Poster #
385

Identification of NF-κB phospho-p50 as a potential predictive biomarker for IRAK4 inhibitor CA-4948 in patients with Non-Hodgkin's lymphoma

Ugolkov A, Samson M, Hok R, von Roemeling R, Martell R
Curis Inc., Lexington, MA

Background

IRAK4, a serine/threonine kinase, was identified as a therapeutic target in hematological malignancies (1). CA-4948, a first-in-class small molecule inhibitor of IRAK4, has been tested in Phase 1 clinical trial with evidence of clinical activity in Non-Hodgkin's lymphoma (NHL) and acute myelogenous leukemia (AML) patients. To support the development of a predictive biomarker assay for CA-4948, we developed an immunohistochemical (IHC) assay and explored expression of potential biomarkers of response to CA-4948 in tumor biopsy samples obtained from NHL patients. IRAK4-mediated activation of NF-κB could play an important role in NF-κB-regulated survival and chemoresistance of cancer cells (1). It has been shown that phosphorylation of NF-κB p50 at Serine 337 is critical for DNA binding and transcriptional activity of NF-κB (2). Our results demonstrate that expression of NF-κB p-p50 S337 in cancer cells could be a potential predictive and pharmacodynamic marker for CA-4948 in human NHL.

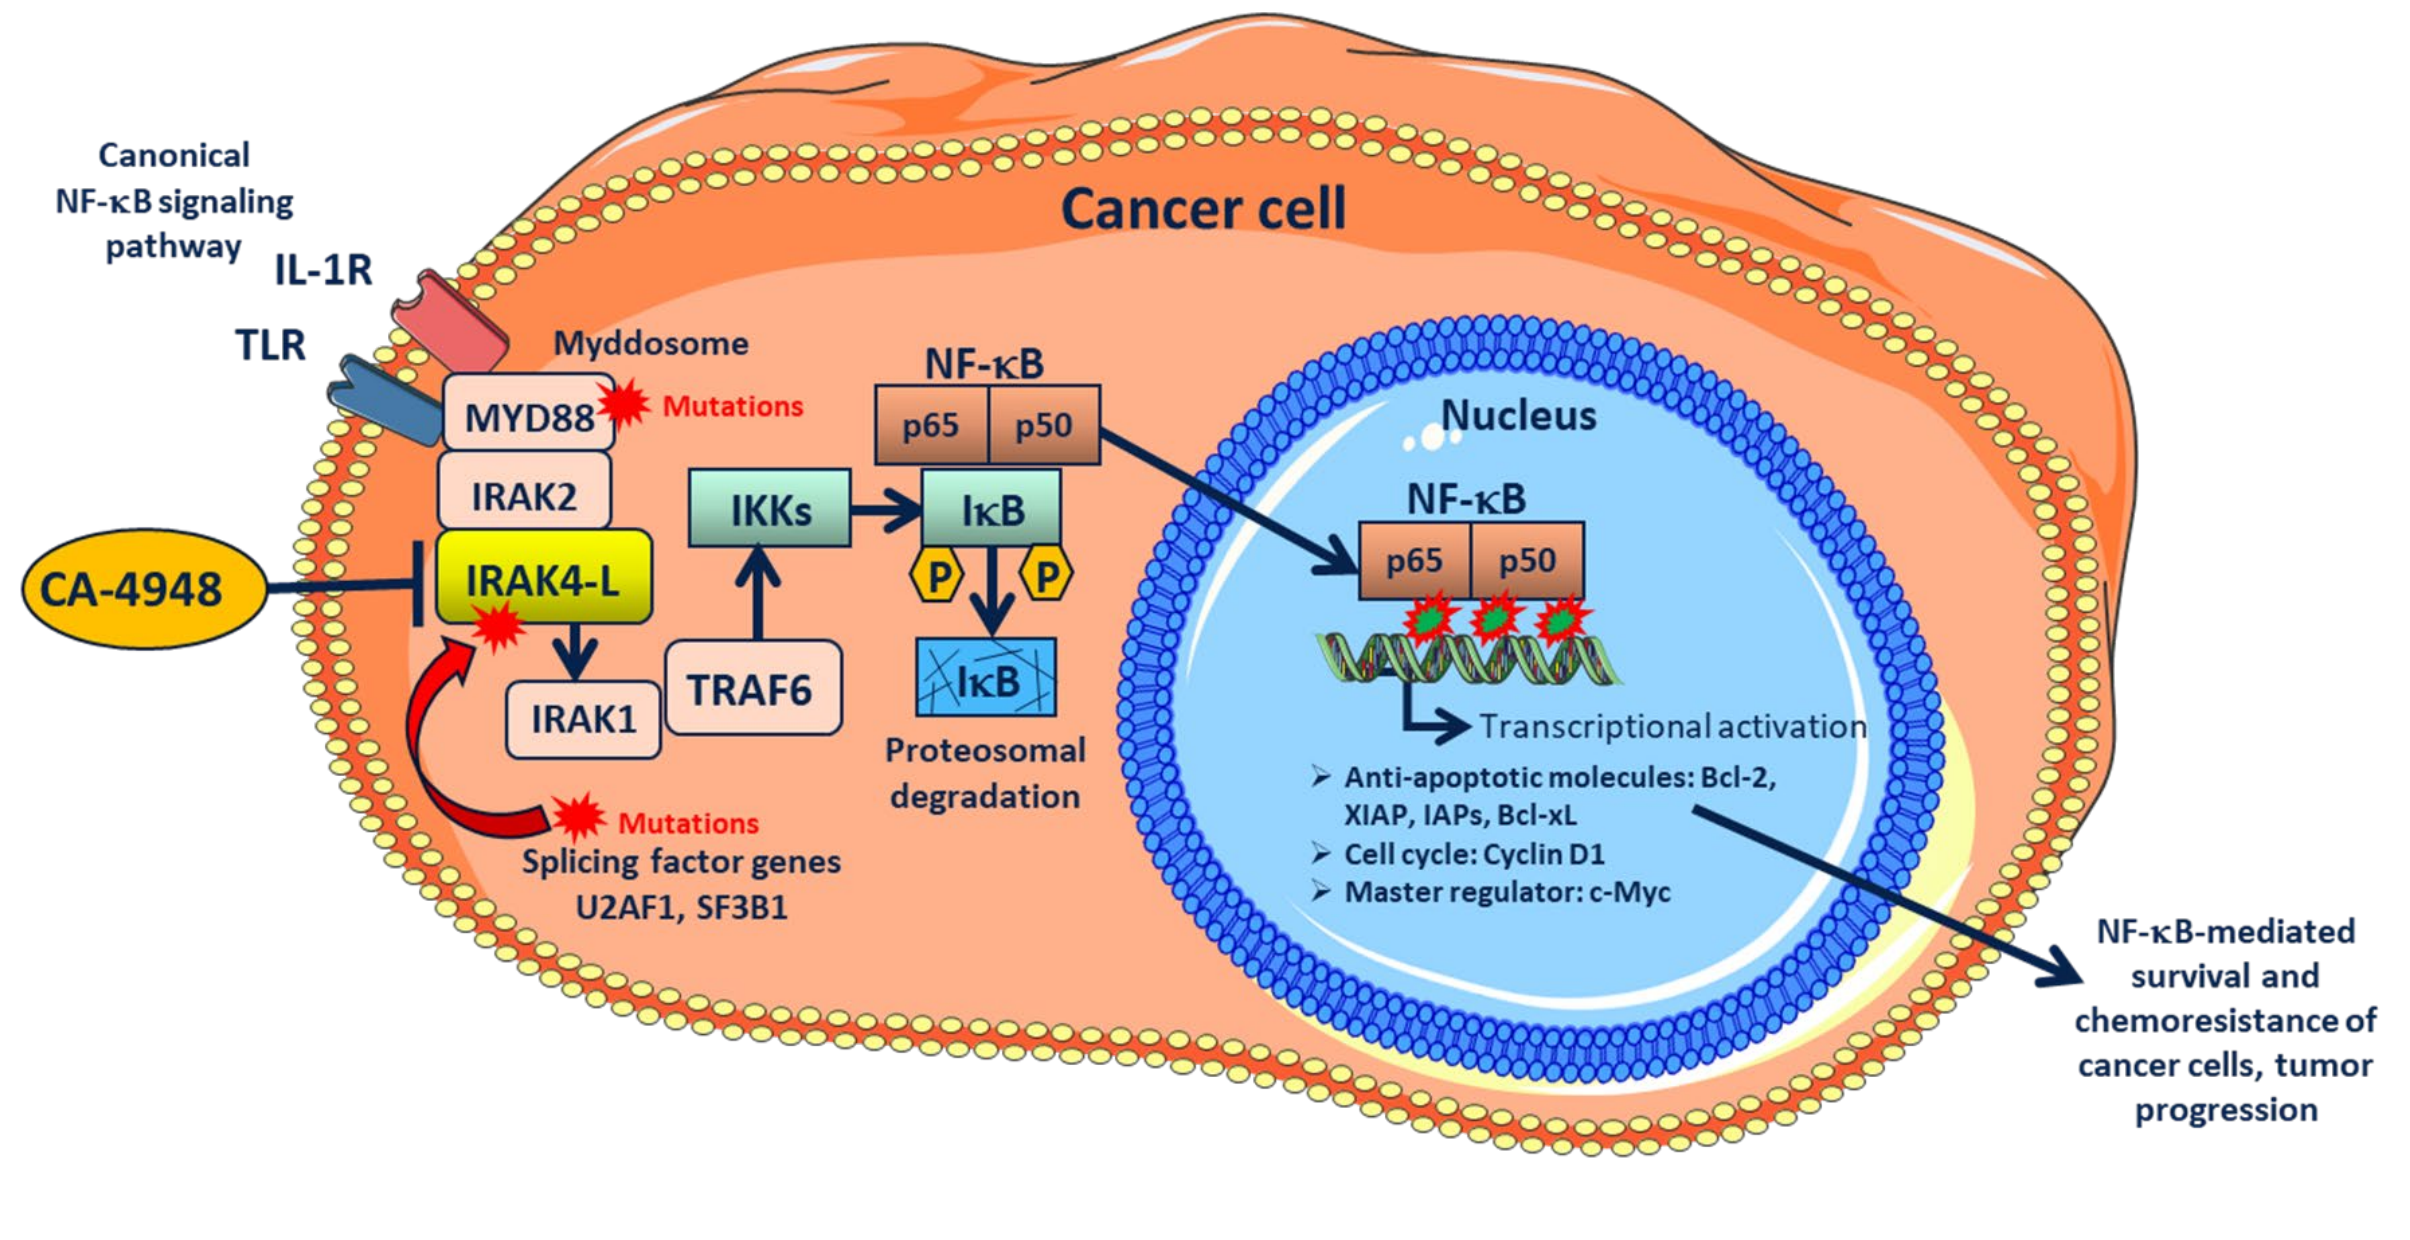
Canonical NF-κB signaling pathway
IL-1R
TLR
Cancer cell
Myddosome
MYD88
Mutations
IRAK2
CA-4948
IRAK4-L
IRAK1
TRAF6
IKKs
NF-κB
p65
p50
IκB
P
P
IκB
Proteosomal degradation
Mutations
Splicing factor genes
U2AF1, SF3B1
Nucleus
NF-κB
p65
p50
Transcriptional activation
Anti-apoptotic molecules: Bcl-2, XIAP, IAPs, Bcl-xL
Cell cycle: Cyclin D1
Master regulator: c-Myc
NF-κB-mediated survival and chemoresistance of cancer cells, tumor progression

Materials and Methods

Immunohistochemical assay has been developed and used for analysis of NF-κB phospho-p50 S337 expression in tumor biopsy samples obtained from 14 patients with NHL before CA-4948 treatment. Patients were defined according to their clinical response to CA-4948 treatment: stable disease (SD), 7 cases and progressive disease (PD), 7 cases. Positive staining was defined as nuclear and/or cytoplasmic expression of NF-κB p-p50 S337 in more than 50% of cancer cells throughout the tumor specimen.

Results

We found nuclear and/or cytoplasmic expression of NF-κB p-p50 in 6 of 7 SD cases (Fig.1, 2) treated with 50 mg QD (3 cases, 2 cases of tumor regression), 50 mg BID (1 case), 200 mg BID (1 case, tumor regression) and 400 mg BID (2 cases). Expression of NF-κB p-p50 was not detected in 6 of 7 cases with PD (Fig. 1, 2) including patients treated with 50 mg QD (1 case), 100 mg QD (1 case), 100 mg BID (3 cases), 200 mg BID (1 case) and 400 mg BID (1 case). We found statistically significant correlation between expression of NF-κB p-p50 in tumor biopsy and SD in NHL patients treated with CA-4948 ($p<0.05$). Analysis of NF-κB p-p50 expression in paired tumor biopsy samples (3 cases) collected before and after the treatment with CA-4948 revealed a significant downregulation of NF-κB p-p50 expression in tumors obtained from CA-4948-treated NHL patients (Fig. 3). In support of our *in vivo* findings, our *in vitro* experiments demonstrated that expression of NF-κB p-p50 was depleted in 3D lymphoma organoids treated with a clinically relevant concentration of CA-4948 (Fig. 4). Our results support further development of NF-κB p-p50 as a potential predictive and pharmacodynamic biomarker of IRAK4 inhibitor CA-4948.

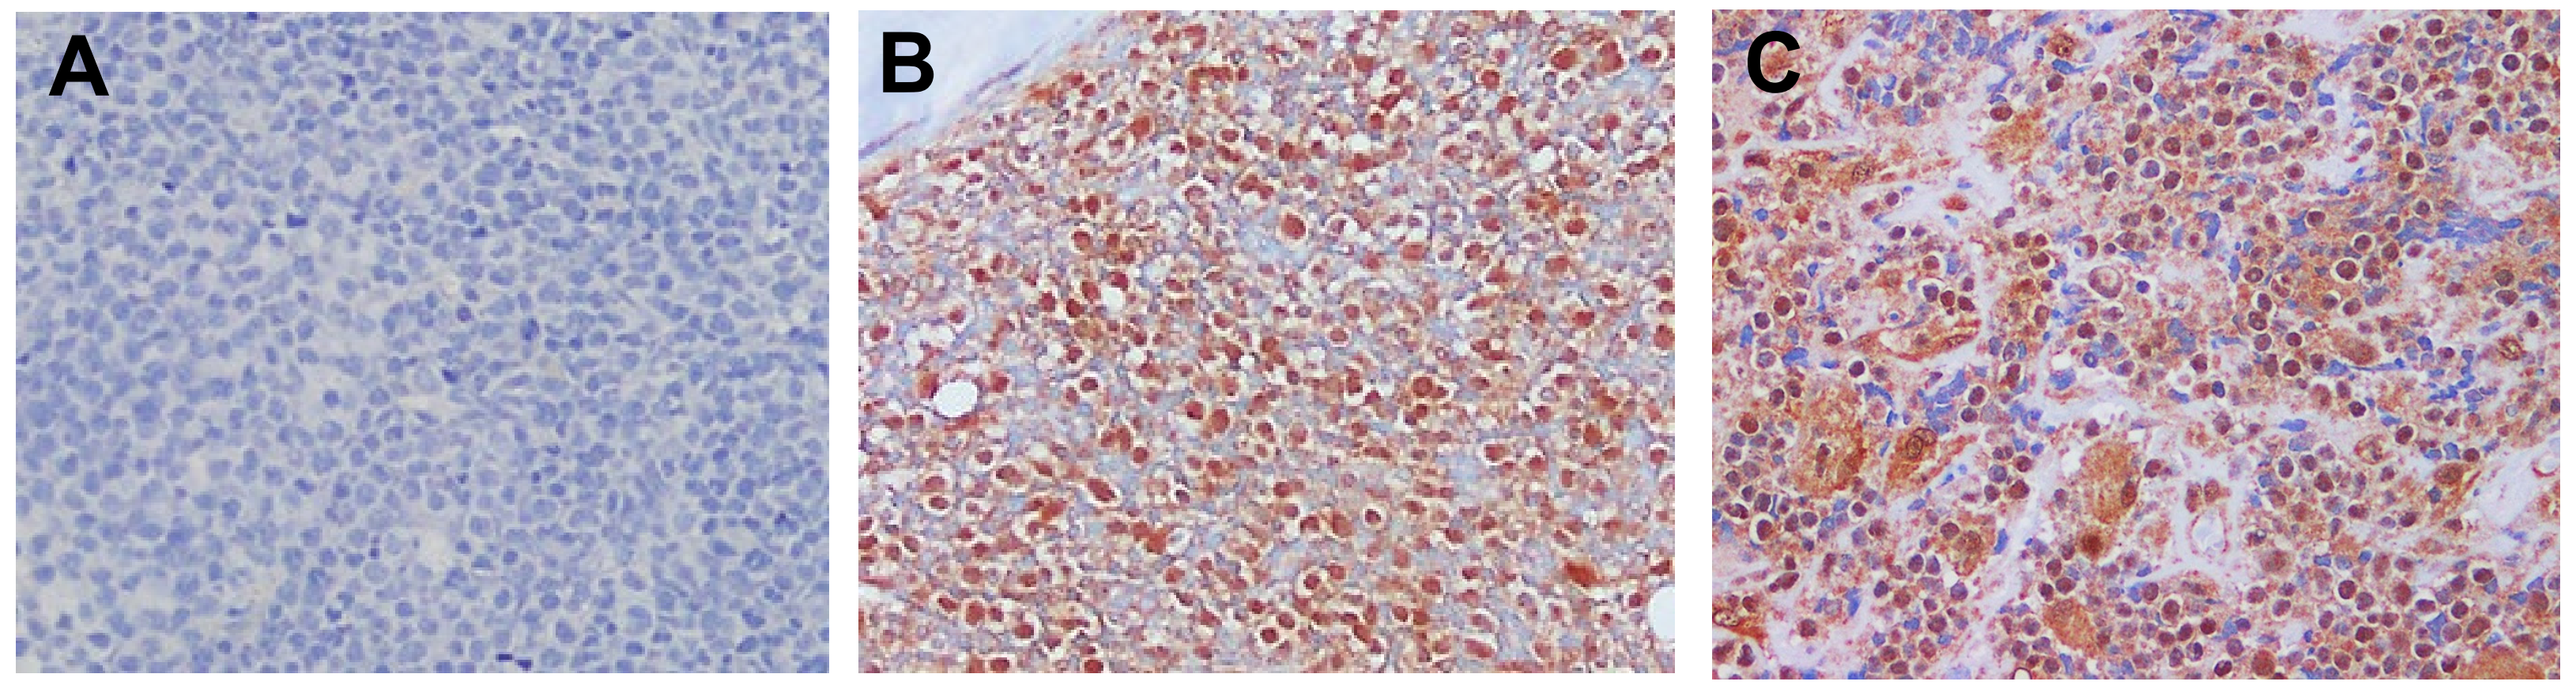
A
B
C

Figure 1. Representative pictures of NF-κB p-p50 S337 expression in NHL biopsy samples. A, negative staining. B and C, positive staining, nuclear/cytoplasmic staining of more than 50% of cancer cells.

NF-κB p-p50 S337

Negative staining: Patient	Negative staining: Best Response	Positive staining: Patient	Positive staining: Best Response
12-1002	+86% PD	19-1001	-35% SD
18-2004	+156% PD	02-1001	-24% SD
02-4004	+75% PD	02-3003	+22% SD
12-4004	+125% PD	12-5007	-35% SD
12-5006	+190% PD	02-6007	+25% SD
13-6001	+98% PD	02-6008	+16% SD
01-4002	+7% SD	15-1001	+65% PD

Figure 2. Positive staining of NF-κB p-p50 is correlated with stable disease in NHL patients treated with CA-4948. Response is shown as a percentage of tumor growth or regression. PD, progressive disease, SD, stable disease.

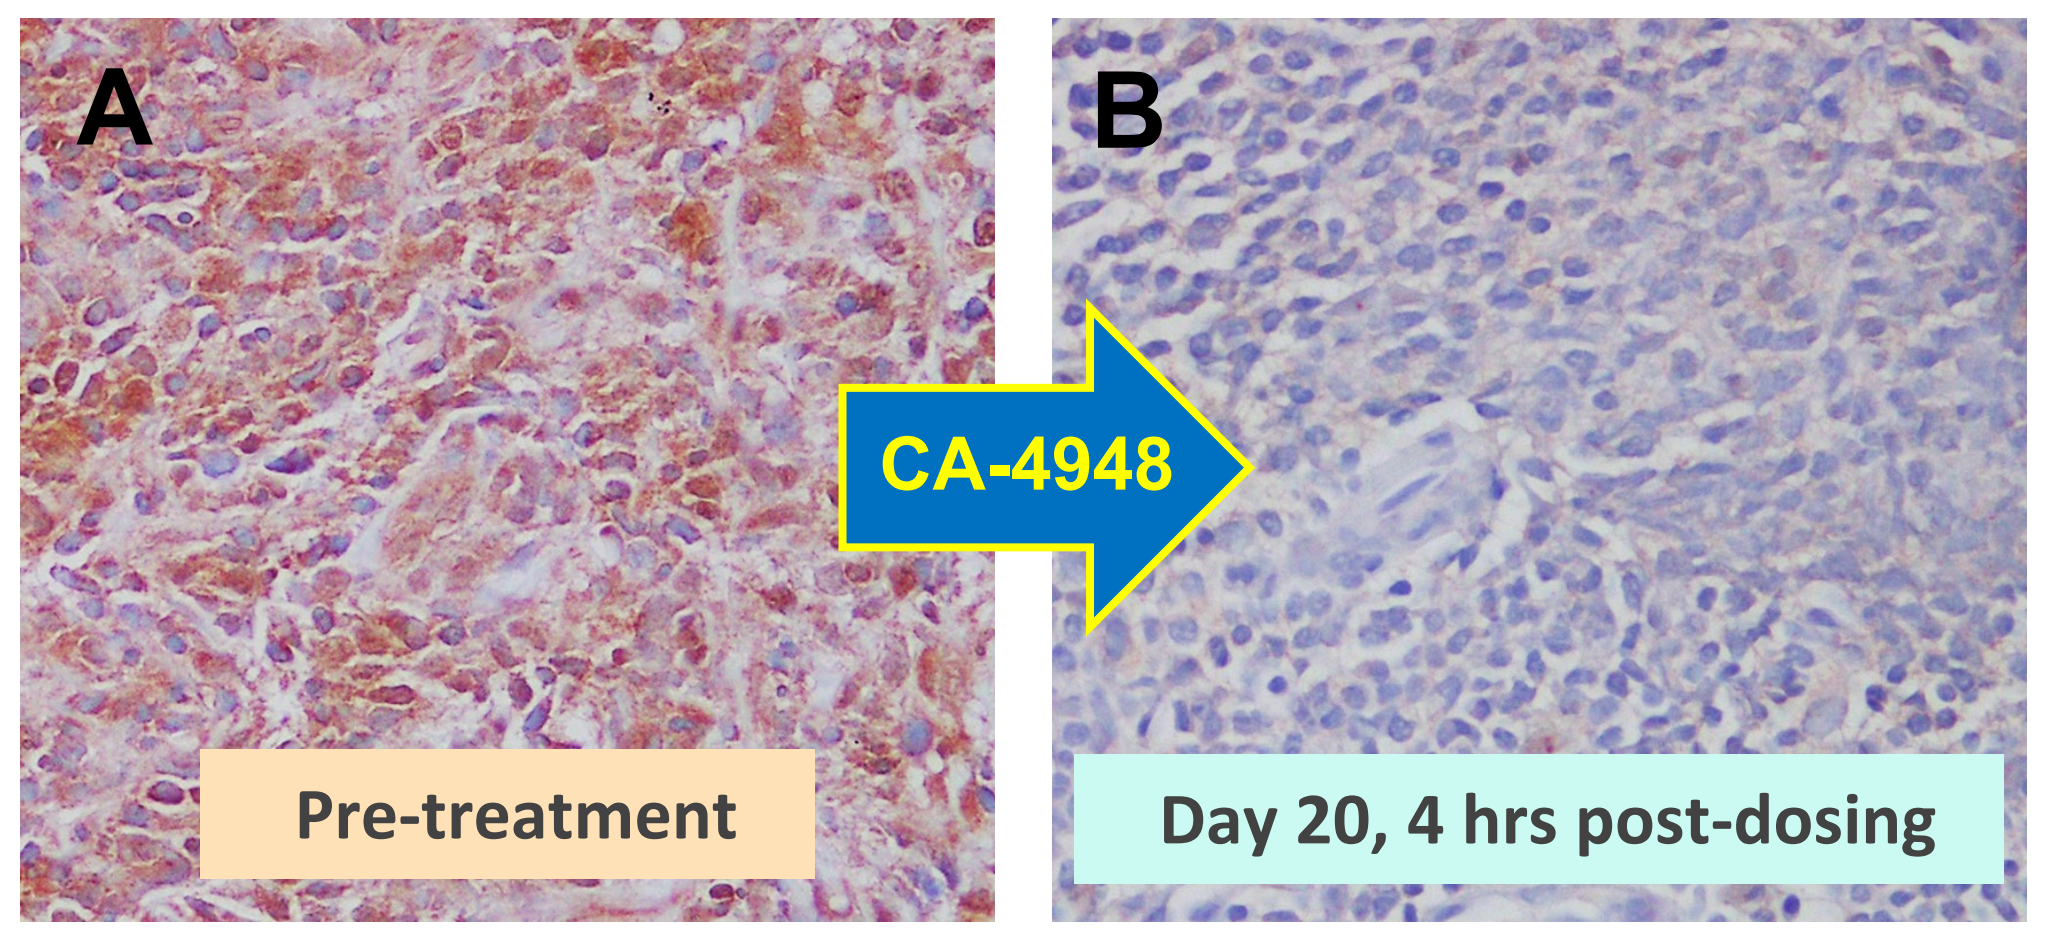
A
B
CA-4948
Pre-treatment
Day 20, 4 hrs post-dosing

Figure 3. Expression of NF-κB p-p50 is downregulated in tumor sample obtained from CA-4948-treated NHL patient. Representative pictures of NF-κB p-p50 S337 expression in lymphoma sample obtained from NHL patient (case 02-6007, stable disease) before (A) and after (B) the treatment with CA-4948.

Results

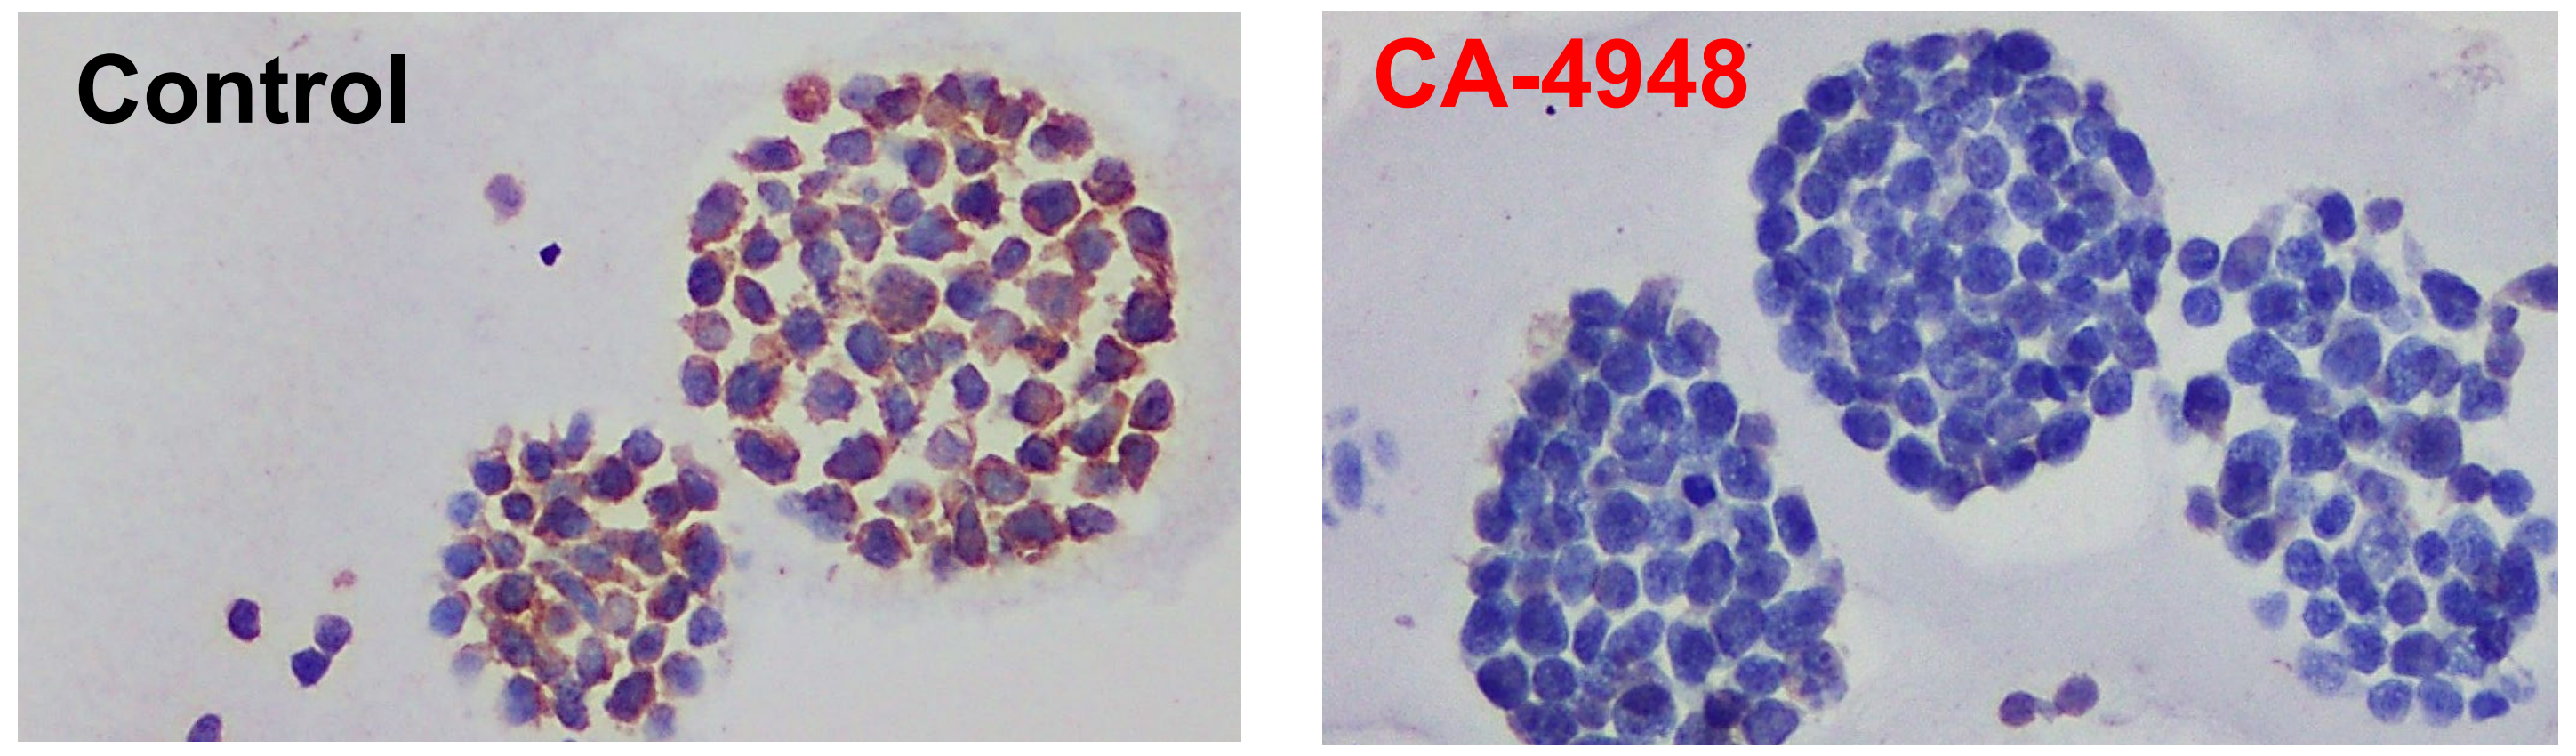
Control
CA-4948

Figure 4. CA-4948 downregulates expression of NF-κB p-p50 in OCI-LY19 NHL cell line. OCI-LY19 lymphoma cells were grown in Matrigel as 3D tumor organoids and treated with clinically relevant concentration of CA-4948 (10 μM) for 48 hours. Tumor organoids were fixed in 10% formalin at the end of the treatment and processed to FFPE sections. Expression of NF-κB p-p50 S337 was analyzed by IHC staining.

Conclusions

Although the cohort size is small, our findings suggest that expression of NF-κB p-p50 could serve as a potential biomarker to predict SD in response to the treatment with IRAK4 inhibitor CA-4948 in NHL patients whereas negative staining could be an exclusion marker for CA-4948 therapy. NF-κB p-p50 selection strategy might be used in future clinical trials to identify NHL patients which are most likely to respond to CA-4948 in combination with chemotherapy or targeted therapeutics.

References

1. Rhyasen GW, Starczynowski DT. IRAK signalling in cancer. Br J Cancer 2015;112(2):232-7.
2. Hou S, Guan H, Ricciardi RP. Phosphorylation of serine 337 of NF-kappaB p50 is critical for DNA binding. J Biol Chem 2003;278(46):45994-8.

Contact and Acknowledgements

We would like to thank the patients, their families and caregivers for their invaluable contribution and participation in this study.

Curis Contact:
Andrey Ugolkov, MD, PhD
Senior Director
Translational Science
augolkov@curis.com

Curis, Inc.
128 Spring Str
Bldg C – Suite 500
Lexington, MA 02421
www.curis.com