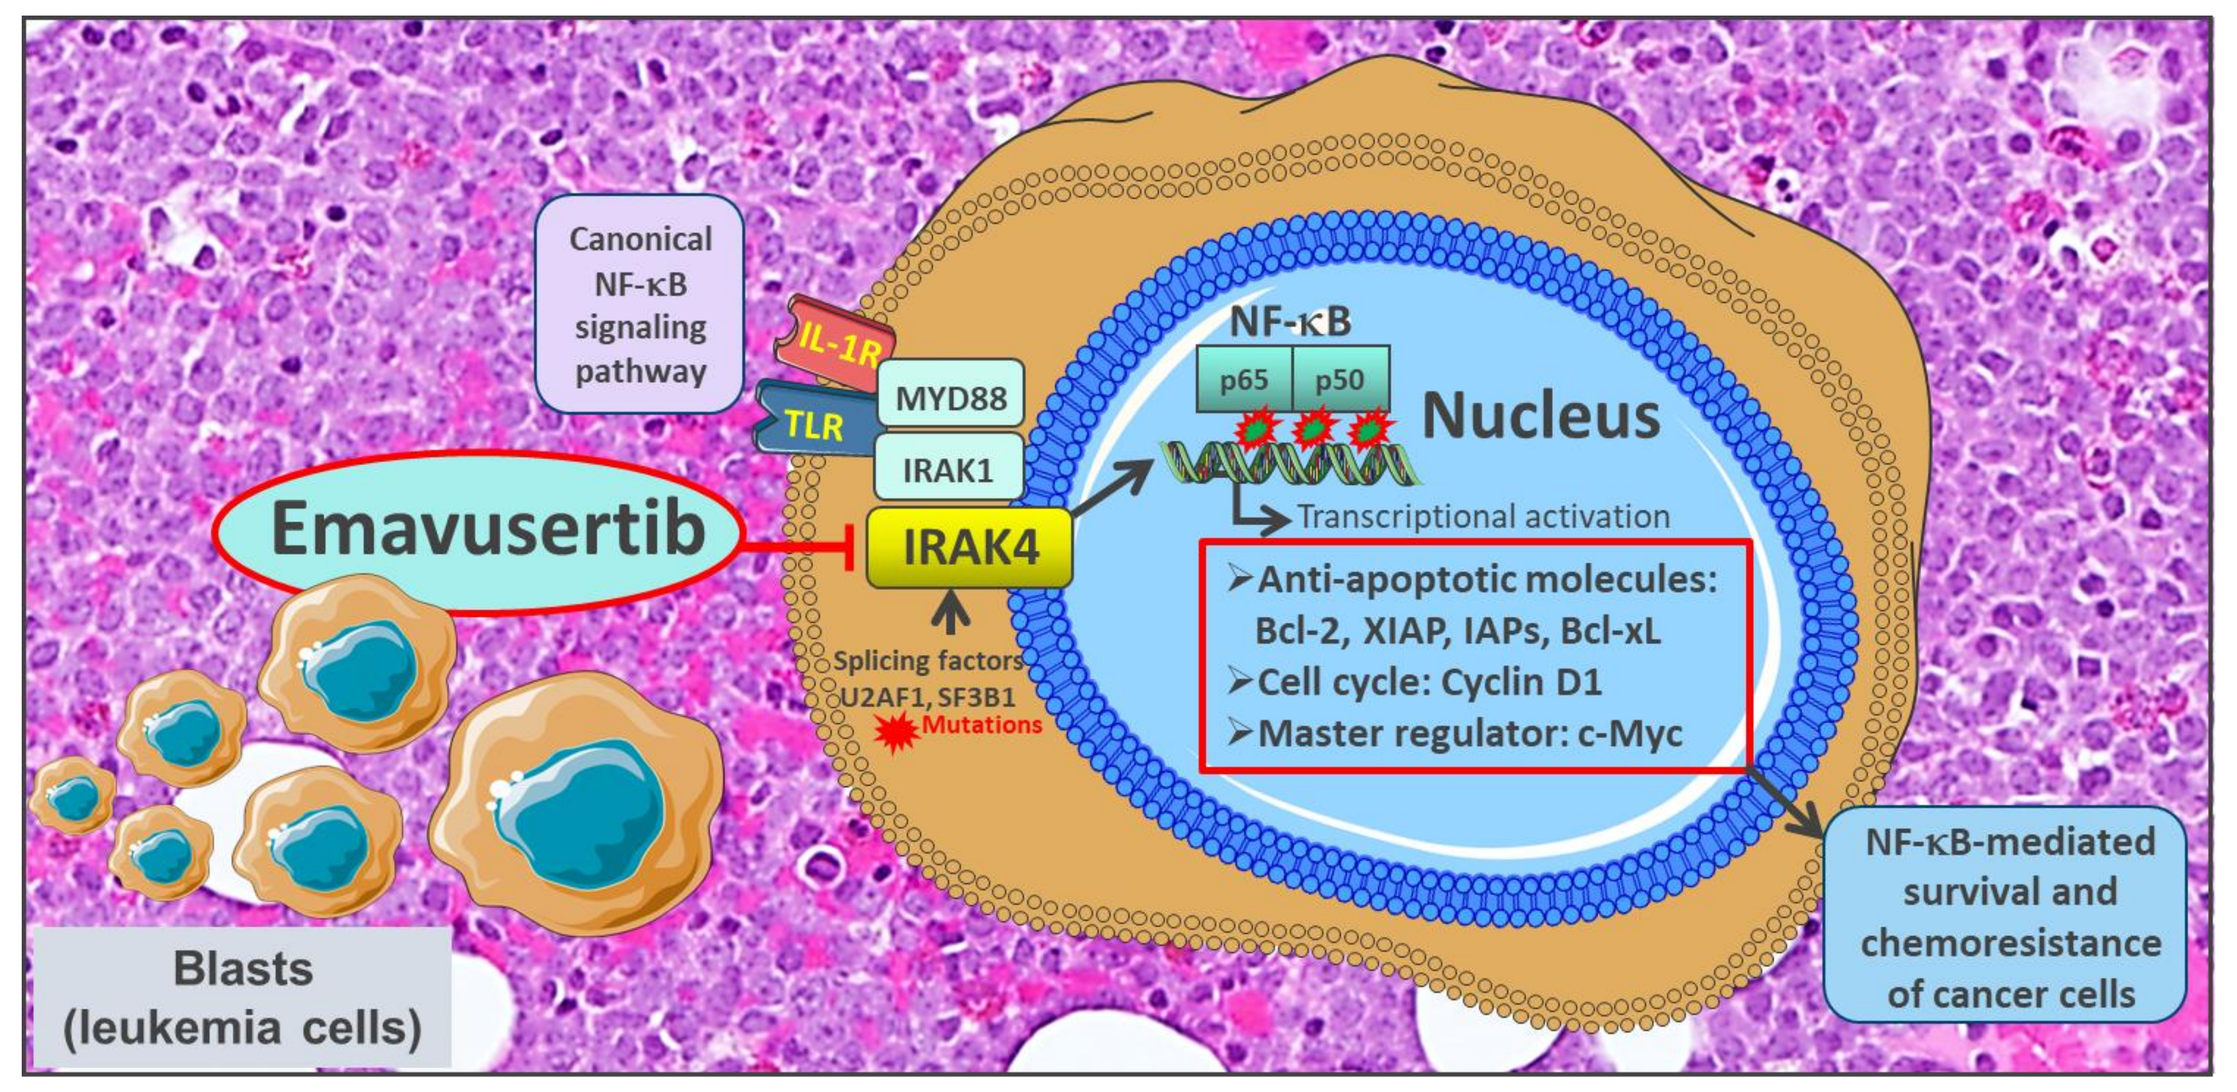
Canonical NF-κB signaling pathway
IL-1R
TLR
MYD88
IRAK1
IRAK4
Emavusertib
NF-κB
p65
p50
Nucleus
Transcriptional activation
➢Anti-apoptotic molecules: Bcl-2, XIAP, IAPs, Bcl-xL
➢Cell cycle: Cyclin D1
➢Master regulator: c-Myc
Splicing factors U2AF1, SF3B1
Mutations
NF-κB-mediated survival and chemoresistance of cancer cells
Blasts (leukemia cells)

Figure 1. *Inhibition of IRAK4 by emavusertib (CA-4948) could lead to suppression of NF-κB-mediated survival, and chemoresistance of malignant cells in human AML. IRAK4 is known as a cytoplasmic protein playing a critical role in formation of myddosome complex and initiation of signal transduction pathways.*

Clip art images reproduced under the terms of the Creative Commons Attribution 3.0 Unported license (CC BY 3.0), https://creativecommons.org/licenses/by/3.0/ from Smart Servier Medical Art, https://smart.servier.com/

OBJECTIVE

The objective of the present study was to analyze expression of IRAK4, NF-κB p-p50 and NF-κB p-p65 by immunohistochemical staining in human AML with further evaluation of these molecules as potential biomarkers for emavusertib therapy in AML patients.

METHODS

We used IHC staining and immunoblotting to determine the expression of IRAK4 and NF-κB proteins in human leukemia cell lines and clinical AML samples. THP-1, HL-60 and K-562 nuclear cell extracts were purchased from Santa Cruz Biotechnology. Nuclear extracts were separated by SDS-PAGE, transferred to nitrocellulose membrane and probed with IRAK4, p-IRAK4 T345/S346, NF-κB p50, NF-κB p65 or Histone H3 antibodies followed by incubation with fluorophore-conjugated secondary antibody. Double immunostaining was performed using DAB (brown), green or black chromogens. CD34/CD117 staining was used as a marker of blasts. Exploratory biomarker evaluations were performed using serial sections of formalin-fixed paraffin-embedded (FFPE) BM clot samples obtained from 19 AML patients. Eight BM AML samples were purchased from Analytical Biological Services and 11 BM samples were obtained at screening from evaluable patients with relapsed/refractory AML enrolled in CA-4948-102 clinical trial (NCT #04278768).

RESULTS

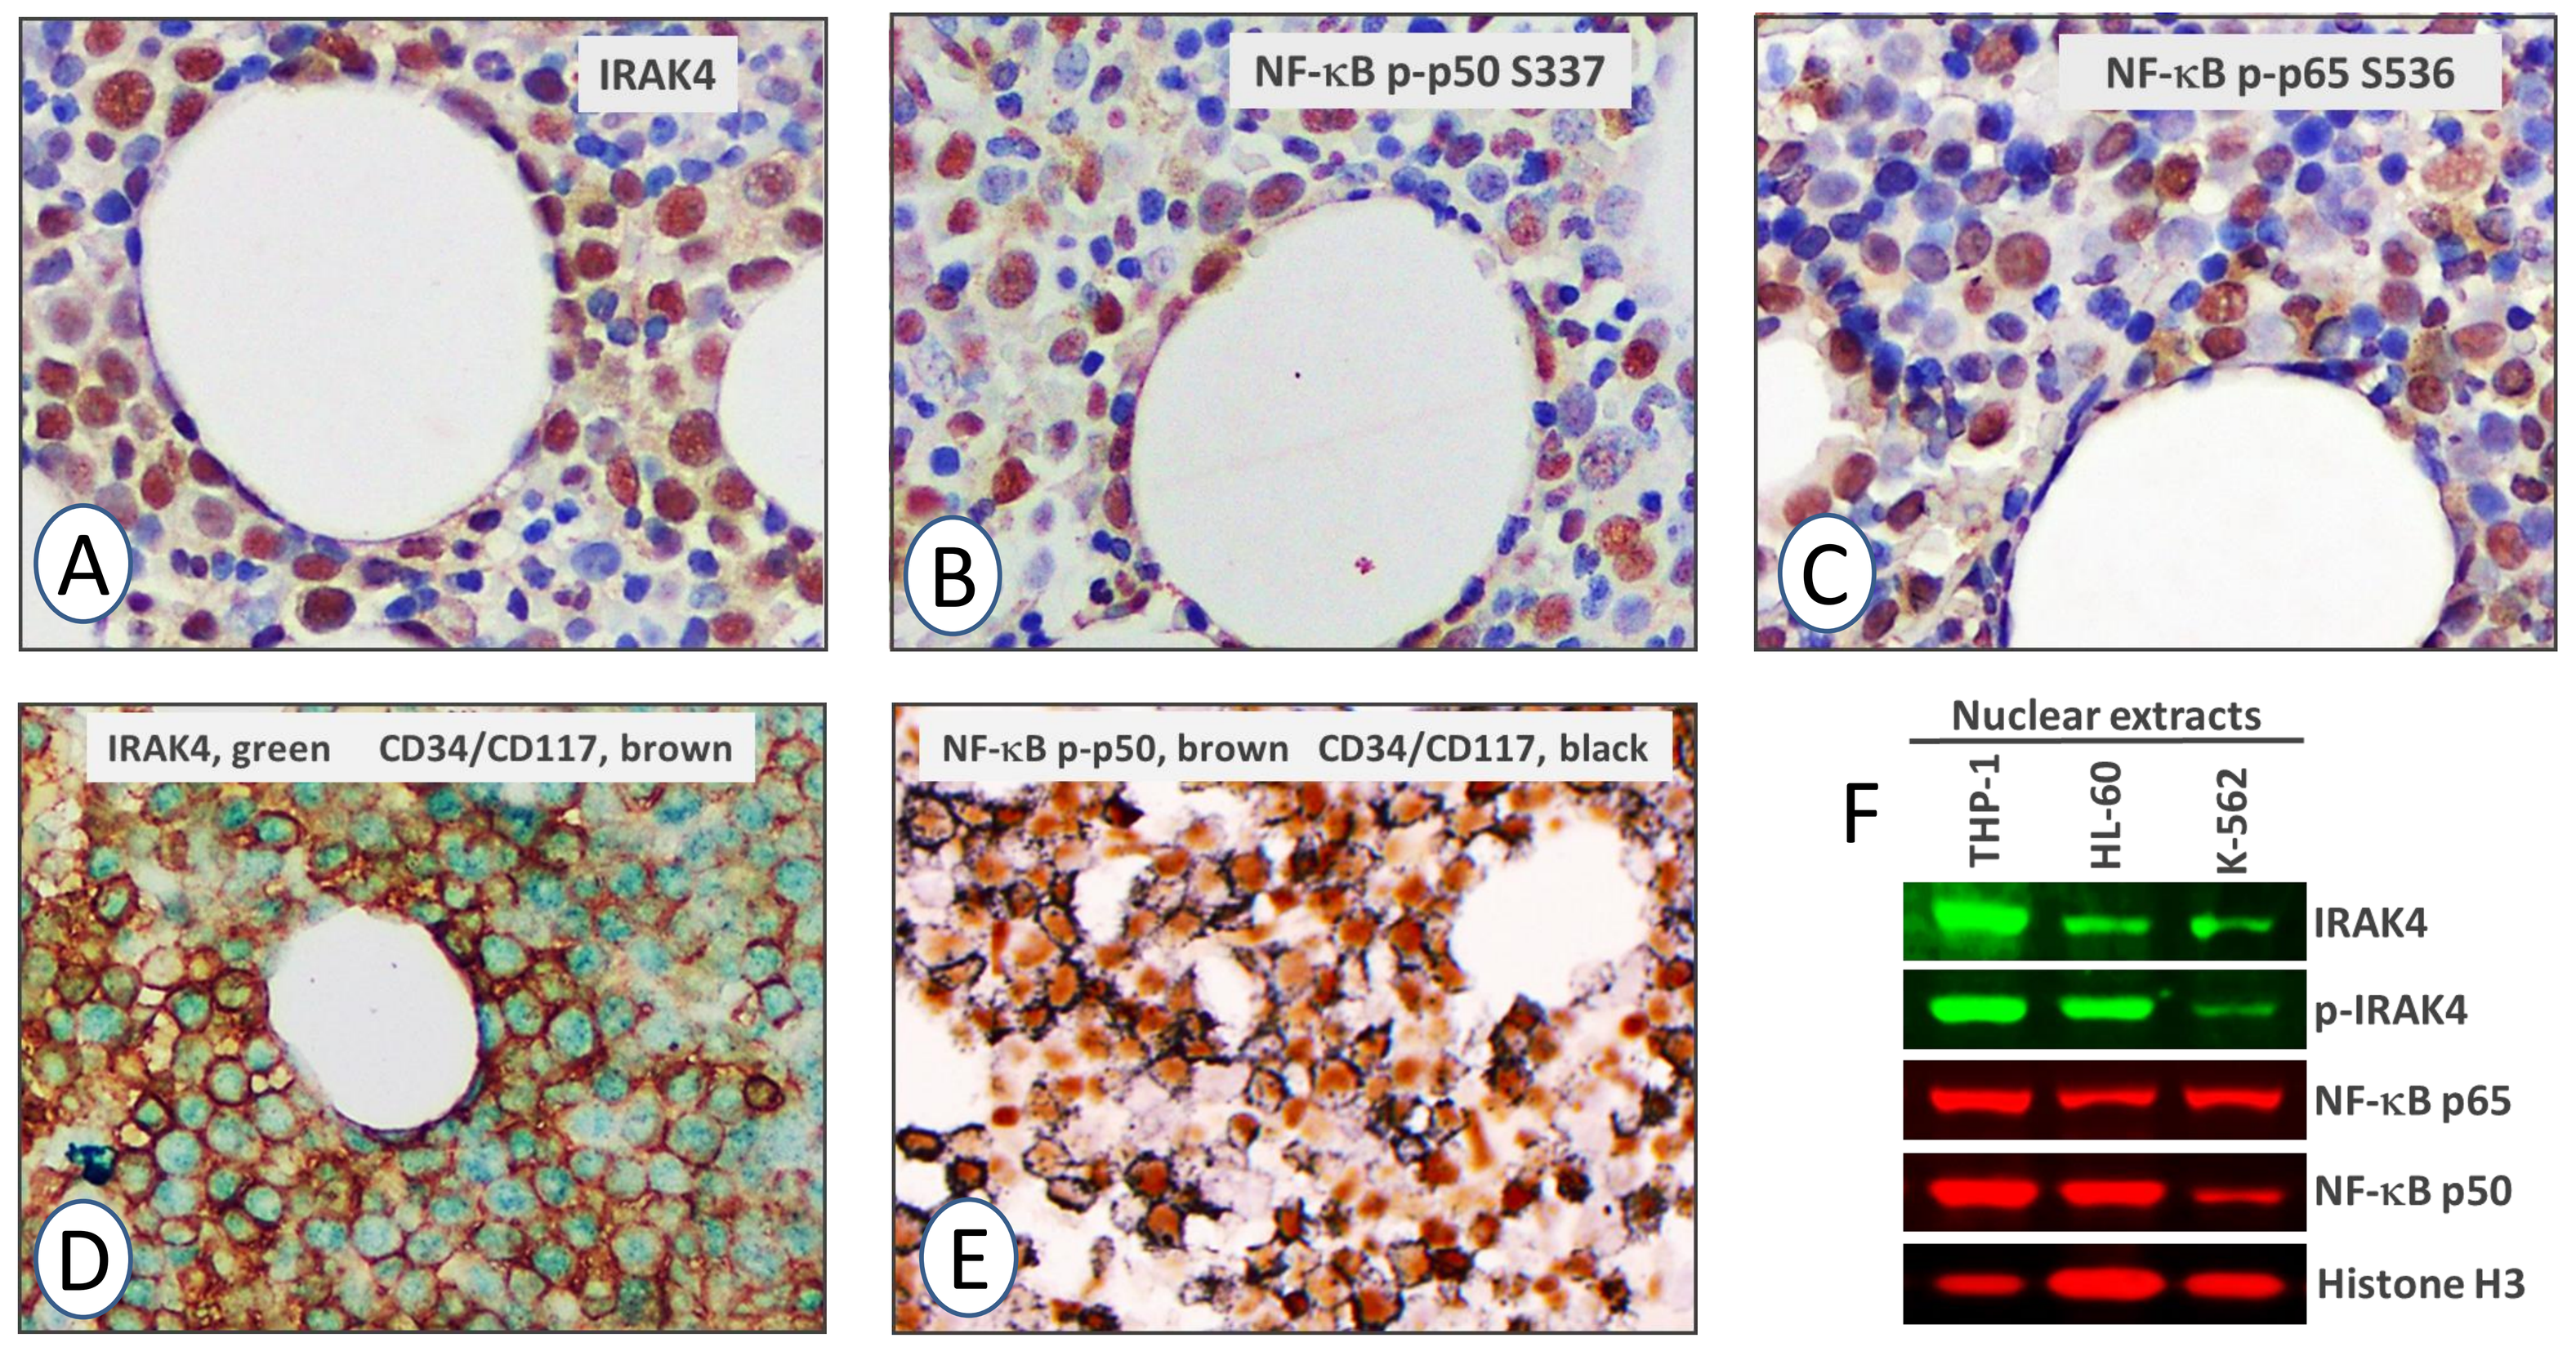
IRAK4
A
NF-κB p-p50 S337
B
NF-κB p-p65 S536
C
IRAK4, green CD34/CD117, brown
D
NF-κB p-p50, brown CD34/CD117, black
E
F
Nuclear extracts
THP-1
HL-60
K-562
IRAK4
p-IRAK4
NF-κB p65
NF-κB p50
Histone H3

Figure 2. *Representative images of IHC staining of AML bone marrow clots. Nuclear expression of IRAK4 (A), NF-κB p-p50 S337 (B) and NF-κB p-p65 S536 (C) was found in blasts and immature myeloid cells in serial sections of AML bone marrow clot sample 1b (triple-positive case). (D) Double immunostaining shows nuclear expression of IRAK4 (green) in CD34/CD117-positive blasts (membranous staining, brown) in AML bone marrow clot sample 4b. (E) Double immunostaining shows nuclear expression of NF-κB p-p50 (brown) in CD34/CD117-positive blasts (membranous staining, black) in AML bone marrow clot sample 4b. (F) Expression of IRAK4, p-IRAK4 T345/S346 (active form of IRAK4), NF-κB p65 and NF-κB p50 proteins was analyzed in nuclear extracts prepared from THP-1, HL-60 and K-562 leukemia cell lines by Western immunoblotting.*

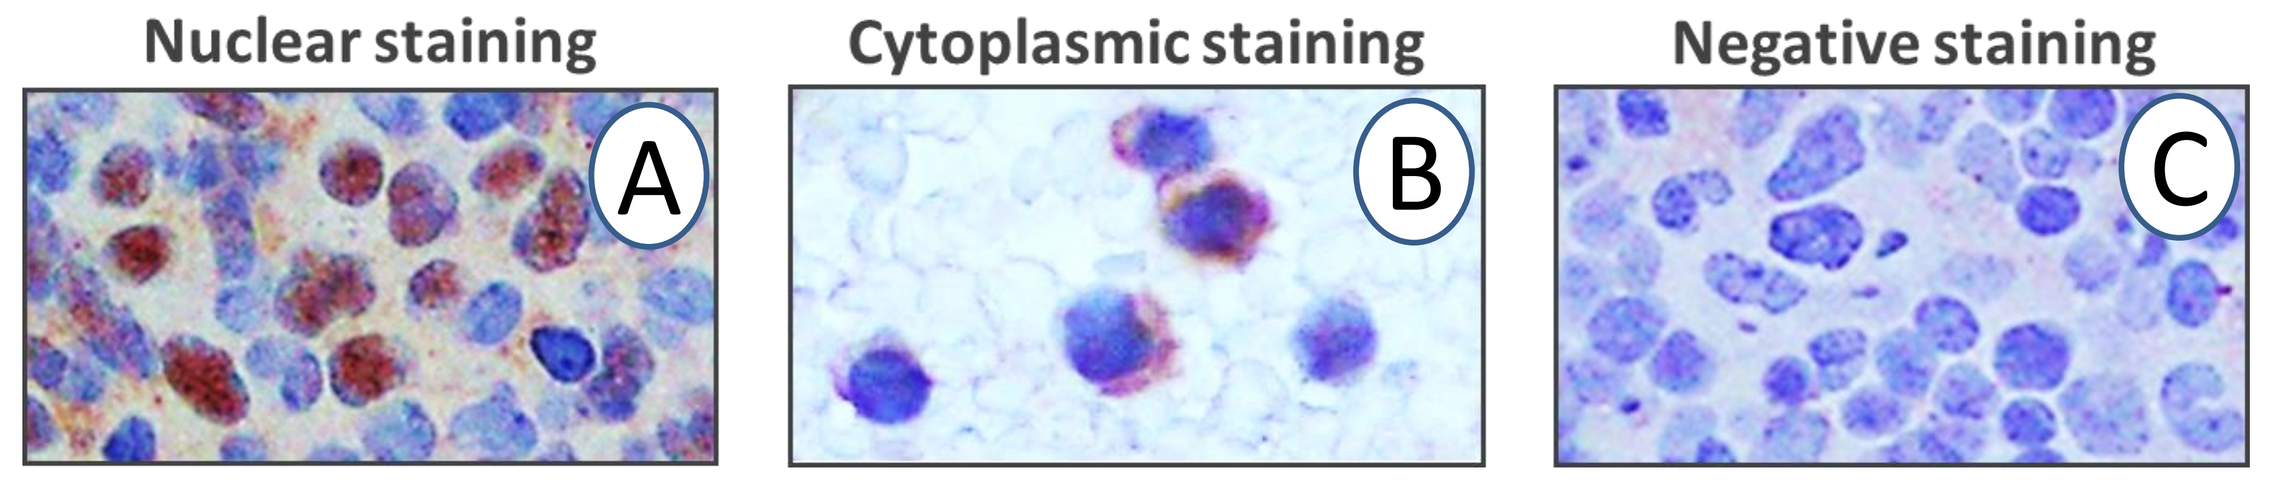
Nuclear staining
A
Cytoplasmic staining
B
Negative staining
C

Figure 3. *Representative images of IRAK4 nuclear (A), cytoplasmic (B) and negative (C) staining of blasts in bone marrow clot samples obtained at screening from AML patients (3 cases).*

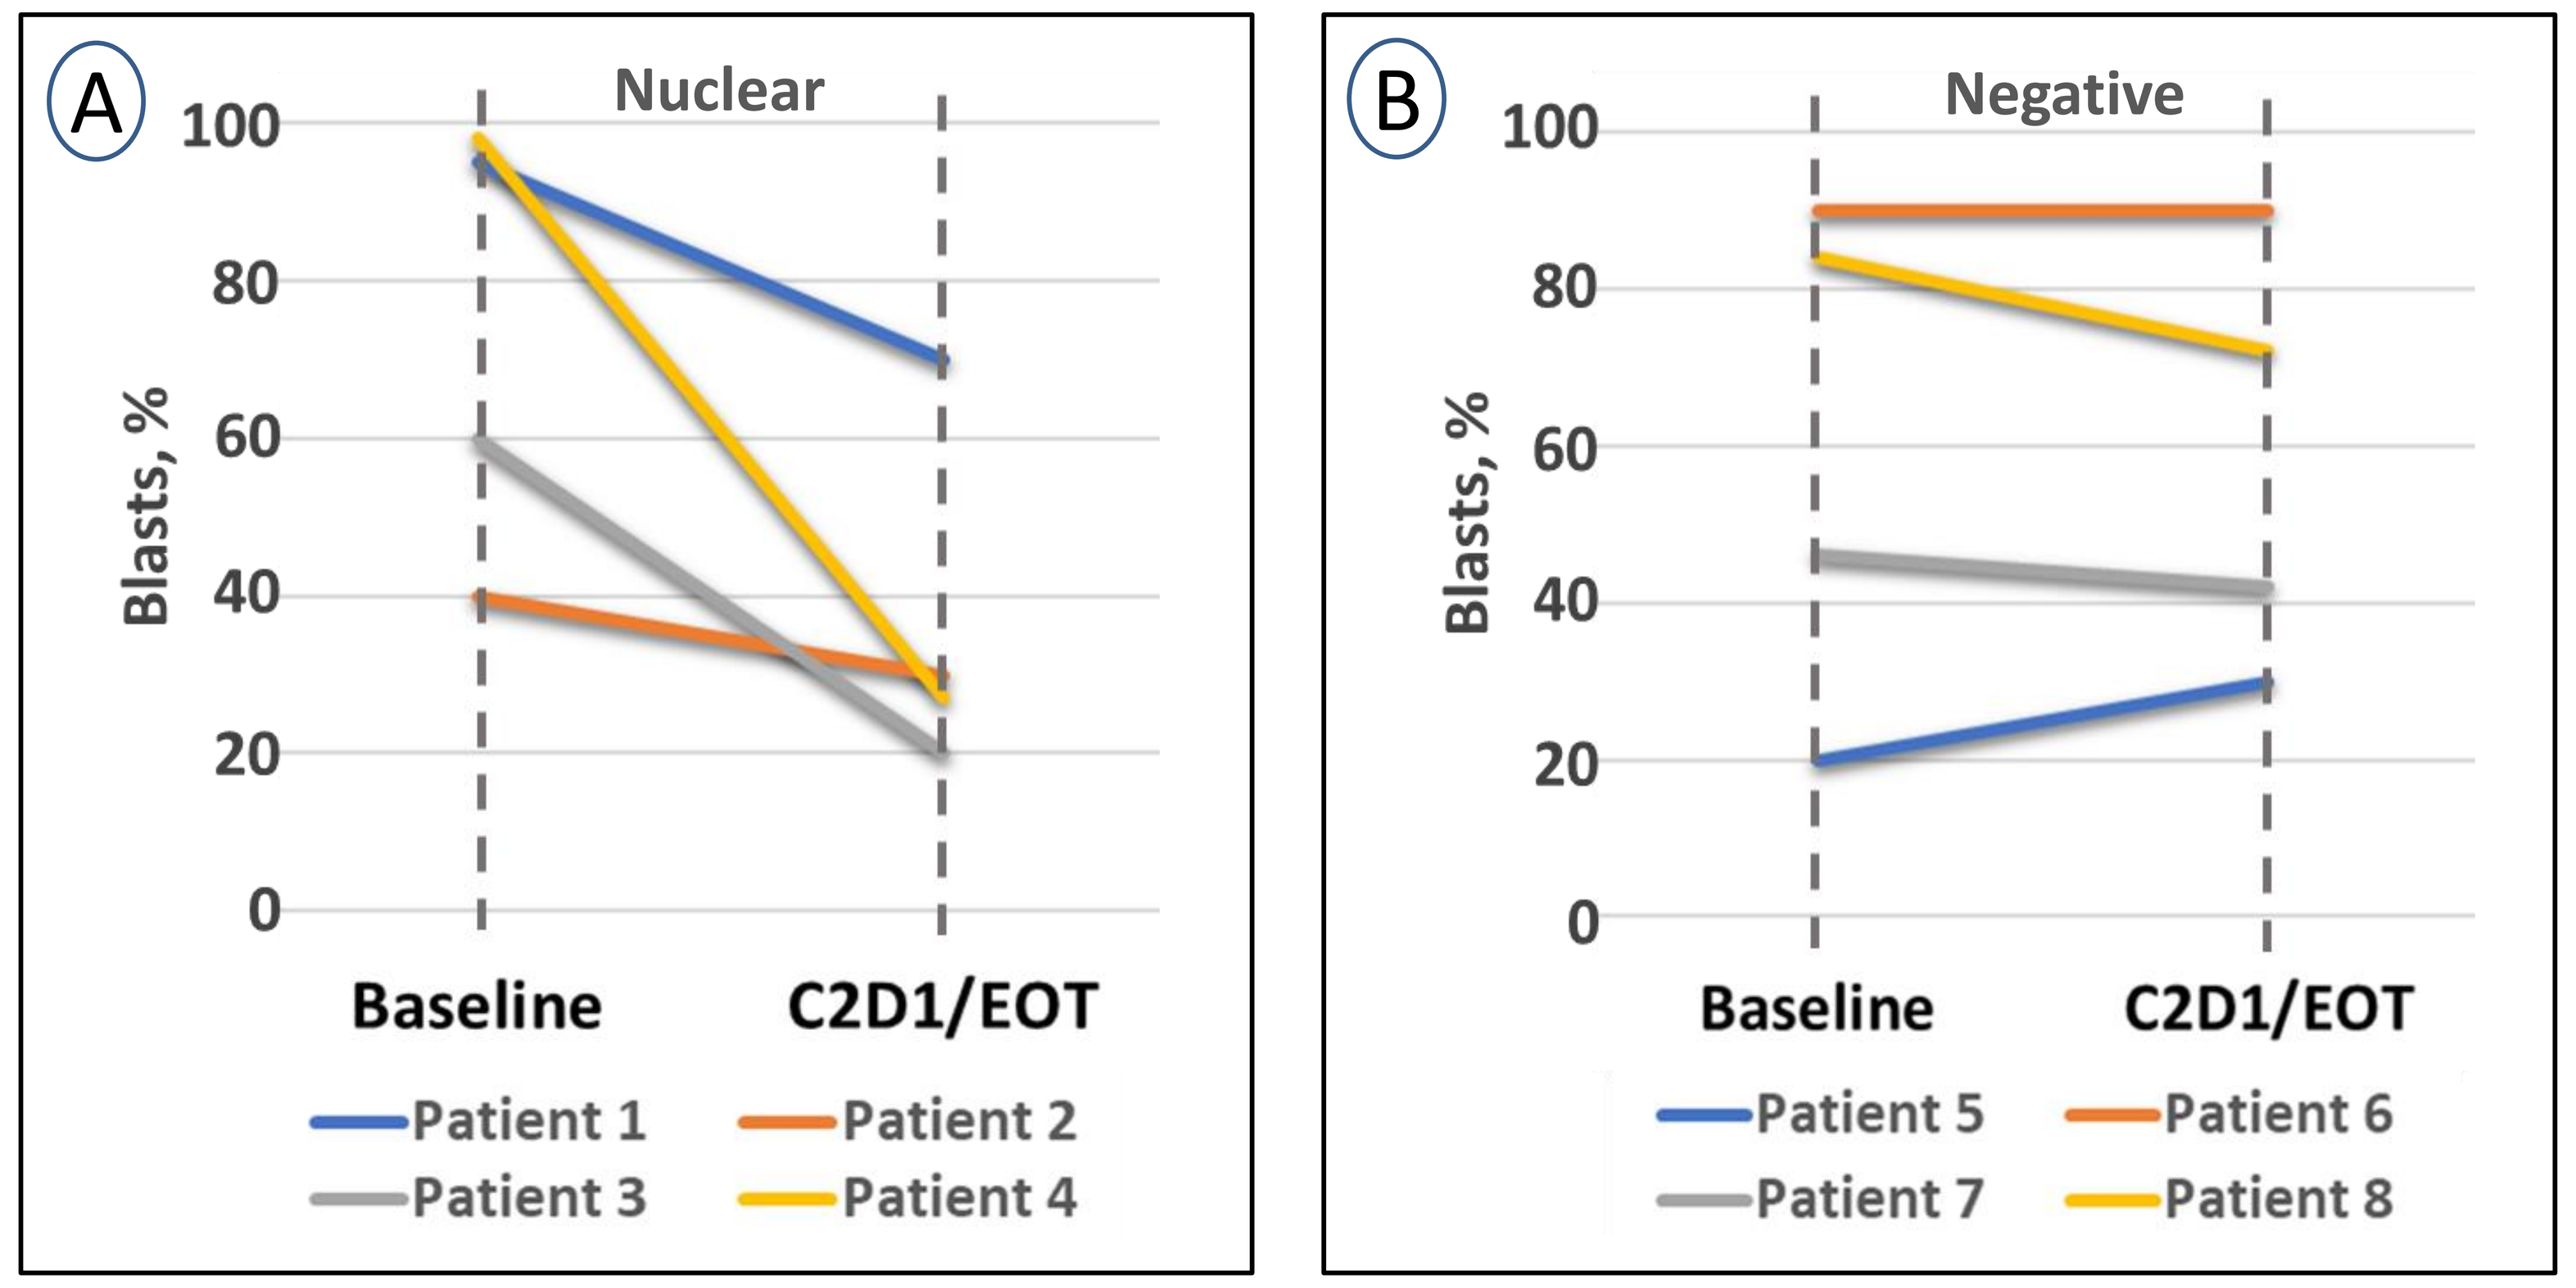
A
Nuclear
Blasts, %
100
80
60
40
20
0
Baseline
C2D1/EOT
Patient 1
Patient 2
Patient 3
Patient 4
B
Negative
Blasts, %
100
80
60
40
20
0
Baseline
C2D1/EOT
Patient 5
Patient 6
Patient 7
Patient 8

Figure 4. *Treatment with emavusertib (clinical trial NCT #04278768) significantly reduced bone marrow blast count in AML patients with triple-positive nuclear staining (IRAK4, NF-κB p-p50 and NF-κB p-p65) as compared to the baseline blast count (A). (B) Triple-negative staining (IRAK4, NF-κB p-p50 and NF-κB p-p65) was observed in bone marrow samples obtained at screening from four AML patients (clinical trial NCT #04278768). C2D1, cycle 2, day1. EOT, end of treatment*

RESULTS

Using IHC staining, we found IRAK4 nuclear expression selectively in blasts and immature myeloid cells in 9/19 AML cases (Fig 2). To the best of our knowledge, this is the first report showing nuclear accumulation of IRAK4 in cancer cells. Using AML BM samples, we found that IRAK4 nuclear expression in blasts was significantly correlated with activation of NF-κB as determined by nuclear accumulation of NF-κB p-p50 and p-p65 (triple-positive nuclear staining) in 9/19 cases (Fig. 2A-E). In support of our findings in clinical AML samples, we detected IRAK4, p-IRAK4, NF-κB p50 and p65 protein expression in nuclear lysates prepared from leukemia cell lines THP-1, HL-60 and K562 (Fig. 2F). Our findings of p-IRAK4 T345/S346 nuclear expression in leukemia cell lines suggest that active form of IRAK4 can accumulate in the nuclei of leukemia cells (Fig. 2F).

Triple-positive nuclear (4 cases), cytoplasmic (3 cases) and negative (4 cases) staining was detected in blasts in bone marrow samples obtained at screening from 11 emavusertib-treated AML patients (Fig. 3). Although the cohort size is small, our preliminary results demonstrate that treatment with emavusertib led to a significant decrease in bone marrow blast count in nuclear triple-positive (IRAK4, NF-κB p-p50 and p-p65) de novo AML cases including case #4 (a decrease in blast count from 98% to 27%) and case #3 (a decrease in blast count from 60% to 20%), whereas no significant decrease in bone marrow blast count was detected in triple-negative cases (Fig. 4).

CONCLUSIONS

Our findings uncovered a previously unknown nuclear expression of IRAK4 in leukemia cells. Our results demonstrated co-expression of nuclear IRAK4, NF-κB p-p50 and p-p65 in blasts suggesting a potential novel mode of interaction between IRAK4 and NF-κB in human AML. Although the role of nuclear IRAK4 in leukemia cells remains to be investigated, our preliminary findings revealed new perspectives into emavusertib treatment stratification and demonstrate the possibility of discovering novel biomarkers through IHC analysis of clinical samples. These may be particularly useful in identifying patients with non-spliceosome gene mutated disease who may benefit from emavusertib.

REFERENCES

1. G. Garcia-Manero, S. Tarantolo, A. Verma, et al. A Phase 1, dose escalation trial with novel oral IRAK4 inhibitor CA-4948 in patients with acute myelogenous leukemia or myelodysplastic syndrome. 2021 EHA Virtual Congress, abstract S165.
2. R. Lindsley, B. Mar, E. Mazzola, et al. Acute myeloid leukemia ontogeny is defined by distinct somatic mutations. Blood 2015;125(9):1367-1376.

CONTACT INFORMATION

Curis Contact:
Andrey Ugolkov, MD, PhD
Senior Director
Translational Science
augolkov@curis.com

Curis, Inc.
128 Spring Str
Building C – Suite 500
Lexington, MA 02421
www.curis.com

EHA2022

HYBRID JUNE 9-17 VIENNA