Phase 2 trial of CA-170, a novel oral small molecule dual inhibitor of immune checkpoints VISTA and PD-1, in patients with advanced solid tumor and Hodgkin lymphoma

Vivek S. Radhakrishnan[1], Sameer Bakhshi[2], Kumar Prabhash[3], Chetan Deshmukh[4], Shona Nag[5], KC Lakshmaiah[6], M Gopichand[7], Vikram Mathews[8], Murali Ramachandra[9], Divyesh Mandavia[9], Akhil Kumar[9], Hongwei Wang[10], Sudeep Gupta[11], Shripad D. Banavali[3]

[1]Tata Medical Center, Kolkata, India; [2]All India Institute of Medical Sciences, New Delhi, India; [3]Tata Memorial Hospital, Mumbai, India; [4]Deena Nath Mangeshkar Hospital, Pune, India; [5]Jehangir Clinical Development Center, Pune, India; [6]Srinivasam Cancer Care Multispecialty Hospitals, Bangalore, India; [7]HCG Curie Center of Oncology, Vijaywada, India; [8]Christian Medical College, Vellore, India; [9]Aurigene Discovery Technologies Limited, Bangalore, India; [10]Curis Inc. Boston, USA; [11]Advanced Centre for Treatment, Research & Education in Cancer, Mumbai, India.

Poster ID: P714

Introduction

V-domain Ig suppressor of T-cell activation (VISTA) and Programmed-death 1 (PD-1) are independent immune checkpoints implicated in various malignancies. Preclinical studies have demonstrated that dual blockade of these pathways is synergistic. CA-170 is a first-in-class oral small molecule that directly targets both VISTA and PD-1/PD-L1 pathways and has shown anti-tumor activity in multiple preclinical models. A Phase 1 dose escalation study (Clinicaltrials.gov NCT02812875)[1] has shown acceptable safety of CA-170 with dose escalated up to 1200 mg BID.

Methods

The Phase II study is a multi-tumor (Head & Neck Squamous Cell Cancer [HNSCC], Non-Small Cell Lung Cancer [NSCLC], MSI-H positive solid tumors and Classical Hodgkin Lymphoma [HL]) trial investigating two dosages (400 mg versus 800 mg) of CA-170. [Clinical Trials Registry of India (CTRI) registration no. - CTRI/2017/12/011026]

Key eligibility criteria include: age ≥ 18 years, ECOG ≤1, adequate organ function, no previous exposure to immuno-oncology agents, and 1-3 prior lines of systemic therapy. Primary endpoint is Response Rates by RECIST 1.1 for solid tumors and by IWG Revised Response Criteria for Classical Hodgkin Lymphoma[2,3]. Additional endpoints include response rates by immune-related Response Criteria (irRC)[4] for solid tumors, Clinical benefit Rate (CBR) - defined as Stable Disease or better, as well as safety and Pharmacokinetics. Analysis for Hodgkin Lymphoma was also done by Lugano criteria[5,6]. CA-170 was given once daily till progression of the disease or intolerable toxicities. Patients with solid tumors were followed with contrast enhanced CT scans and those with Hodgkin lymphoma with PET/CT.

Results

Enrolment started in February 2018, and as of Oct 15, 2018, a total of 62 patients have been enrolled. The tumor type distribution and baseline demographics of patients is provided in Tables 1 and 2.

Table 1: Tumor distribution

Tumor Type	No. of Patients
HNSCC	20
NSCLC	24
MSI-H positive solid tumors	5
Classical Hodgkin Lymphoma	13

Table 2: Baseline Demographics

Parameters	Values
Age, Median (range) years	53 (20-81)
Males, N (%)	48 (77)
ECOG status 1, N (%)	41 (66)
Median lines of previous therapy	2
Median duration of disease before enrollment (months)	14
Patients at 400 mg	30
Patients at 800 mg	32

Baseline demographics were similar in the 400 and 800 mg groups

Correspondence

*Dr. Divyesh Mandavia, MD.
Medical Lead – Clinical Development
Aurigene Discovery Technologies Limited
Email: divyesh_m@aurigene.com
Phone: +91- 80-71025588;Website: www.aurigene.com

Efficacy Evaluation

Efficacy evaluation is done in “Evaluable Population” - defined as patients who have had at least one follow up scan during study or were withdrawn due to clinical progression before the initial follow up scan. As of Oct. 15, 2018, 37 patients constitute “Evaluable Population”. Table 3 and Figure 1 provide efficacy evaluation from these patients.

Table 3: Clinical Benefit Rate (CBR) by irRC in solid tumors and by Lugano criterion for Hodgkin lymphoma

Tumor type	HNSCC (N=13)	NSCLC (N=10)	MSI-H positive solid tumors (N=5)	Hodgkin Lymphoma (N=9)	Total (N=37)
CBR at 400 mg; n/N (%)	4/7 (57.1)	6/7 (85.7)	0/1 (0)	4/4 (100)	**14/19 (73.7)**
CBR at 800 mg; n/N (%)	2/6 (33.3)	1/3 (33.3)	2/4 (50)	3/5 (60)	**8/18 (44.4)**
CBR (Total); n/N (%)	6/13 (46.2)	7/10 (70)	2/5 (40)	7/9 (77.8)	**22/37 (59.5)**

Overall CBR is 59.5% - with a strong trend towards superior benefit at lower dosage. The tendency of improved CBR is seen across all tumor types, except MSI-H where the sample size is smallest (5), and only one (1) patient was randomized to the lower dose (400 mg). The rates of CBR with 400 mg dosage is similar to the rates achieved with PD-1/PD-L1 antibodies[7]. In addition, clear efficacy signals seen among four patients also showed higher activity at 400 mg – out of three (3) patients with Hodgkin lymphoma achieving PR, two (2) received 400 mg; the HNSCC patient having a 48% reduction was also at 400 mg (Figures 1, 2 and 3).

Figure 1: Percentage change from baseline in SPD by irRC criterion in solid tumors and in SUVmax by Lugano criterion in Hodgkin lymphoma.

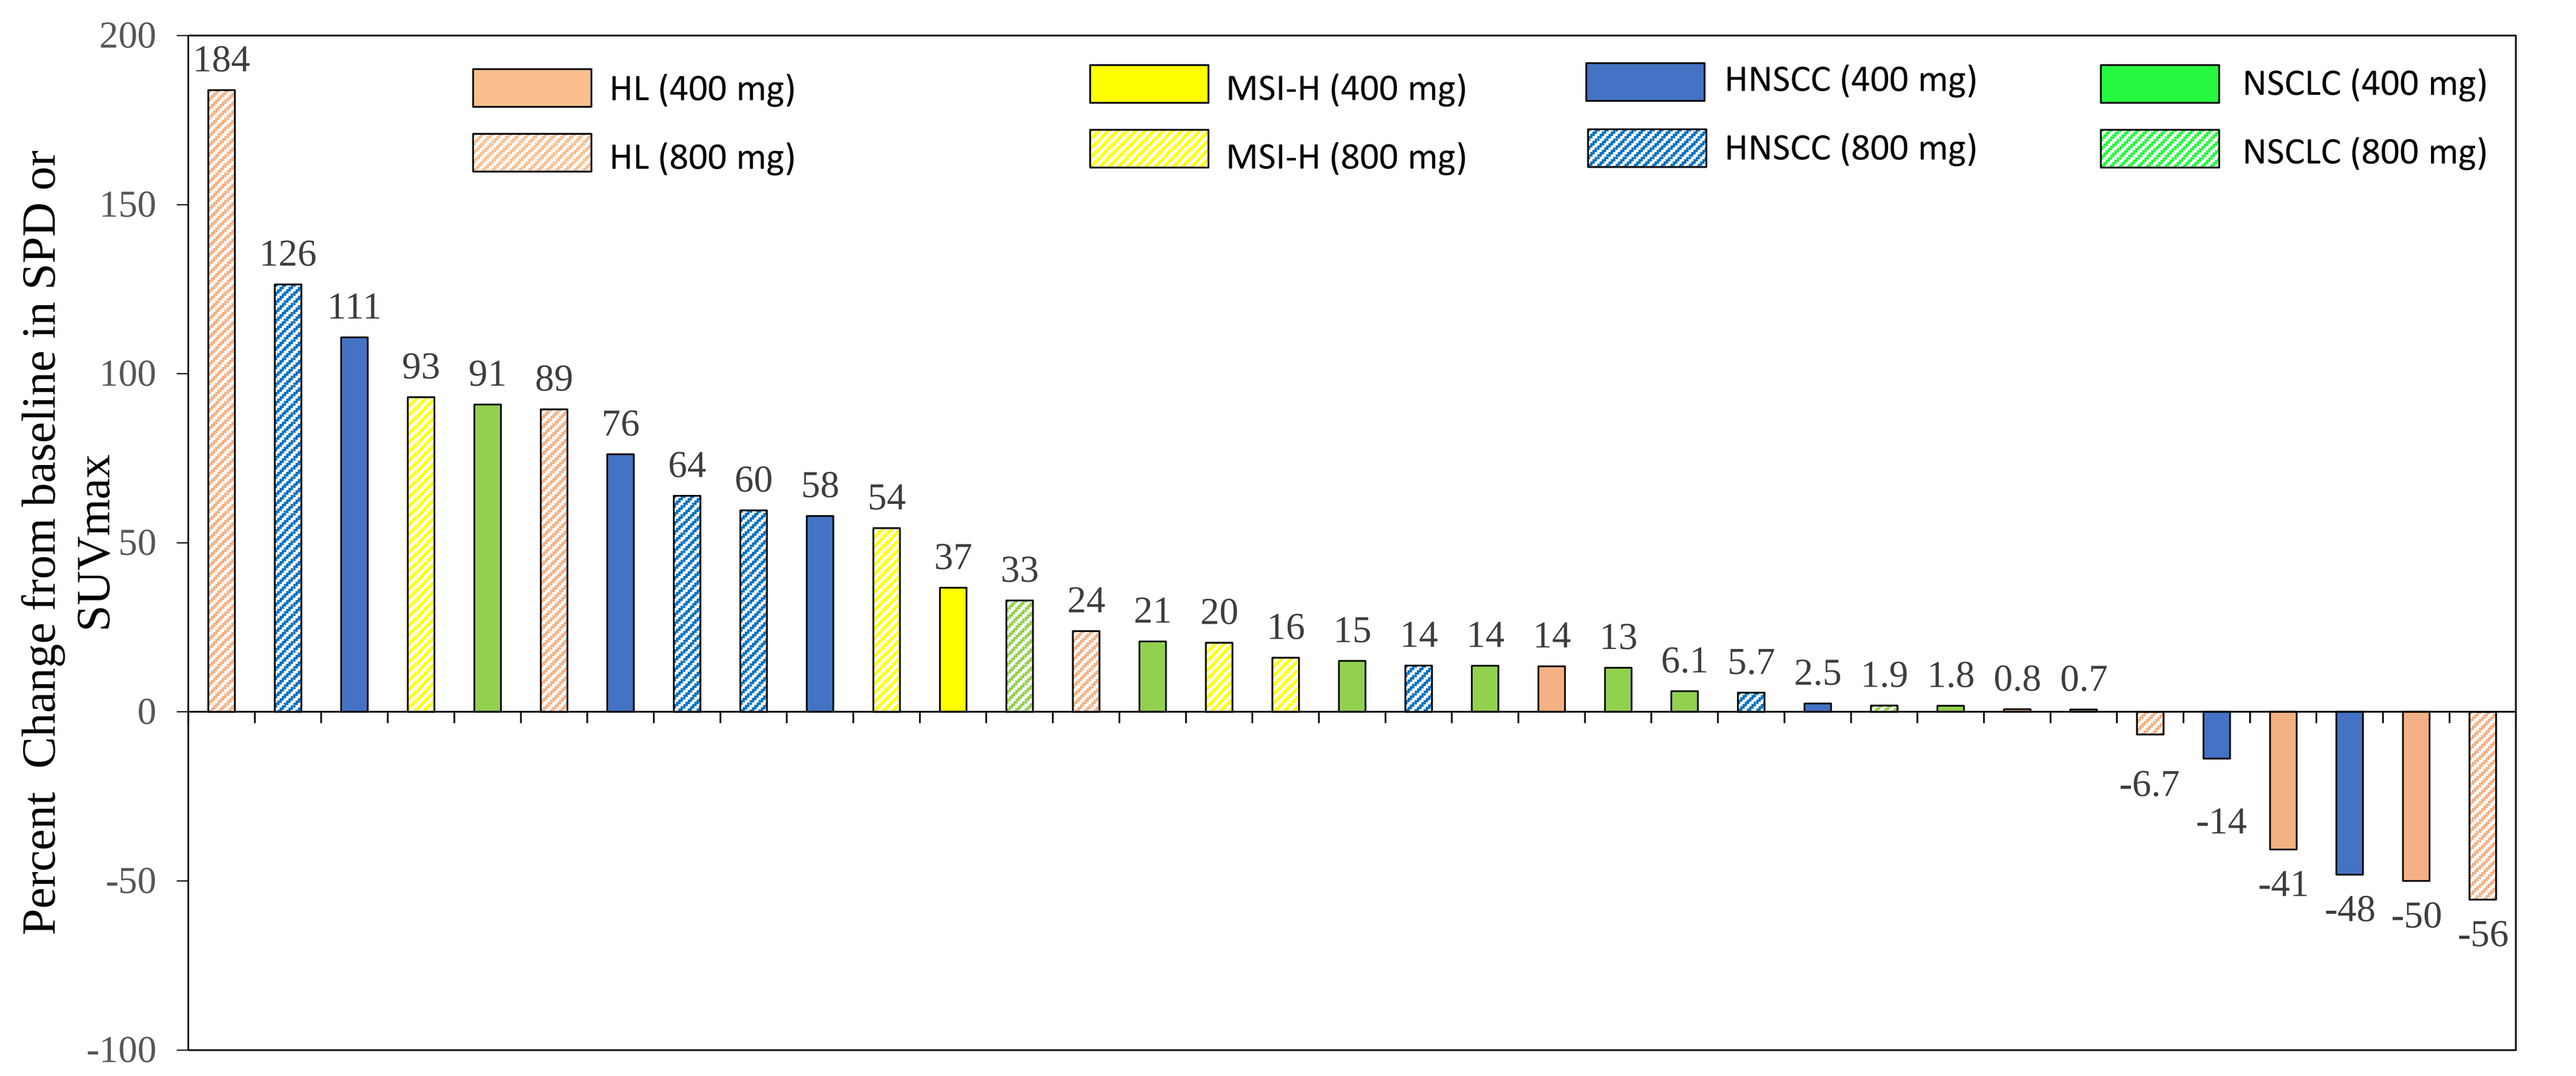

Percent Change from baseline in SPD or SUVmax
200
150
100
50
0
-50
-100
HL (400 mg)
HL (800 mg)
MSI-H (400 mg)
MSI-H (800 mg)
HNSCC (400 mg)
HNSCC (800 mg)
NSCLC (400 mg)
NSCLC (800 mg)
184 126 111 93 91 89 76 64 60 58 54 37 33 24 21 20 16 15 14 14 14 13 6.1 5.7 2.5 1.9 1.8 0.8 0.7 -6.7 -14 -41 -48 -50 -56

Figure 2: Treatment effect in Hodgkin Lymphoma (patient at 400 mg)

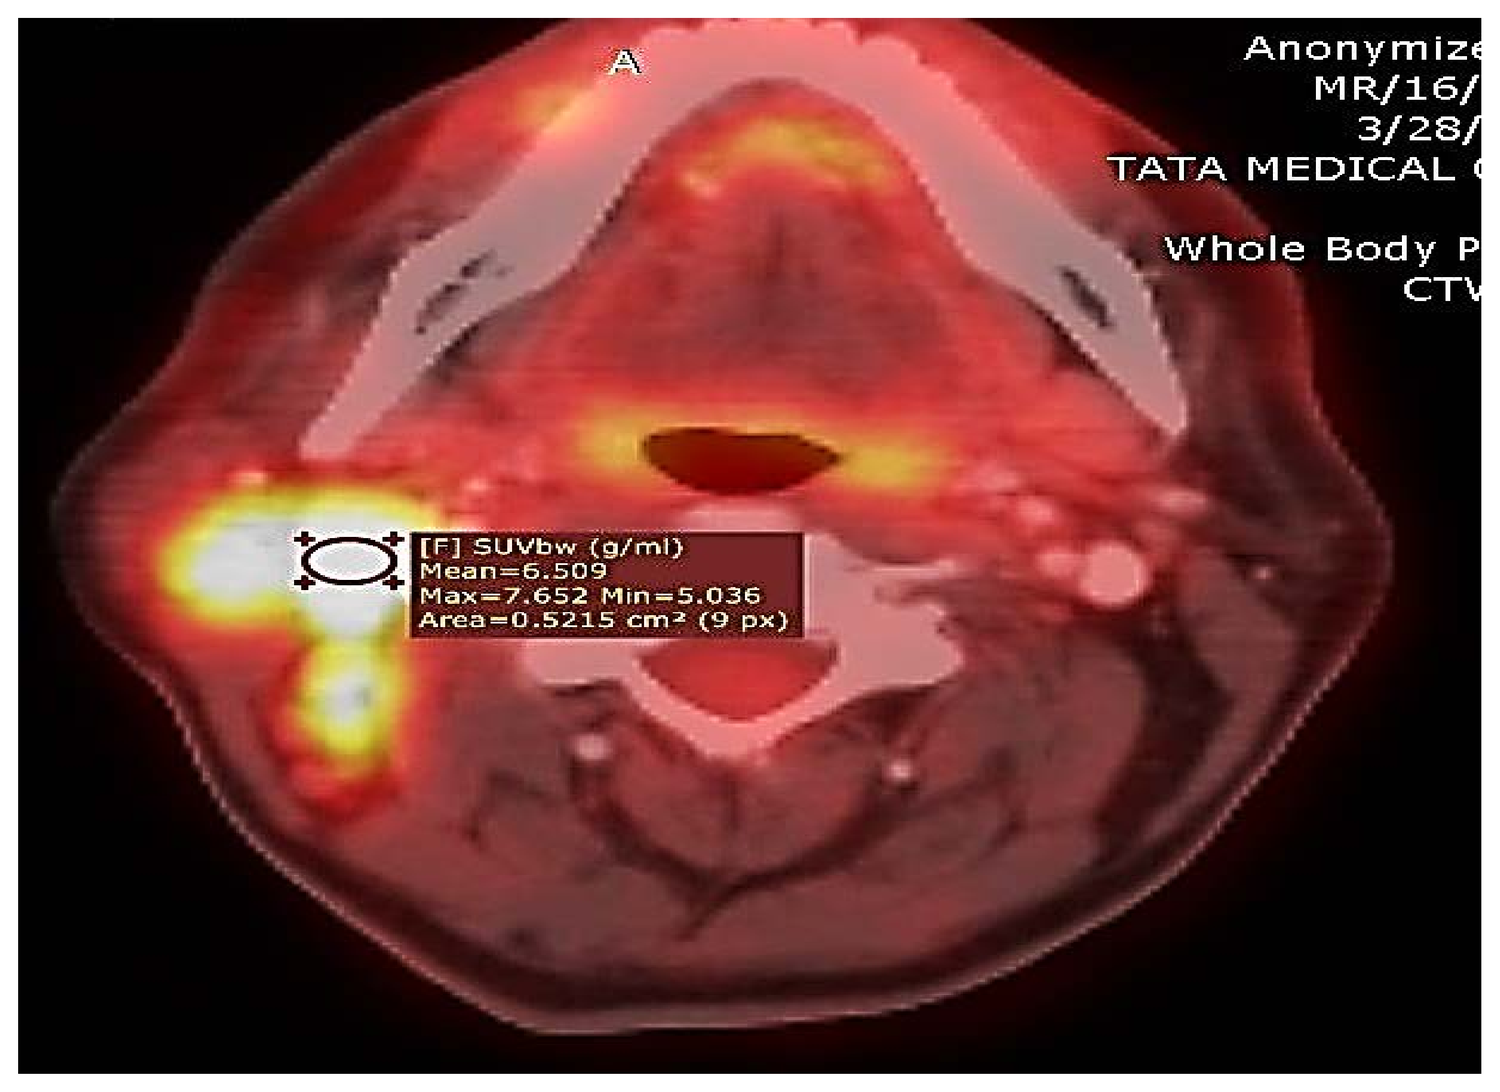

Baseline lesion

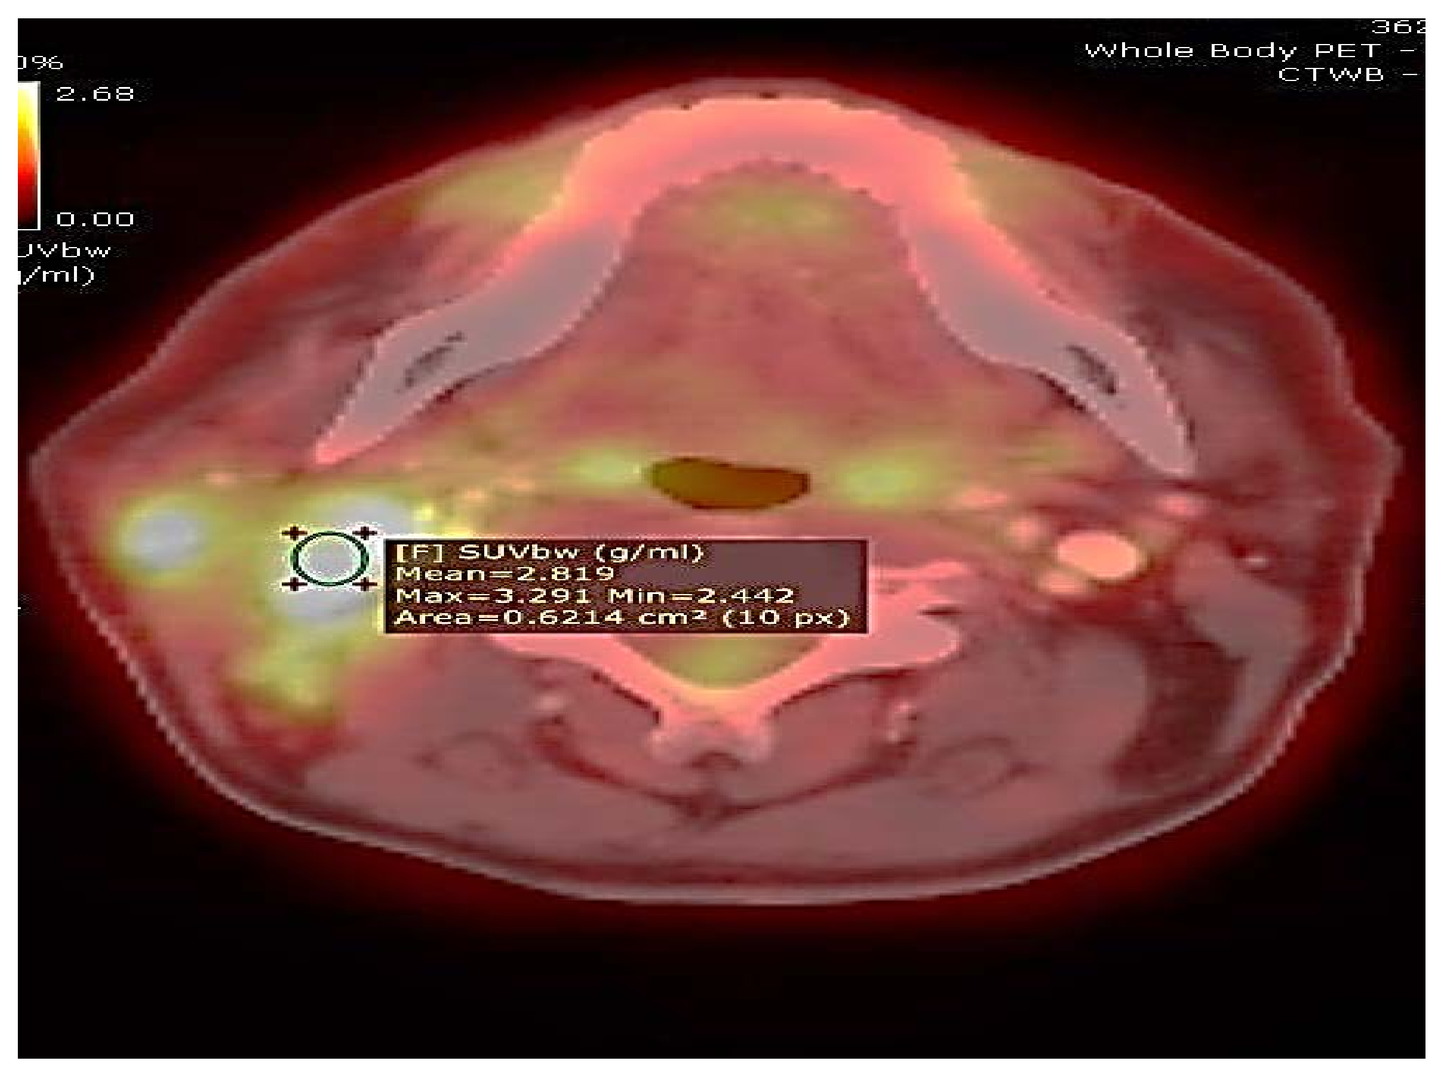

Same lesion at cycle 4 with 57% reduction in SUV

Figure 3: Treatment effect in Head and Neck Cancer (patient at 400 mg)

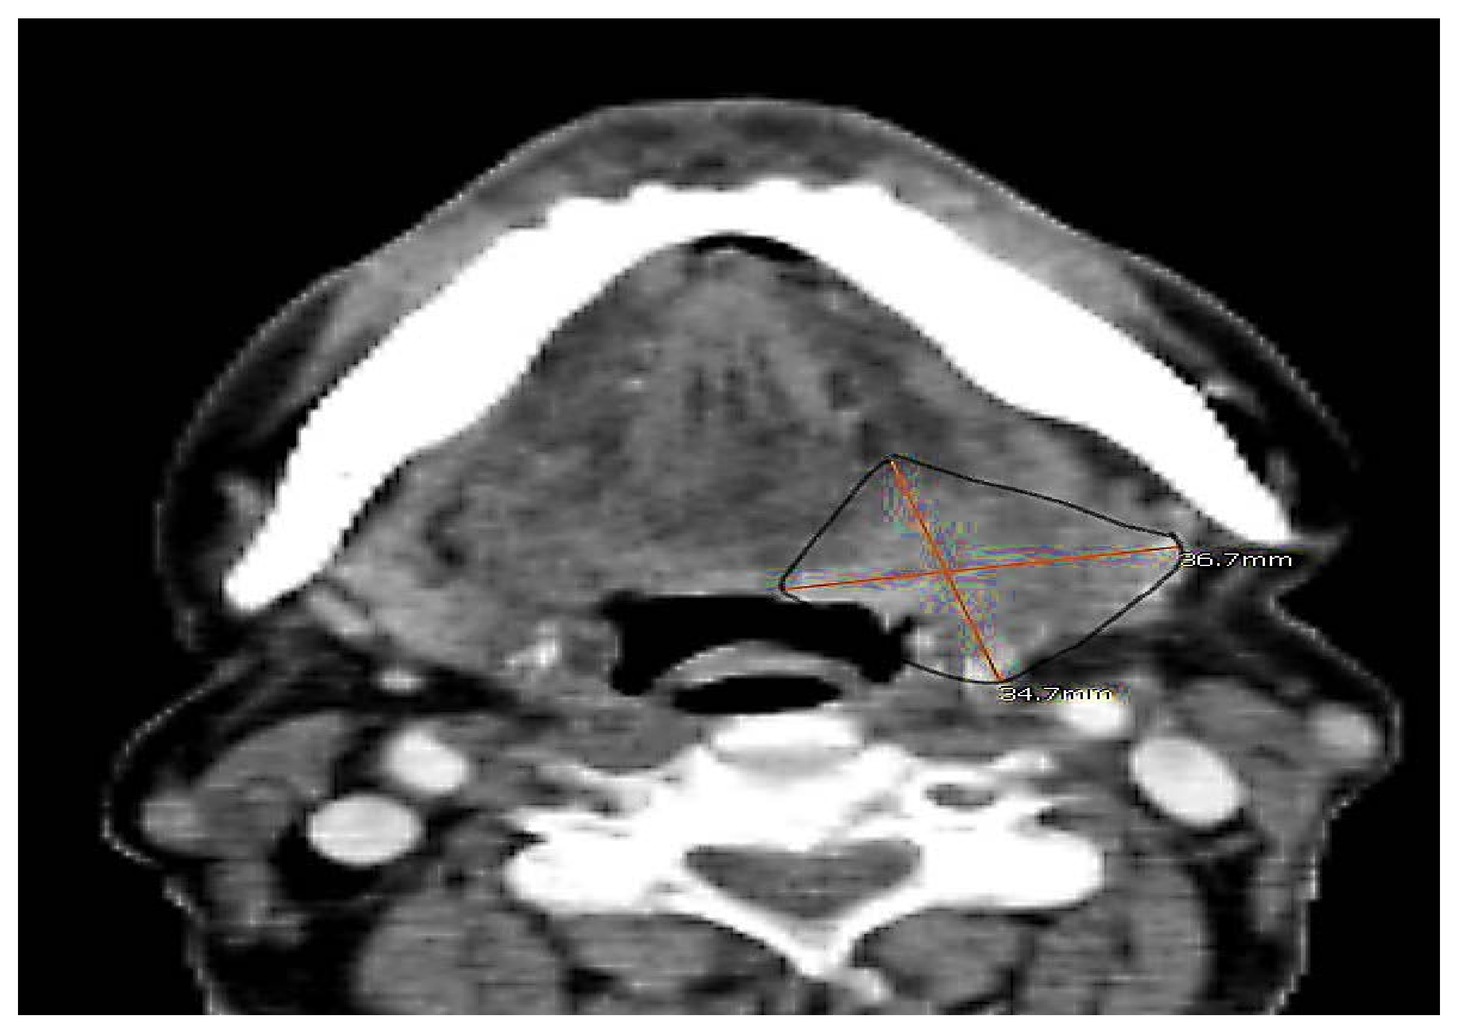

Baseline lesion

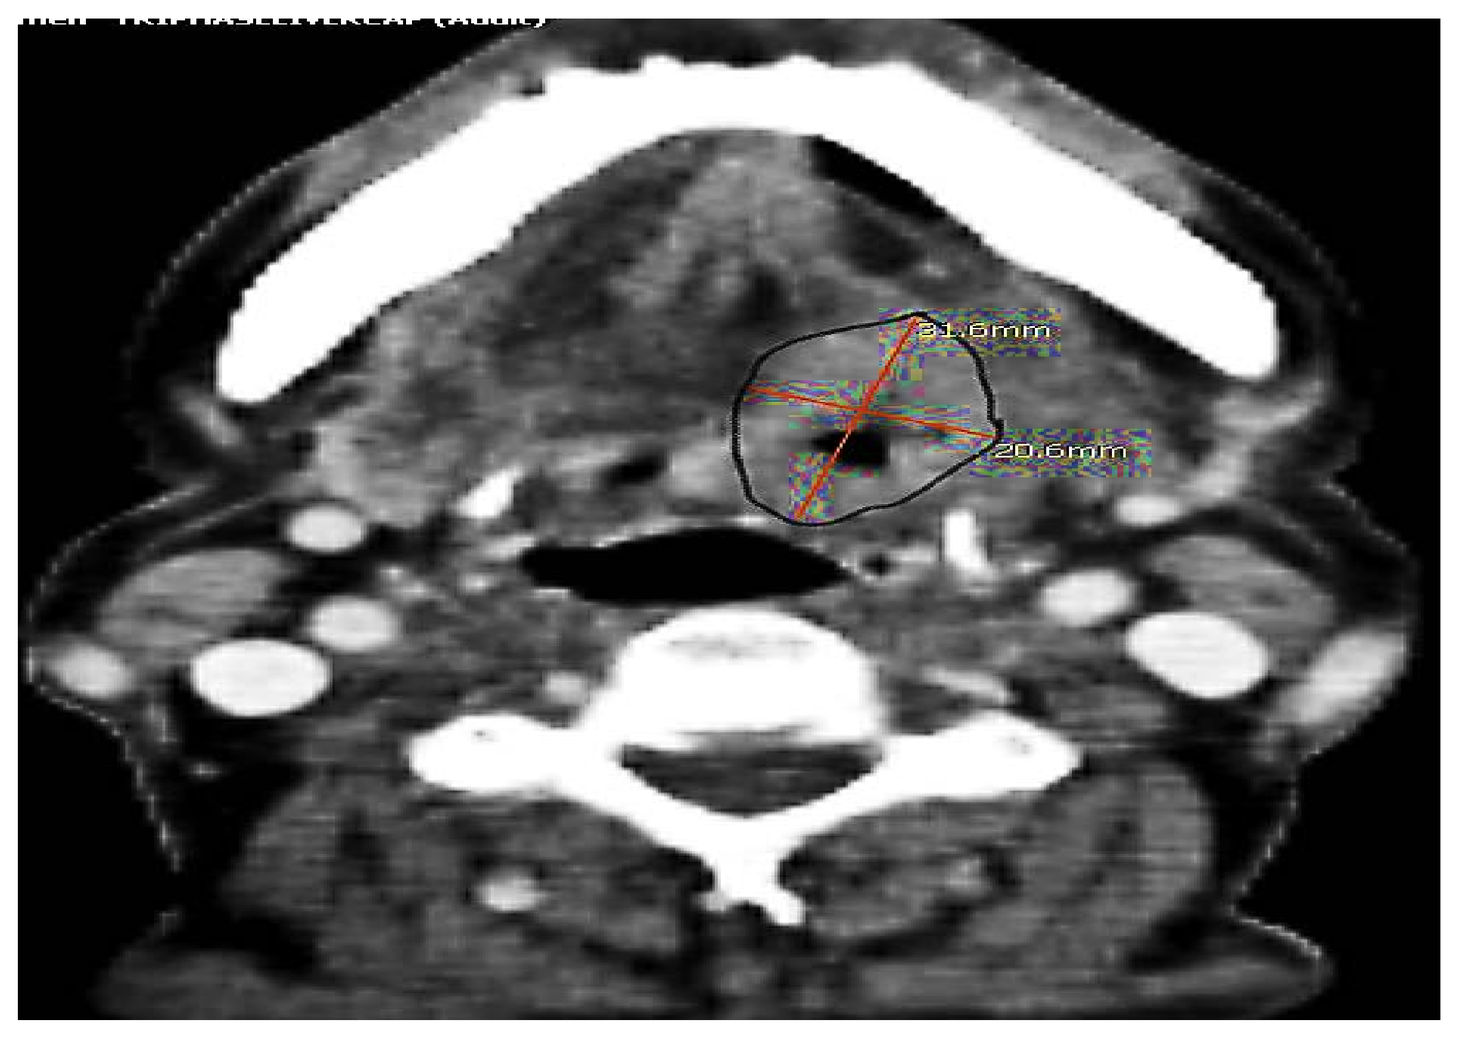

Same lesion at cycle 2 with 48% reduction in SPD

In Vitro Immune Activation with CA-170

Figure 4: Functional antagonism of PD-L1/L2 and VISTA with CA-170

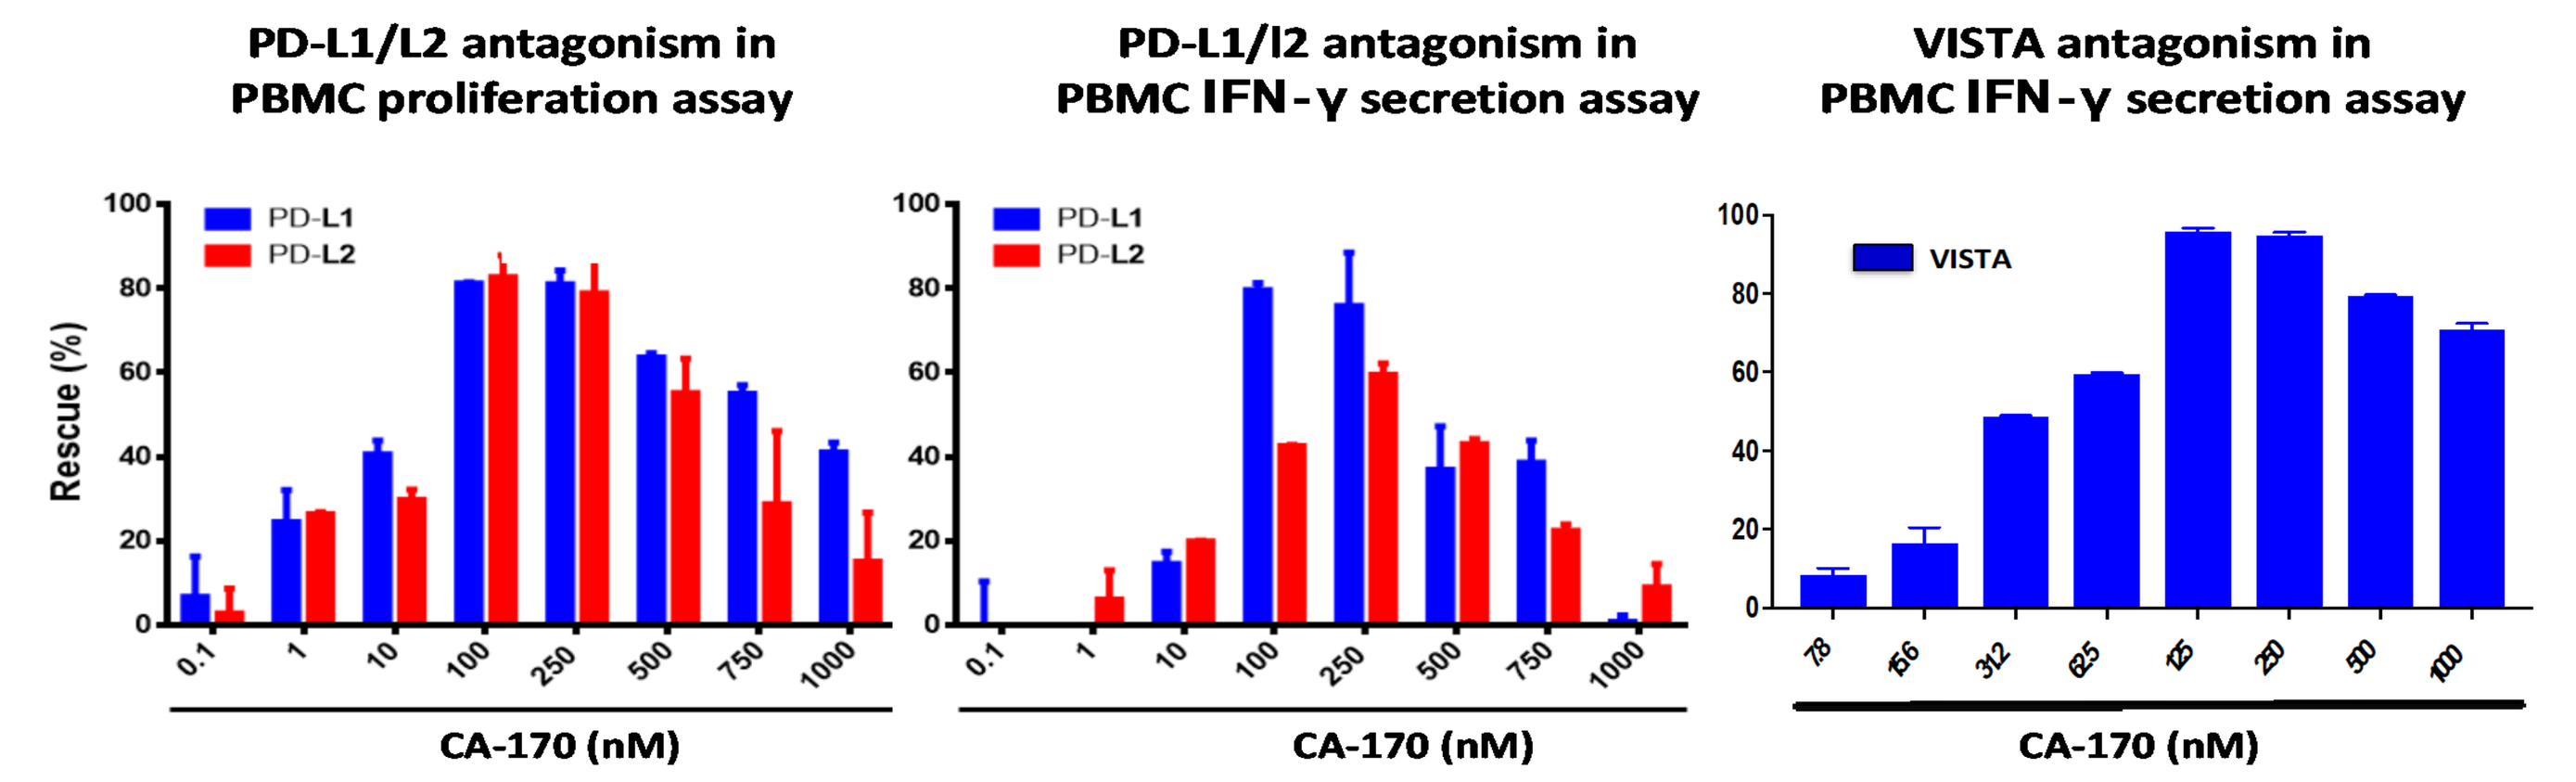

PD-L1/L2 antagonism in PBMC proliferation assay
PD-L1/l2 antagonism in PBMC IFN-γ secretion assay
VISTA antagonism in PBMC IFN-γ secretion assay
Rescue (%)
PD-L1
PD-L2
VISTA
CA-170 (nM)

CA-170 shows a bell shaped immune activation in PBMC-based assays with decrease in activity at higher concentration as monitored by rescue of proliferation or IFN-Y secretion inhibited by PD-L1/L2 or VISTA. Similar bell-shaped response has been reported for other immunomodulators[8].

Safety

Safety is evaluated among all 62 patients who received CA-170. Overall, CA170 has been well tolerated, with Immune Related Adverse Events (irAEs) seen in eight patients – five (5) with increase in TSH or worsening hypothyroidism (2 at 400 mg and 3 at 800 mg), two (2) with skin rash (both at 400 mg) and one (1) with Grade 3 neutropenia and Grade 3 anemia (at 400 mg).

The patient (with related Grade 3 neutropenia / Grade 3 anemia) developed lower grade cytopenias within Cycle 1. The counts continued to worsen, necessitating CA-170 interruption in Cycle 3. The hematological values improved after drug interruption, and the counts worsened again when CA-170 was resumed (Figure 5).

Figure 5: Changes in Absolute Neutrophil Count (ANC) and Hemoglobin in a HNSCC Patient (400 mg)

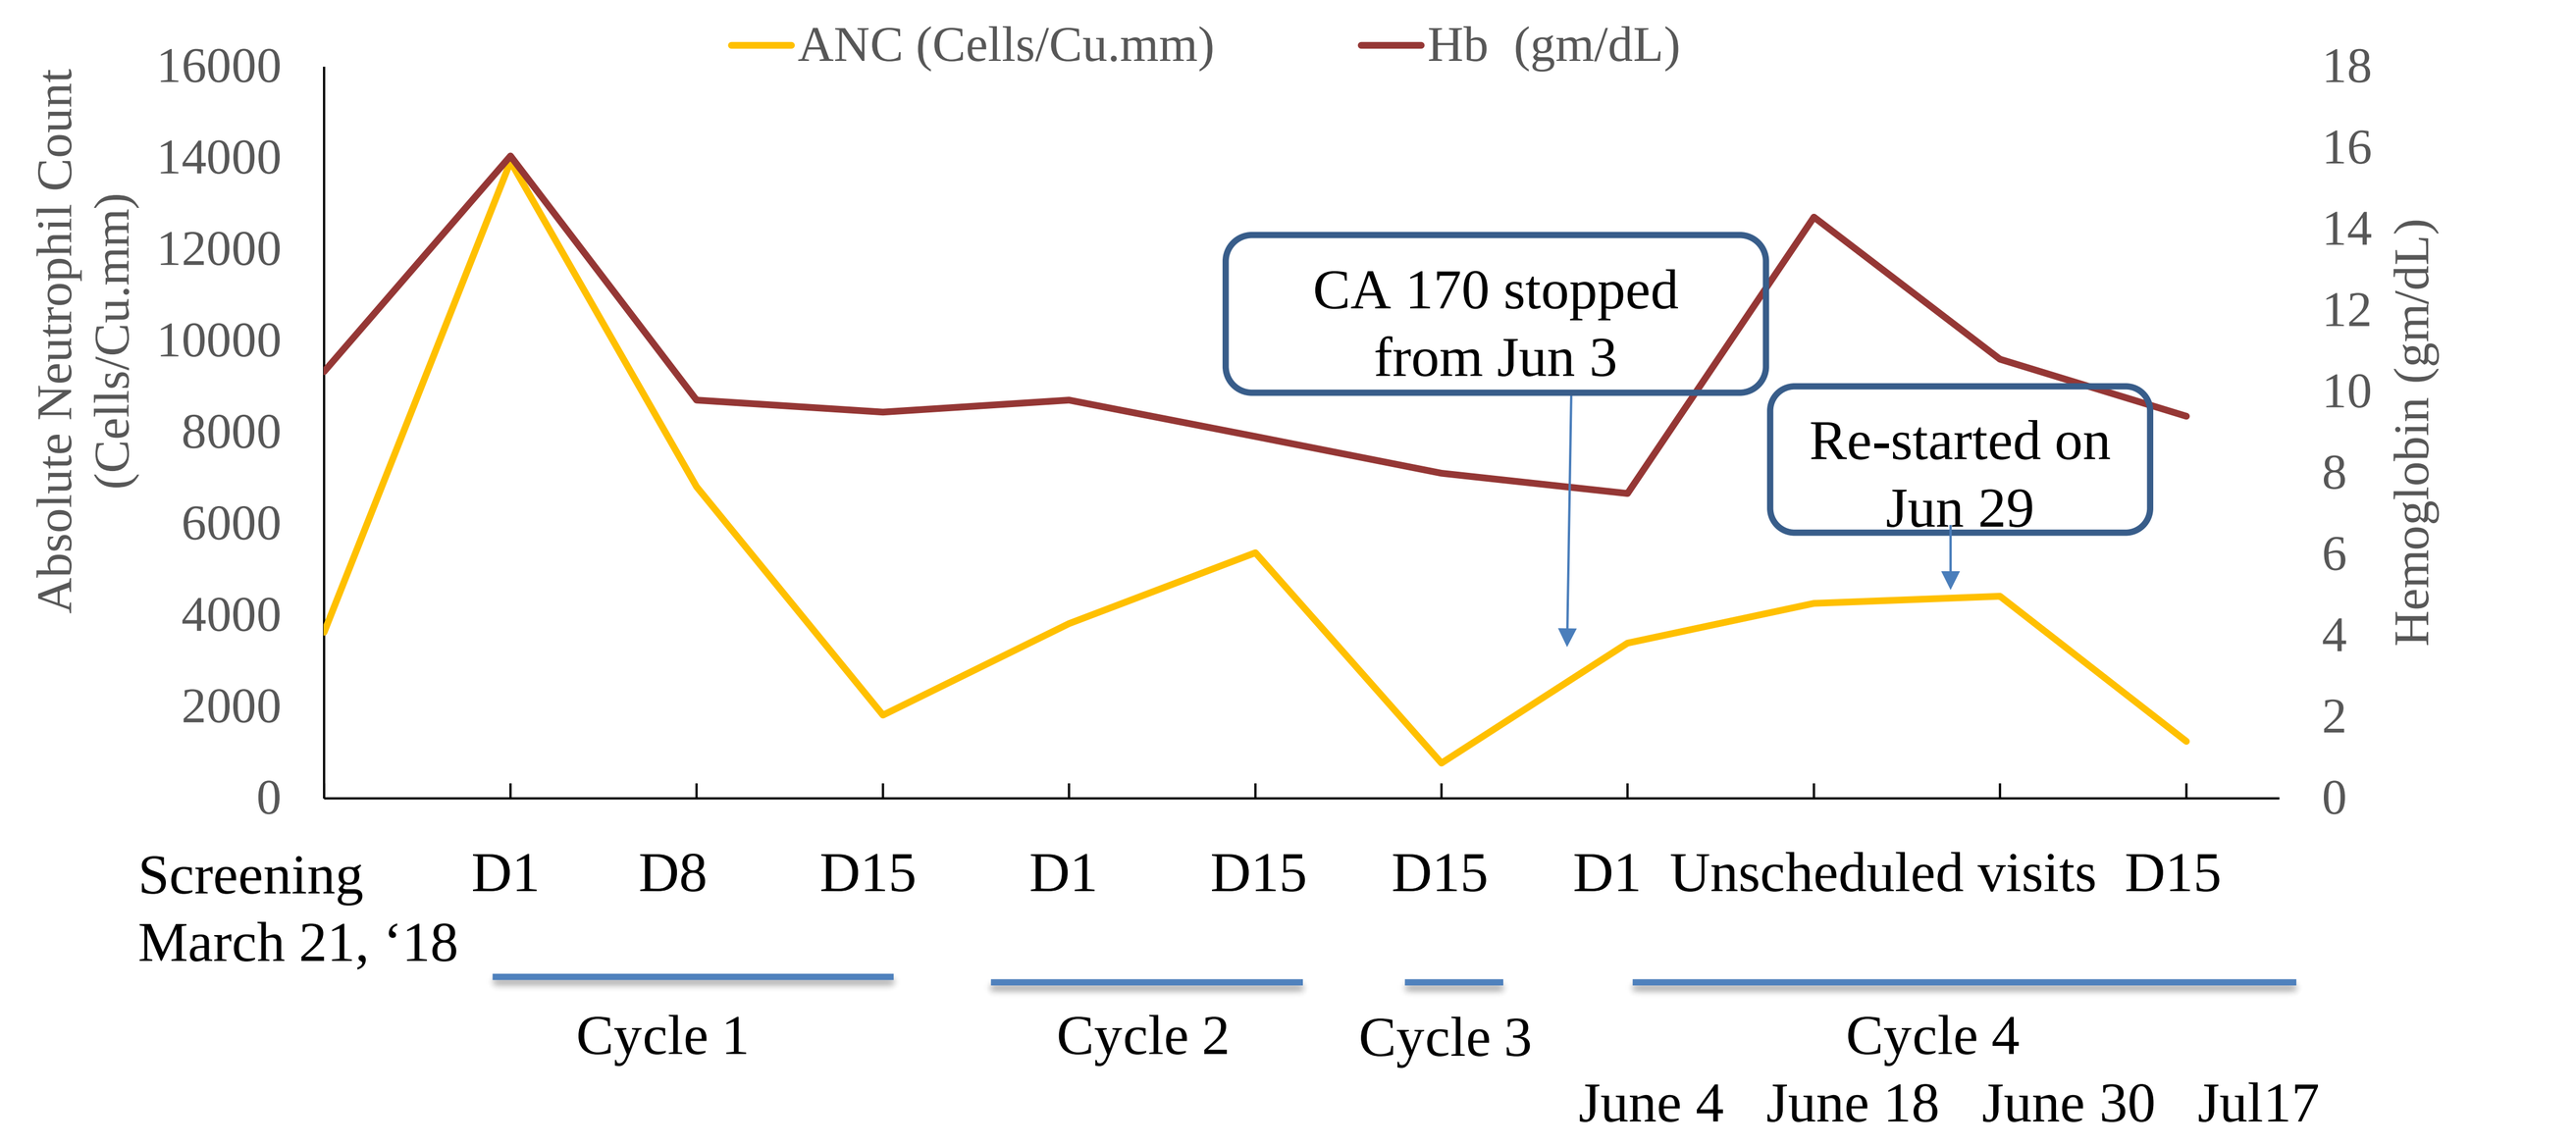

ANC (Cells/Cu.mm)
Hb (gm/dL)
Absolute Neutrophil Count (Cells/Cu.mm)
Hemoglobin (gm/dL)
CA 170 stopped from Jun 3
Re-started on Jun 29
Screening March 21, ‘18
D1 D8 D15 D1 D15 D15 D1 Unscheduled visits D15
Cycle 1
Cycle 2
Cycle 3
Cycle 4
June 4 June 18 June 30 Jul17

Conclusions

- The Clinical Benefit Rate (CBR) with CA-170 is in a similar range as PD-1/PD-L1 antibodies[7] with Objective Response Rates being lower.
- Better efficacy results are observed at the lower dosage (400 mg), consistent with pre-clinical findings showing bell-shaped curve of immune activation with CA-170 (Figure 4).
- Higher proportion of patients at 400 mg developed Immune Related Adverse Events (irAEs).
- Immune related hematological events, reported either as fatal or of prolonged duration with antibodies[9,10], have been reversible and of shorter duration.
- The CA-170 dose and dosing regimen are being optimized in relevant tumor types to achieve higher responses, in view of short half life[1] and PD data showing bell shaped immune activation.

References

1. Bang YJ, et al., abstract poster 341, Phase 1 trial of CA-170, a first-in-class, orally available, small molecule immune checkpoint inhibitor (ICI) dually targeting PD-L1 and VISTA, in patients with advanced solid tumors or lymphomas. SITC 2017.
2. Eisenhauer EA, et al. New response evaluation criteria in solid tumors: Revised RECIST guideline (version 1.1). Eur J Can. 2009;45:228-247.
3. Cheson BD, et al. International Harmonization Project on Lymphoma. Revised response criteria for malignant lymphoma. J Clin Oncol. 2007;25(5):579-86.
4. Wolchok JD, et al. Guidelines for the Evaluation of Immune Therapy Activity in Solid Tumors: Immune-Related Response Criteria. Clin Cancer Res. 2009;15(23):7412–20.
5. Cheson BD, et al. Recommendations for initial evaluation, staging, and response assessment of Hodgkin and non-Hodgkin lymphoma: the Lugano classification. J Clin Oncol. 2014 Sep 20;32(27):3059-68.
6. Van Heertum RL, et al. Lugano 2014 criteria for assessing FDG-PET/CT in lymphoma: an operational approach for clinical trials. Drug Des Devel Ther. 2017 Jun 13;11:1719-1728.
7. Alberto Carretero-González, et al. Analysis of response rate with ANTI PD1/PD-L1 monoclonal antibodies in advanced solid tumors: a meta-analysis of randomized clinical trials. Oncotarget 2018 Feb 2; 9(9): 8706–8715.
8. Fisher et al. Targeting of 4-1BB by monoclonal antibody PF-05082566 enhances T-cell function and promotes anti-tumor activity. Cancer Immunol Immunother 2012, 61:1721
9. Atwal D, et al. Pembrolizumab-Induced Pancytopenia: A Case Report. Perm J 2017;21:17-004
10. Zachary Wright et al. High-grade neutropenia in a patient successfully treated with nivolumab for refractory primary mediastinal B-cell lymphoma. Blood Adv. 2017 Jul 25; 1(17): 1306–1308.